Poster # **372P**

Frederick M. Howard | frederick.howard@bsd.uchicago.edu

A NATALEE data–based machine learning model to predict distant recurrence and treatment effect in real-world patients with HR+/HER2– early breast cancer without CDK4/6 inhibitor treatment

Frederick M. Howard,[1] Peter A. Fasching,[2] Cesar A. Santa-Maria,[3] Elgene Lim,[4,5] Joseph A. Sparano,[6] Maryam B. Lustberg,[7] Thomas Bachelot,[8] Oleg Blyuss,[9] Christine Brezden-Masley,[10] Yeon Hee Park,[11] Murat Akdere,[12] Fen Ye,[13] Kristyn Pantoja,[14] Patricia Dominguez Castro,[15] Christoph Kurz,[16] Pedram Razavi[17]

[1]Department of Medicine, University of Chicago, Chicago, IL, USA; [2]University Hospital Erlangen Comprehensive Cancer Center Erlangen-EMN, Friedrich Alexander University Erlangen-Nuremberg, Erlangen, Germany; [3]Breast and Gynecological Malignancies Group, Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins, Baltimore, MD, USA; [4]Garvan Institute of Medical Research, Sydney, NSW, Australia; [5]Faculty of Medicine, St Vincent's Clinical School, University of New South Wales, Sydney, NSW, Australia; [6]Icahn School of Medicine at Mount Sinai, Tisch Cancer Institute, New York, NY, USA; [7]Department of Medicine, Yale University School of Medicine, New Haven, CT, USA; [8]Department of Medical Oncology, Centre Léon-Bérard, Lyon, France; [9]Centre for Cancer Screening, Prevention, and Early Diagnosis, Wolfson Institute of Population Health, Queen Mary University of London, London, UK; [10]Division of Medical Oncology and Hematology, Faculty of Medicine, Mount Sinai Hospital, University of Toronto, Toronto, ON, Canada; [11]Division of Hematology-Oncology, Department of Medicine, Samsung Medical Center, Sungkyunkwan University School of Medicine, Seoul, Republic of Korea; [12]Novartis Pharma AG, Basel, Switzerland; [13]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [14]Novartis Pharmaceuticals Corporation, Cambridge, MA, USA; [15]Novartis Ireland, Dublin, Ireland; [16]Novartis Pharma GmbH, Munich, Germany; [17]Memorial Sloan Kettering Cancer Center, New York, NY, USA

KEY FINDINGS & CONCLUSIONS

- This analysis validated an RW data–trained ML model[5] using NATALEE trial data to predict distant recurrence in RW patients with HR+/HER2– EBC
- Additionally, a NATALEE data–trained model was able to predict DR and RIB treatment effect in RW patients with HR+/HER2– EBC
- These findings support the use of both RW and clinical trial data in personalized prediction of DR risk and treatment benefit
- This approach can inform optimization of clinical decision-making in the HR+/HER2– EBC population
- Developing models with more extensive and diverse data and additional clinical and genomic variables may improve model performance and generalizability
- This NATALEE-trained ML model has the potential to estimate individual RW benefit from CDK4/6i treatment and guide treatment personalization for patients

Scan to obtain:
- Poster

https://tinyurl.com/MHoward372P

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study is sponsored by Novartis Pharma AG.
Poster presented at the ESMO Congress 2025, 17-21 October 2025; Berlin, Germany

INTRODUCTION

- Despite standard-of-care adjuvant endocrine therapy (ET), many patients with hormone receptor–positive (HR+)/human epidermal growth factor receptor 2–negative (HER2−) early breast cancer (EBC) still experience distant recurrence (DR)[1-3]
- The phase 3 NATALEE trial demonstrated a statistically significant invasive disease-free survival benefit with ribociclib (RIB) + nonsteroidal aromatase inhibitor (NSAI) vs NSAI alone in patients with high-risk HR+/HER2− EBC,[3] with a sustained benefit after all patients were off RIB (HR, 0.72; 95% CI: 0.61-0.84; *P*<.0001)[4]
- We previously trained a machine learning (ML) model on real-world (RW) data that was able to accurately predict DR risk (concordance index [C-index], 0.86; integrated Brier score [IBS], 0.05)[5]
- This study aimed to externally validate this model using NATALEE data and assess whether RIB treatment effect can be predicted in an RW cohort of patients with HR+/HER2− EBC

METHODS

- Previously, an elastic net–penalized Cox proportional hazards (PH) model was trained with data from RW patients aged ≥18 years in the US-based, electronic health record–derived deidentified Flatiron Health Research Database[6] with stage I-III HR+/HER2− EBC who had undergone surgery and initiated adjuvant ET, but not cyclin-dependent kinase 4/6 inhibitor (CDK4/6i) treatment (1 January 2011 to 30 April 2024)[5]
- To validate and extend the ML model, the current analysis was conducted in 5 steps (**Figure 1**), using NATALEE trial data (data cutoff: 29 April 2024)
- The Harrell C-index (0.5-1.0; 1.0 = perfect concordance) and IBS (0-0.25; 0 = perfect concordance) were used to compare predicted vs actual outcomes for each model; *P* values were determined by Wald test and are reported at the α=0.05 level

Figure 1. Five-Step Study Design

1. Comparison of RW and NATALEE populations →
- Key variables were compared by distribution analysis of variables
- Kaplan-Meier curves were used to compare survival outcomes

2. Adaptation of RW data–trained ML model to NATALEE data →
- The RW data–trained ML model was refitted using NATALEE trial data to weight predictor variables

3. External validation of the RW data–trained ML model on NATALEE data →
- The generalizability of the RW data–trained ML model was validated with the NATALEE NSAI alone–treated cohort using 10 predictor variables

4. Estimation of RIB treatment effect using a NATALEE data–trained ML model →
- A weighted Cox PH model was fitted with the overall NATALEE trial population using the 10 predictor variables as well as RIB treatment status as a covariate

5. Determination of the predictive ability of the NATALEE data–trained ML model using RW data
- The NATALEE data–trained Cox PH model was applied to RW patients with stage I-III EBC, assuming all patients received NSAI or RIB + NSAI

EBC, early breast cancer; ML, machine learning; NSAI, nonsteroidal aromatase inhibitor; PH, proportional hazards; RIB, ribociclib; RW, real world.

RESULTS

Characteristics and DR in the RW and NATALEE NSAI Alone Populations

- A total of 7842 RW patients with stage I-III HR+/HER2– EBC who initiated adjuvant ET but not CDK4/6i treatment in the Flatiron Health database were eligible according to the inclusion and exclusion criteria[5]
 - Given that the NATALEE trial does not include patients with stage I EBC, only RW patients with stage II/III disease were included in analysis of cohort similarity
- Baseline characteristics in patients who received ET in the NATALEE NSAI alone treatment arm (n=2441)[4] were similar to those of the RW patients with stage II/III EBC (n=3276), except for age (median, 62 years in RW patients, 52 years in NATALEE NSAI patients; **Figure 2**)
- The percentage of patients free from DR at 48 months was 92.2% (382 DR events) in the RW cohort with stage II/III EBC and 87.3% (246 DR events) in the NATALEE NSAI alone arm cohort (**Figure 3**)

Figure 2. Characteristics of the RW Population With Stage II/III EBC and NSAI Alone–Treated NATALEE Population

