Poster #PS2150

Catherine Thieblemont | catherine.thieblemont@aphp.fr

Four-Year Update of Phase 2 ELARA Trial: Clinical Outcomes of Tisagenlecleucel in Patients With High-Risk Relapsed/ Refractory Follicular Lymphoma

Catherine Thieblemont, MD[1]; Martin Dreyling, MD[2]; Michael J. Dickinson, MBBS, D Med Sci, FRACP, FRCPA[3]; Joaquin Martinez-Lopez, MD, PhD[4]; Arne Kolstad, MD, PhD[5]; Jason Butler, MBBS[6]; Monalisa Ghosh, MD[7]; Leslie L. Popplewell, MD, FACP, MPH[8]; Julio C. Chavez, MD[9]; Emmanuel Bachy, MD, PhD[10]; Koji Kato, MD, PhD[11]; Hideo Harigae, MD, PhD[12]; Marie José Kersten, MD, PhD[13]; Charalambos Andreadis, MD, MS[14]; Peter A. Riedell, MD[15]; P. Joy Ho, MBBS, FRACP, FRCPA[16]; Jose Antonio Pérez Simon, MD, PhD[17]; Andy Chen, MD, PhD[18]; Loretta J. Nastoupil, MD[19]; Bastian von Tresckow, MD[20, 21]; Andrés José María Ferreri, MD[22]; Takanori Teshima, MD, PhD[23]; Piers E.M. Patten, MBChB, FRCP, FRCPath, PhD[24]; Joseph P. McGuirk, DO[25]; Fritz Offner, MD, PhD[26]; Andreas Viardot, MD, PhD[27]; Pier Luigi Zinzani, MD, PhD[28, 29]; Ram Malladi, MD, PhD[30]; Aiesha Zia, MSc[31]; Rakesh Awasthi, PhD[32]; Davide Germano, PhD[33]; Roberto Javier Ramos, MD[32]; Pei Hsu, MD[31]; Stephen J. Schuster, MD[34]; Nathan H. Fowler, MD[35]

[1]Service d'hémato-oncologie, Hôpital Saint-Louis, Paris, France; [2]Department of Medicine III, Ludwig-Maximilians-University Hospital, Munich, Germany; [3]Sir Peter MacCallum Department of Oncology, The University of Melbourne, Melbourne, VIC, Australia; [4]Hospital Universitario 12 de Octubre, CIBER-ONC CB16/12/00369, CNIO, Madrid, Spain; [5]Oslo University Hospital, Oslo, Norway; [6]Royal Brisbane Hospital, Herston, QLD, Australia; [7]Division of Hematology/Oncology, University of Michigan, Ann Arbor, MI; [8]Department of Hematology and Hematopoietic Cell Transplantation, City of Hope, Atlanta, GA; [9]Department of Malignant Hematology, Moffitt Cancer Center, Tampa, FL; [10]Clinical Hematology, Hospices Civils de Lyon and Claude Bernard Lyon 1 University, Lyon, France; [11]Department of Hematology, Oncology, and Cardiovascular Medicine, Kyushu University Hospital, Fukuoka, Japan; [12]Department of Hematology, Tohoku University Graduate School of Medicine, Sendai, Japan; [13]Cancer Center Amsterdam, Amsterdam University Medical Centers, University of Amsterdam, Amsterdam, Netherlands; [14]Helen Diller Family Comprehensive Cancer Center, University of California San Francisco, San Francisco, CA; [15]David and Etta Jonas Center for Cellular Therapy, University of Chicago, Chicago, IL; [16]Royal Prince Alfred Hospital & University of Sydney, Sydney, NSW, Australia; [17]Department of Hematology, University Hospital Virgen del Rocio-IBIS, Universidad de Sevilla, Sevilla, Spain; [18]Oregon Health and Science University, Portland, OR; [19]The University of Texas MD Anderson Cancer Center, Houston, TX; [20]Department I of Internal Medicine, Medical Faculty and University Hospital Cologne, University of Cologne, Cologne, Germany; [21]Department of Hematology and Stem Cell Transplantation, West German Cancer Center (WTZ), University Hospital Essen, University of Duisburg-Essen, Essen, Germany; [22]Unit of Lymphoid Malignancies, Department of Onco-Haematology, IRCCS San Raffaele Scientific Institute, Milano, Italy; [23]Department of Hematology, Hokkaido University, Sapporo, Japan; [24]King's College London, London, United Kingdom; [25]University of Kansas Medical Center, Westwood, KS; [26]Universitair Ziekenhuis Ghent, Ghent, Belgium; [27]Department of Internal Medicine III, University Hospital of Ulm, Ulm, Germany; [28]Universitaria di Bologna, Istituto Di Ematologia, Bologna, Italy; [29]Institute of Hematology "Seràgnoli," University of Bologna, Bologna, Italy; [30]Hospitals NHS Foundation Trust, Cambridge University, Cambridge, United Kingdom; [31]Novartis Pharma AG, Basel, Switzerland; [32]Novartis Pharmaceuticals, East Hanover, NJ; [33]Autolus Therapeutics, Basel, Switzerland; [34]Abramson Cancer Center, University of Pennsylvania, Philadelphia, PA; [35]BostonGene, Waltham, MA