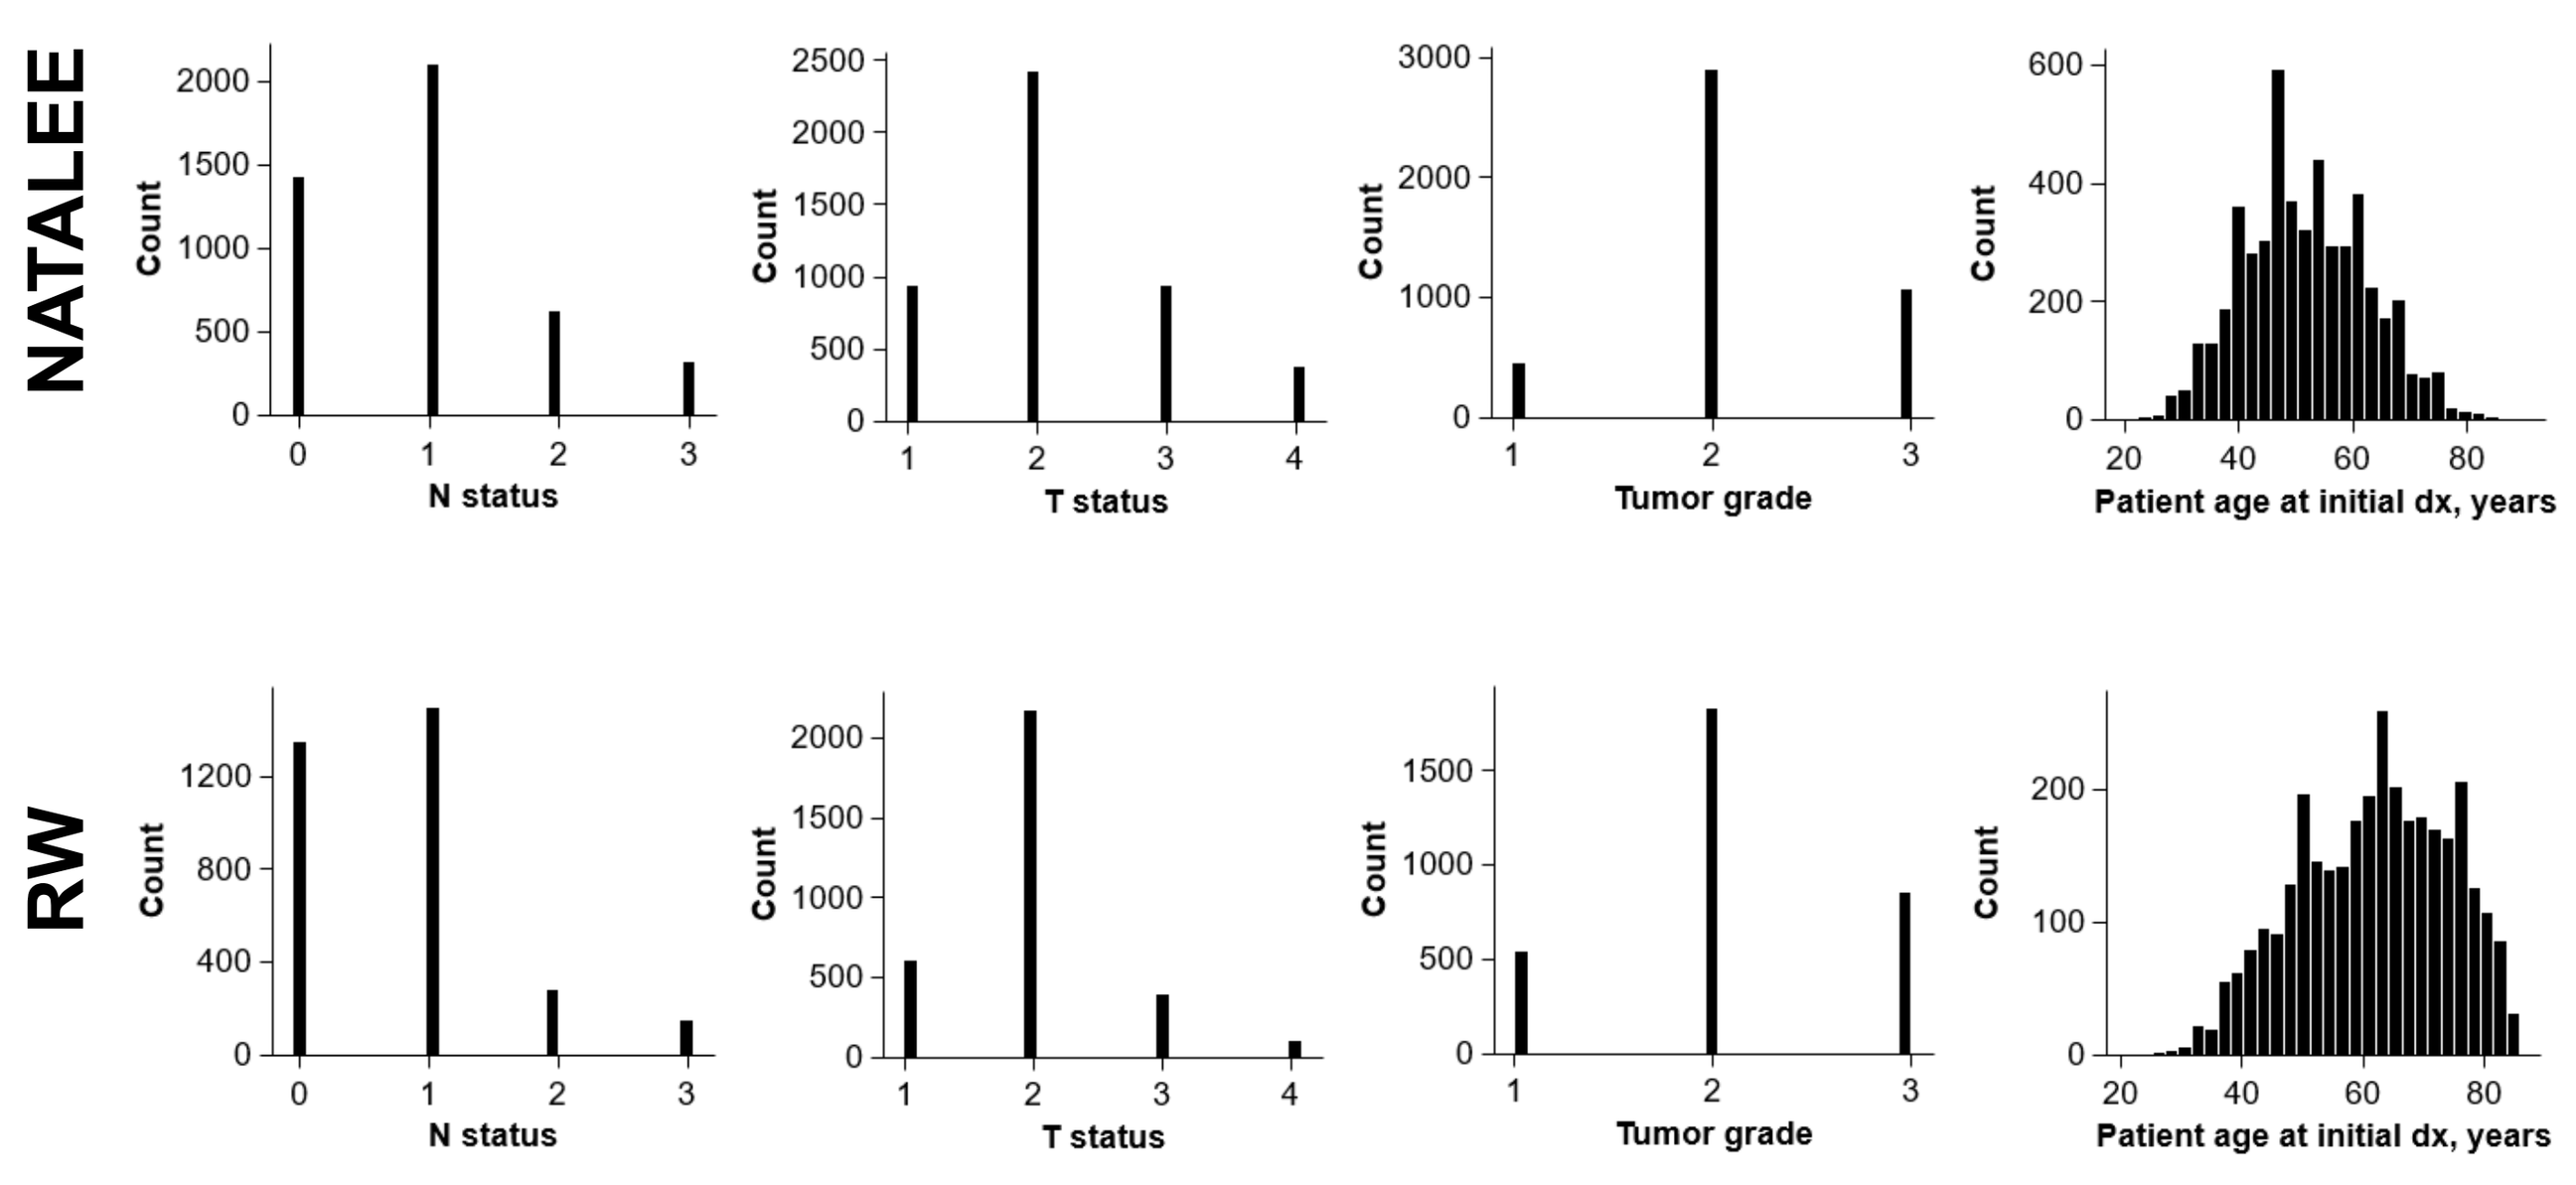
NATALEE
RW
Count
N status
T status
Tumor grade
Patient age at initial dx, years

EBC, early breast cancer; dx, diagnosis; N, nodal; NSAI, nonsteroidal aromatase inhibitor; RW, real world; T, tumor.

Figure 3. Percentage Free From DR of the RW Population With Stage II/III EBC and NSAI Alone–Treated NATALEE Population[a]

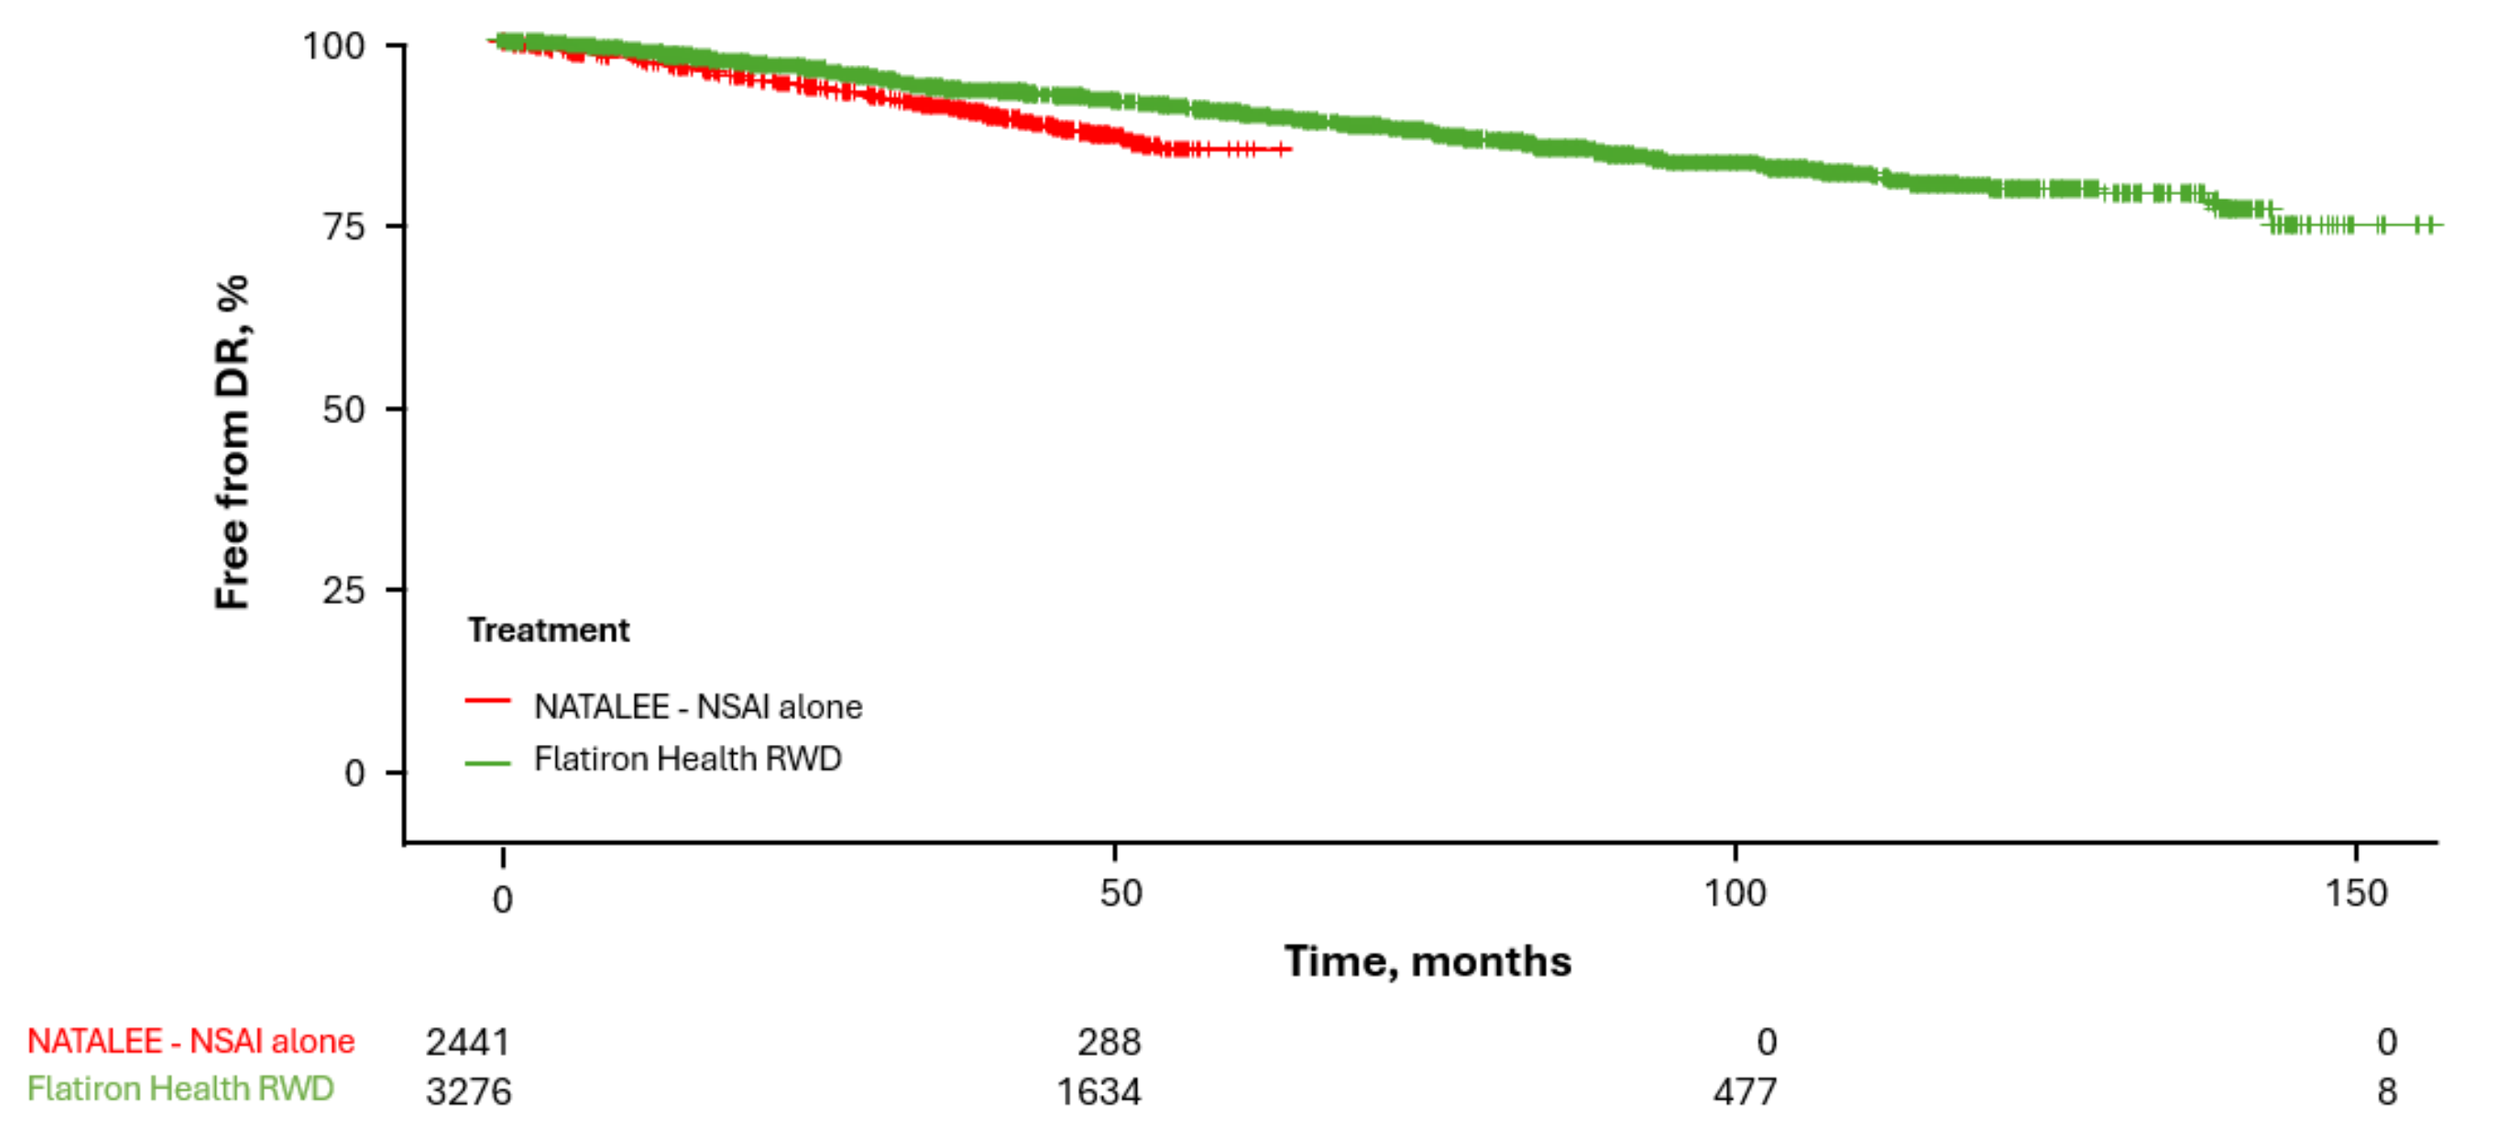
Free from DR, %
Time, months
Treatment
NATALEE - NSAI alone
Flatiron Health RWD
NATALEE - NSAI alone 2441 288 0 0
Flatiron Health RWD 3276 1634 477 8