KEY FINDINGS & CONCLUSIONS

- Updated long-term follow-up from the ELARA trial continues to demonstrate robust durable responses >4 years post infusion, alongside a favorable safety profile
- Subgroup analyses suggest that most baseline high-risk disease characteristics (double-refractory disease, bulky disease, POD24, and high FLIPI) are not associated with inferior CRR, 48-month PFS, or 48-month OS
 - Although lower CRR, 48-month PFS, and 48-month OS rates were reported for patients with high tumor burden, it is important to remember high-risk subgroup analyses were exploratory and some subgroups (ie, high tumor burden) had very limited patient numbers
- High frequencies of MRD-negative status were achieved among MRD-evaluable patients, showing that tisagenlecleucel therapy can achieve deep response in treated patients

Scan to obtain:
- Poster

https://tinyurl.com/ThieblemontPS2150

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study is sponsored by Novartis Pharma AG.
Poster presented at the 30th European Hematology Association (EHA) Annual Congress; June 12-15, 2025; Milan, Italy.

INTRODUCTION

- Treatment strategies for patients with relapsed/refractory follicular lymphoma (r/r FL) require consideration of prior therapies and patient-related factors[1]
 - Patients with r/r FL typically have worsening treatment outcomes with increasing lines of therapy[2]
 - Patients with high-risk disease characteristics (eg, progression of disease within 2 years of frontline systemic therapy [POD24]) have an increased chance of suboptimal response to later-line therapies such as R-CHOP (rituximab plus cyclophosphamide, doxorubicin, vincristine, prednisone)[3,4]
- Chimeric antigen receptor (CAR)-T cell therapy has demonstrated durable responses with acceptable safety profile in the treatment of patients with r/r FL, including those with high-risk disease characteristics[1,5-7]
- Tisagenlecleucel is an autologous cluster of differentiation (CD)-directed CAR-T cell therapy approved for the treatment of patients with r/r FL who have received ≥2 lines of prior therapy[8]
- Here we report the 4-year follow-up of efficacy, safety, and pharmacokinetics findings with a focus on patients with high-risk disease characteristics

METHODS

Study design

- ELARA study design, including eligibility criteria and key end points, can be found in **Figure 1**

Figure 1. ELARA Study Design

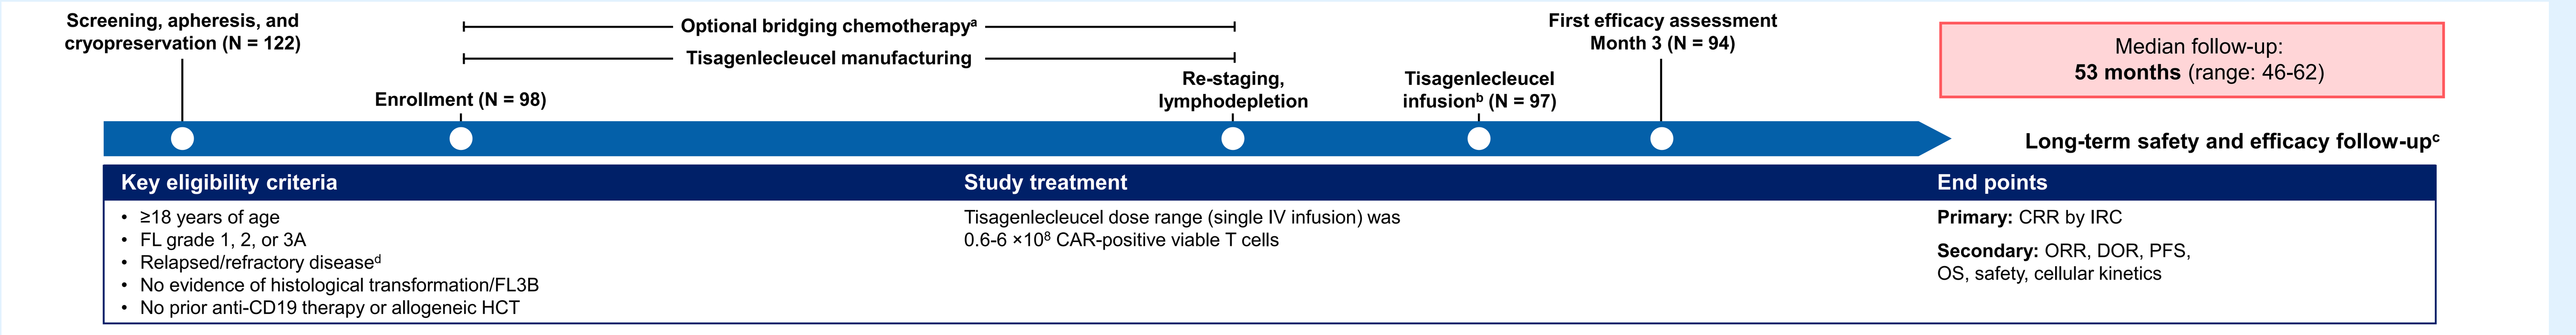
Screening, apheresis, and cryopreservation (N = 122)
Optional bridging chemotherapy[a]
Tisagenlecleucel manufacturing
Enrollment (N = 98)
Re-staging, lymphodepletion
Tisagenlecleucel infusion[b] (N = 97)
First efficacy assessment Month 3 (N = 94)
Median follow-up: 53 months (range: 46-62)
Long-term safety and efficacy follow-up[c]
Key eligibility criteria
≥18 years of age
FL grade 1, 2, or 3A
Relapsed/refractory disease[d]
No evidence of histological transformation/FL3B
No prior anti-CD19 therapy or allogeneic HCT
Study treatment
Tisagenlecleucel dose range (single IV infusion) was 0.6-6 ×10[8] CAR-positive viable T cells
End points
Primary: CRR by IRC
Secondary: ORR, DOR, PFS, OS, safety, cellular kinetics

CAR, chimeric antigen receptor; CD, cluster of differentiation; CRR, complete response rate; DOR, duration of response; FL, follicular lymphoma; FL3B, FL grade 3B; HCT, hematopoietic cell transplant; IRC, independent review committee; IV, intravenous; ORR, overall response rate; OS, overall survival; PFS, progression-free survival.
[a]Disease was reassessed prior to infusion for all patients requiring bridging therapy. [b]Infusion was conducted on an in- or outpatient basis at investigator discretion. [c]Every 3 months until month 12, and every 6 months until end of study. [d]Refractory to ≥2nd line of systemic therapy (including an anti-CD20 antibody and alkylating agent) or relapsed within 6 months after ≥2nd line of therapy or after an autologous HSCT.

- Bridging therapy was allowed and was followed by disease re-evaluation before tisagenlecleucel infusion
- Cellular kinetics were determined by measurement of transgene levels by quantitative polymerase chain reaction
- Minimal residual disease (MRD) levels were determined via clonoSEQ® next-generation sequencing assay performed at Adaptive Biotechnologies (Seattle, WA, USA)

RESULTS

Baseline Characteristics

- As of March 27, 2024, 97 patients were infused; 94 patients were evaluable for efficacy with a median follow-up of 53 months (range: 46-62)
 - The last patient in the ELARA trial was infused May 22, 2020
- Baseline and primary disease characteristics of the infused set can be found in **Table 1**
 - Median age was 57 years (range: 29-73), with 25% of infused patients ≥65 years of age
 - Median number of prior therapies was 4 (range: 2-13)
 - Thirty-six percent of patients received a prior autologous hematopoietic cell transplant (HCT)
 - Seventy-eight percent of patients were refractory to the last line of therapy
- Key patient subgroups at high risk among efficacy-evaluable patients (N = 94):
 - Disease refractory to ≥2 prior regimens: 72%
 - Bulky disease (>7 cm or at least 3 lesions >3 cm): 66%
 - POD24 from first anti-CD20 monoclonal antibody (mAb)-containing therapy: 65%
 - High Follicular Lymphoma International Prognostic Index (FLIPI; ≥3): 61%
 - High tumor burden (total metabolic tumor volume >510 mL[9]): 21%