DR, distant recurrence; EBC, early breast cancer; ET, endocrine therapy; NSAI, nonsteroidal aromatase inhibitor; RW, real world; RWD, real-world data. [a]Index date was at the start of adjuvant treatment for the RW population and at randomization for the population from the NATALEE trial (which allowed up to 12 months of prior ET at randomization).

External Validation of the RW Data–Trained ML Model on NATALEE Data

- The ML model developed on data for RW patients with stage I-III EBC was retrained with weighting of the 10 predictor variables based on differences between the RW population with stage II/III EBC and overall NATALEE population (**Figure 4**)
- Post weighting, the ML model was externally validated on the NATALEE NSAI alone arm cohort, with lower but still discriminatory performance (C-index, 0.66; IBS, 0.06)
 - Sensitivity analyses yielded similar results with model training on only patients with stage II/III EBC in the RW cohort (C-index, 0.65; IBS, 0.06) or without variable weighting (C-index, 0.66; IBS, 0.06)

Figure 4. Adjustment by Weighting of Predictor Variables for Population Differences in the RW Patients With Stage II/III EBC vs the Overall NATALEE Cohort

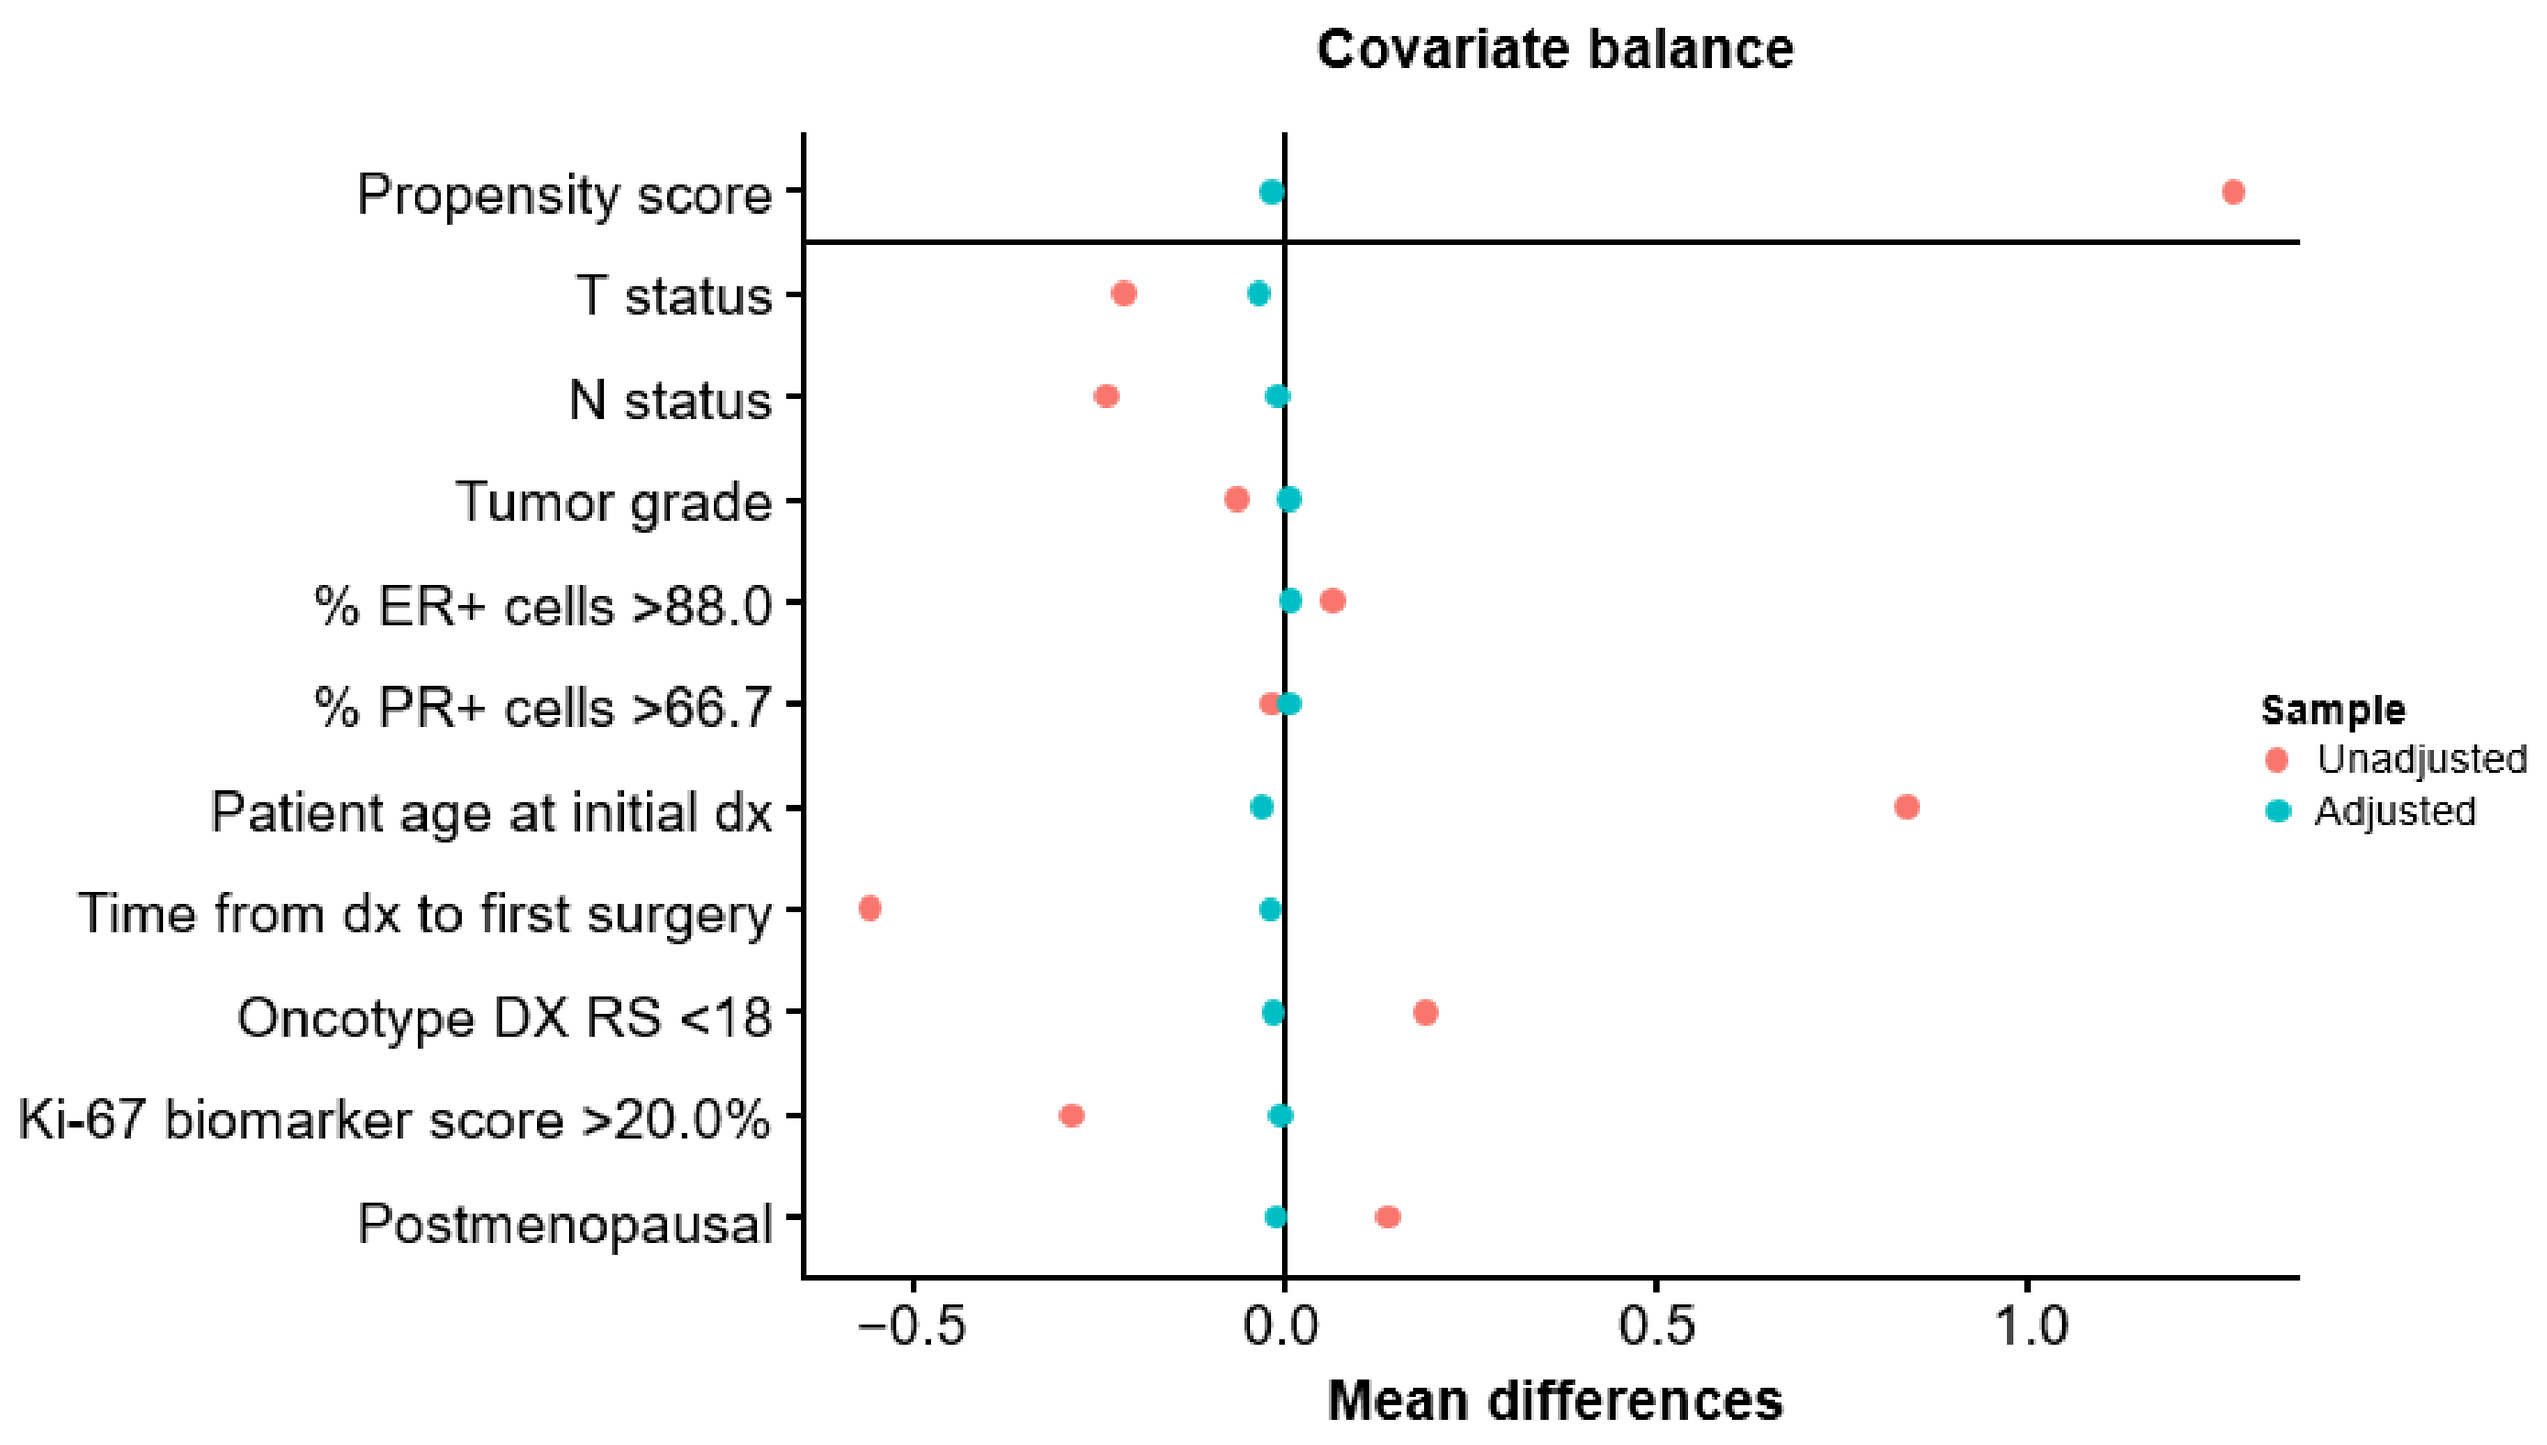
Covariate balance
Propensity score
T status
N status
Tumor grade
% ER+ cells >88.0
% PR+ cells >66.7
Patient age at initial dx
Time from dx to first surgery
Oncotype DX RS <18
Ki-67 biomarker score >20.0%
Postmenopausal
Mean differences
Sample
Unadjusted
Adjusted

dx, diagnosis; EBC, early breast cancer; ER, estrogen receptor; N, nodal; PR, progesterone receptor; RS, Recurrence Score; RW, real world; T, tumor.