Table 1. Disease History and Baseline Patient Characteristics

	Infused set (N = 97), n (%)
Median age (range), years	57.0 (29-73)
18 to <65 years	73 (75)
≥65 years	24 (25)
ECOG PS ≥1 prior to infusion	42 (43)
Stage at study entry III-IV	83 (86)
Bone marrow involvement	37 (38)
Bulky disease[a]	63 (65)
FLIPI high at study entry (≥3)	58 (60)
Median no. of prior therapies (range)	4 (2-13)
POD24[b]	61 (63)
Refractory disease to last line of therapy	76 (78)
Refractory to ≥2 regimens	69 (71)
Double refractory: anti-CD20 mAb + alkylating agent	66 (68)
Refractory to PI3K inhibitors	14 (14)
Prior autologous HCT	35 (36)

CD, cluster of differentiation; ECOG PS, Eastern Cooperative Oncology Group performance status; FLIPI, Follicular Lymphoma International Prognostic Index; HCT, hematopoietic cell transplant; mAb, monocolonal antibody; PI3K, phosphatidylinositol-3-kinase; POD24, progression of disease within 2 years of frontline systemic therapy.
[a]Bulky disease defined as 1 lesion >7 cm or at least 3 lesions >3 cm.
[b]POD24 from first anti-CD20 mAb-containing therapy or rituximab monotherapy.

Safety

- No new safety signals were reported since the last data cut. Adverse events (AEs) of special interest can be found summarized by timing post infusion in **Table 2**
 - Among the 47 patients who experienced cytokine release syndrome (CRS), all first events occurred within 8 weeks after infusion. Two patients experienced a second late onset CRS event; one in the context of a new anticancer therapy
 - Most cytopenias had their onset in the first-year post infusion
 - Rates of infections (including grade 3) increased over time

Table 2. AEs of Special Interest by Timing After Tisagenlecleucel Infusion

	Infused set (N = 97), n (%)					
	Within 8 weeks of infusion		>8 weeks to 1 year post infusion		>1 year post infusion	
Preferred term	All grades	Grade ≥3	All grades	Grade ≥3	All grades	Grade ≥3
Patients with at least one AE	88 (90.7)	68 (70.1)	70 (72.9)	42 (43.8)	38 (45.2)	24 (28.6)
Patients with at least one infection[a]	20 (20.6)	6 (6.2)	37 (38.5)	12 (12.5)	31 (36.9)	16 (19.0)
Cytokine release syndrome	47 (48.5)	0	1 (1.0)	0	1 (1.2)	1 (1.2)
Cytopenias						
Neutropenia	33 (34.0)	32 (33.0)	21 (21.9)	18 (18.8)	5 (6.0)	5 (6.0)
Neutrophil count decreased	15 (15.5)	15 (15.5)	7 (7.3)	7 (7.3)	2 (2.4)	1 (1.2)
Anemia	23 (23.7)	13 (13.4)	7 (7.3)	5 (5.2)	4 (4.8)	3 (3.6)
Thrombocytopenia	15 (15.5)	9 (9.3)	8 (8.3)	5 (5.2)	3 (3.6)	2 (2.4)
Platelet count decreased	7 (7.2)	4 (4.1)	4 (4.2)	2 (2.1)	0	0
White blood cell count decreased	18 (18.6)	13 (13.4)	12 (12.5)	7 (7.3)	2 (2.4)	0
Leukopenia	7 (7.2)	7 (7.2)	3 (3.1)	3 (3.1)	1 (1.2)	0
Febrile neutropenia	10 (10.3)	10 (10.3)	2 (2.1)	2 (2.1)	2 (2.4)	2 (2.4)
Lymphocyte count decreased	10 (10.3)	8 (8.2)	2 (2.1)	0	0	0
Lymphopenia	6 (6.2)	6 (6.2)	4 (4.2)	4 (4.2)	1 (1.2)	1 (1.2)
Hypogammaglobulinemia	9 (9.3)	0	7 (7.3)	1 (1.0)	2 (2.4)	0
Patients with at least one serious neurological AE	10 (10.3)	1 (1.0)	2 (2.1)	2 (2.1)	2 (2.4)	1 (1.2)