Development of NATALEE Data–Trained ML Models to Predict DR Risk to Validate the Modeling Approach and to Predict RIB Treatment Effect

- To further validate this ML approach, another elastic net–penalized ML model developed with the same 10 predictor variables was trained on the NATALEE NSAI alone treatment arm, yielding a similar DR prediction accuracy (C-index, 0.70; IBS, 0.06)
- A weighted Cox PH model that included RIB treatment status as an additional 11th predictor variable trained on the overall NATALEE cohort also had a good predictive ability for DR risk (C-index, 0.68; IBS, 0.06)
 - The HRs for the contribution of the 10 variables used in previous model training indicated that the variables remained predictive (**Figure 5**), showing a similar direction and magnitude for each variable as in the original model[5]
- RIB treatment had an HR of 0.66 (95% CI: 0.55-0.80) in this model, representing a 34% reduction in relative DR risk with RIB + NSAI vs NSAI in the NATALEE cohort across the follow-up period (**Figure 5**)

Figure 5. Treatment Effect of RIB on Relative DR Risk in the Overall NATALEE Trial Population

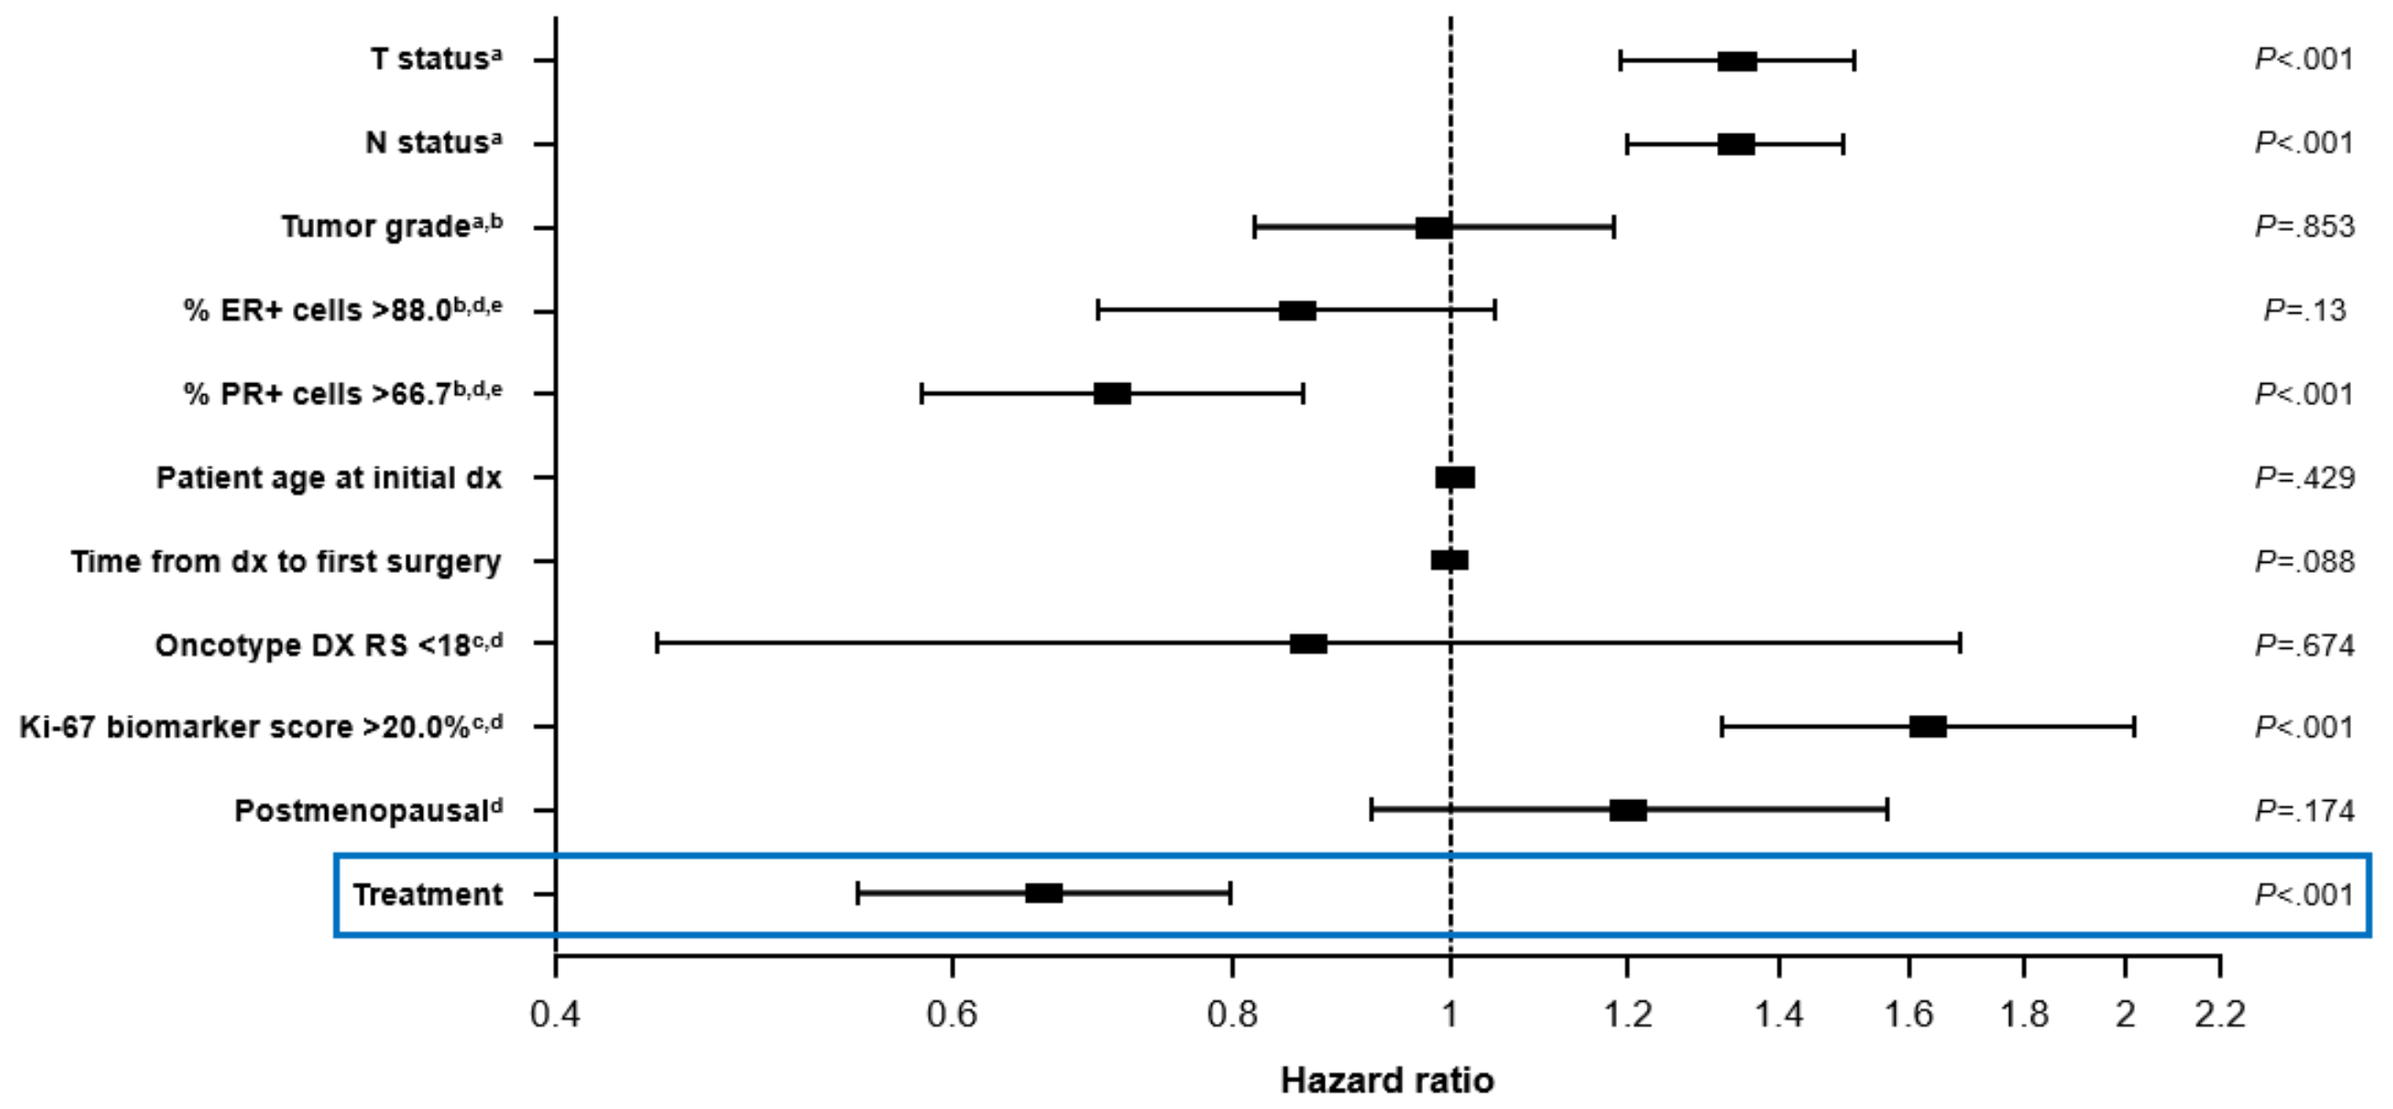
T status[a] P<.001
N status[a] P<.001
Tumor grade[a,b] P=.853
% ER+ cells >88.0[b,d,e] P=.13
% PR+ cells >66.7[b,d,e] P<.001
Patient age at initial dx P=.429
Time from dx to first surgery P=.088
Oncotype DX RS <18[c,d] P=.674
Ki-67 biomarker score >20.0%[c,d] P<.001
Postmenopausal[d] P=.174
Treatment P<.001
Hazard ratio

Blue box: Treatment was included as an 11th predictor variable in model training. DR, distant recurrence; dx, diagnosis; ER, estrogen receptor; ML, machine learning; N, nodal; PR, progesterone receptor; RIB, ribociclib; RS, Recurrence Score; T, tumor. [a]Ordinal variable treated as continuous. Linear assumption supported by data. Hazard ratio indicates change with one step of variable (eg, N0 to N1). [b]Multivariate imputation by chained equations used to impute missing values. [c]Unknown/not documented category created when summarizing baseline variables with >30% missing data was excluded from model analyses. [d]Binary variable indicating condition that is met. Hazard ratio indicates change associated with meeting stated condition. [e]Cutoff associated with recurrence according to ML decision tree method.

NATALEE Data–Trained ML Model Application to RW Cohort to Predict the RIB Treatment Effect in an RW Population

- Application of the weighted NATALEE data–trained ML model to the overall RW cohort (patients with stage I-III EBC and without CDK4/6i treatment) resulted in a predicted 2.4% reduction in absolute DR risk with RIB treatment at 36 months, and this reduction in absolute DR risk increased to 3.2% at 48 months (**Figure 6**)
 - Sensitivity analysis performed by training an unweighted Cox PH model on the NATALEE population produced a similar predicted reduction in absolute DR risk (36 months, 2.4%; 48 months, 3.2%)

Figure 6. Treatment Effect of RIB on Absolute DR Risk in the RW Population

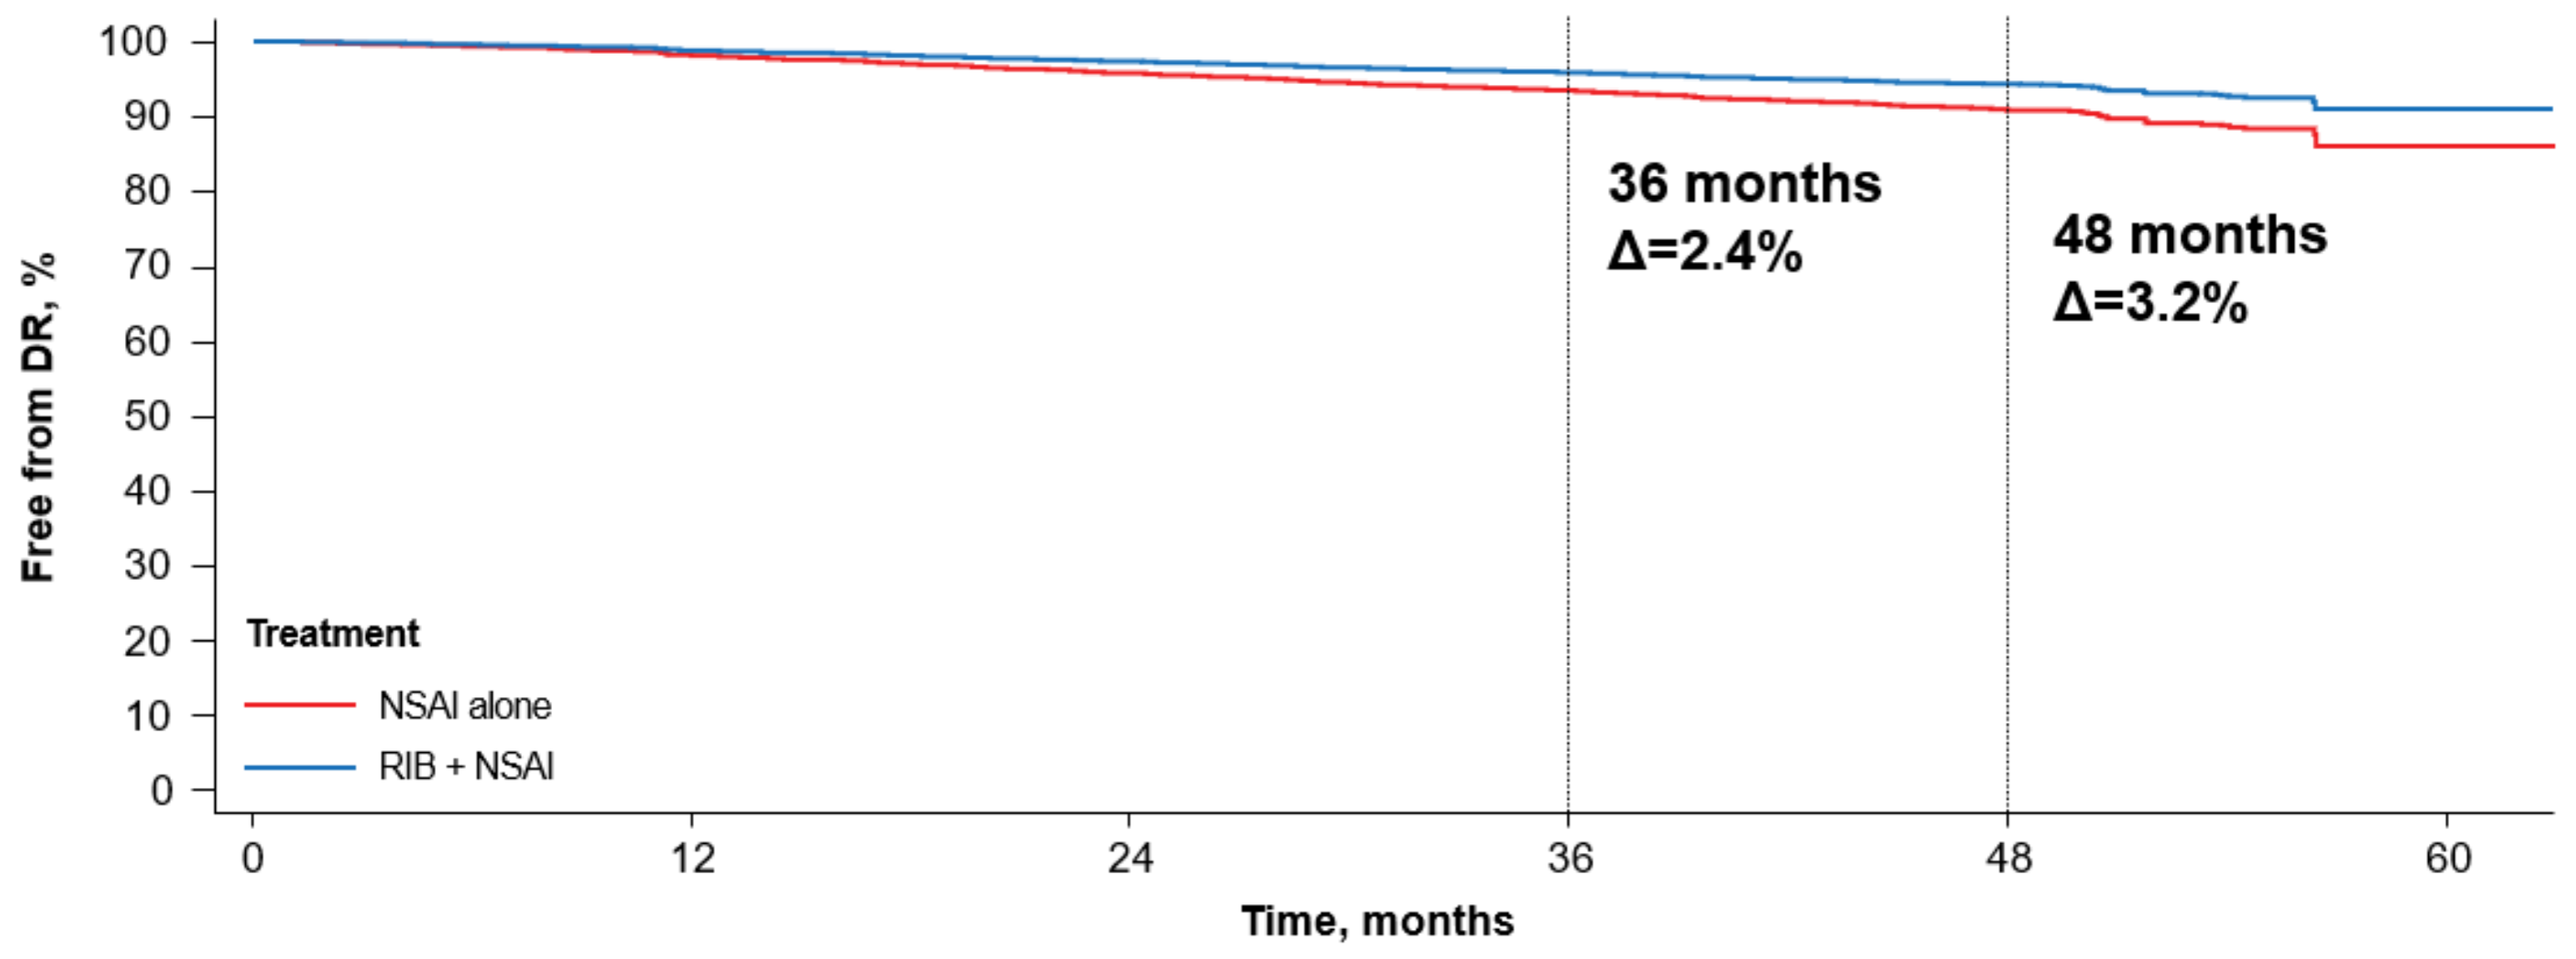
36 months
Δ=2.4%
48 months
Δ=3.2%
Free from DR, %
Time, months
Treatment
NSAI alone
RIB + NSAI

Weighted NATALEE data–trained Cox PH model using the overall NATALEE population to predict absolute DR risk in an RW population, assuming all patients were treated with RIB + NSAI or NSAI alone.
DR, distant recurrence; NSAI, nonsteroidal aromatase inhibitor; PH, proportional hazards; RIB, ribociclib; RW, real world.

Limitations

- Inherent RW data variability, as well as the use of a US-centric database, could lead to unexpected bias in model performance
- Potential differences in variables (definitions, missingness, etc) and in index date definition in RW and NATALEE populations should be noted
- These limitations could affect the accuracy of the NATALEE data–trained model when applied to the RW cohort, so the results should be interpreted with caution and further validated

References

1. Gnant M et al. *J Clin Oncol*. 2022;40(3):282-293.
2. Johnston SRD et al. *Lancet Oncol*. 2023;24(1):77-90.
3. Slamon D et al. *N Engl J Med*. 2024;390(12):1080-1091.
4. Fasching PA et al. *JAMA Oncol*. 2025;e253700.
5. Sparano JA et al. SGBCC 2025. Poster P258.
6. Database Characterization Guide: Flatiron Health. Accessed 1 October 2025. https://flatiron.com/database-characterization.

Acknowledgments

The authors thank the patients enrolled in these studies and their families as well as the study investigators.

Medical editorial assistance was provided by Nucleus Global and was funded by Novartis Pharmaceuticals Corporation. The authors had final responsibility for the poster.

Disclosures

F.M. Howard reports personal fees from Novartis, AG, and Leica Biosystems. **P.A. Fasching** reports personal fees from Novartis, Daiichi-Sankyo, AstraZeneca, Eisai, and Merck Sharp & Dohme; grants from Lilly, Seagen, Roche, Agendia, Gilead, Mylan, Menarini, Veracyte, and Guardant Health; grants from BioNTech and Cepheid; grants and personal fees from Pfizer; and other from Translational Research in Oncology (TRIO). **C.A. Santa-Maria**, **E. Lim**, and **O. Blyuss** report personal fees from Novartis. **J.A. Sparano** reports personal fees from AstraZeneca, Genentech, Lyell, Novartis, and Pfizer. **M.B. Lustberg** reports personal fees from Novartis, Gilead, AstraZeneca, Daiichi Lilly, and Menarini. **T. Bachelot** reports personal fees and nonfinancial support from Roche; grants, personal fees, and nonfinancial support from Novartis, AstraZeneca/Daiichi, and Pfizer; and grants and personal fees from Seagen. **C. Brezden-Masley** reports grants and other from Astellas, AstraZeneca, Gilead, Eli Lilly, Merck, Novartis, Pfizer, and Knight; other from Amgen, Beigene, and Daiichi Sankyo; nonfinancial support and other from BMS; and other from Sanofi. **Y.H. Park** reports grants and personal fees from MSD and AstraZeneca; grants, personal fees, and nonfinancial support from Pfizer and Roche; personal fees from Novartis; grants from Gencurix and Genome Insight; and personal fees from Daiichi-Sankyo, Gilead, and Lilly. **M. Akdere**, **F. Ye**, **K. Pantoja**, **C. Kurz**, and **P. Dominguez Castro** report employment and stock ownership from Novartis. **P. Razavi** reports grants from Grail/Illumina; grants and other from Novartis, AstraZeneca, Epic Sciences, Tempus, and Invitata; grants from Invitae/ArcherDX and Biotheranostics; and other from Foundation Medicine, Pfizer, Daiichi, Natera, Odyssey Biosciences, Paige.AI, and SAGA Diagnostics.