AE, adverse event.
[a]Grade ≥3 infections reported within 8 weeks of infusion included bacteremia, *Campylobacter* infection, *Escherichia* sepsis, human herpesvirus 6 encephalitis, neutropenic sepsis, pseudomonal sepsis, and staphylococcal sepsis. Grade ≥3 infections reported >8 weeks to 1 year post infusion included pneumonia, COVID-19, lower respiratory tract infection, tooth infection, coronavirus pneumonia, diverticulitis, pneumonia influenza, progressive multifocal leukoencephalopathy, pseudomonal bacteremia, and sepsis. Grade ≥3 infections reported >1 year post infusion included COVID-19, pneumonia, sinusitis, COVID-19 pneumonia, encephalitis, infection, pneumonia respiratory syncytial viral, rhinovirus infection, sepsis, skin infection, and varicella zoster virus infection.

- Second primary malignancies (defined as any new cancer occurring post infusion regardless of tisagenlecleucel relationship) were reported in 6 patients (6.2%) and included basal cell carcinoma (N = 2), squamous cell carcinoma (N = 2), acute myeloid leukemia (N = 1), bladder transitional cell carcinoma (N = 1), Bowen's disease (N = 1), malignant melanoma (N = 1), metastatic squamous cell carcinoma (N = 1), and myelodysplastic syndrome (N = 1)
- As of data cutoff, 19 patients have died during the study: 8 due to progressive disease, 10 due to AEs (1 patient each; acute myeloid leukemia, bladder transitional cell carcinoma, cardiac arrest, CRS, encephalitis, gastrointestinal hemorrhage, infection, metastatic squamous cell carcinoma, pneumonia, and sepsis), and 1 from euthanasia

Efficacy

- Subgroup analyses suggest that most baseline high-risk disease characteristics (double-refractory disease, bulky disease, POD24, and high FLIPI) were not associated with inferior overall or complete response following tisagenlecleucel infusion in patients with r/r FL (**Figure 2**)

Figure 2. Complete Response Rate by Subgroups

		CRR, n/N (%)	[95% CI]
All patients	All patients (N = 94)	65/94 (69.1)	[58.8-78.3]
Age	<65 years (N = 70)	46/70 (65.7)	[53.4-76.7]
	≥65 years (N = 24)	19/24 (79.2)	[57.8-92.9]
Sex	Female (N = 30)	22/30 (73.3)	[54.1-87.7]
	Male (N = 64)	43/64 (67.2)	[54.3-78.4]
Race	Asian (N = 11)	9/11 (81.8)	[8.2-97.7]
	White (N = 79)	54/79 (68.4)	[56.9-78.4]
Ethnicity	Not Hispanic or Latino (N = 87)	60/87 (69.0)	[58.1-78.5]
FLIPI at study entry	Low/Intermediate (N = 37)	29/37 (78.4)	[61.8-90.2]
	High (N = 57)	36/57 (63.2)	[49.3-75.6]
Histological grade	1-2 (N = 85)	59/85 (69.4)	[58.5-79.0]
	3A (N = 9)	6/9 (66.7)	[29.9-92.5]
Number of prior lines	≤2 lines (N = 24)	14/24 (58.3)	[36.6-77.9]
	3-4 lines (N = 43)	35/43 (81.4)	[66.6-91.6]
	>4 lines (N = 27)	16/27 (59.3)	[38.8-77.6]
PI3K inhibitor use	Pre-treated (N = 19)	15/19 (78.9)	[54.4-93.9]
	Naive (N = 75)	50/75 (66.7)	[54.8-77.1]
Prior HSCT therapy	Yes (N = 35)	22/35 (62.9)	[44.9-78.5]
	Relapsed ≤12 months (N = 14)	10/14 (71.4)	[41.9-91.6]
	Relapsed >12 months (N = 21)	12/21 (57.1)	[34.0-78.2]
	No (N = 59)	43/59 (72.9)	[59.7-83.6]
Disease status to last line	Refractory (N = 74)	52/74 (70.3)	[58.5-80.3]
	Relapsed (N = 17)	11/17 (64.7)	[38.3-85.8]
POD24	Yes (N = 61)	37/61 (60.7)	[47.3-72.9]
	No (N = 33)	28/33 (84.8)	[68.1-94.9]
Bulky disease at baseline	Yes (N = 62)	41/62 (66.1)	[53.0-77.7]
	No (N = 32)	24/32 (75.0)	[56.6-88.5]
Bridging therapy	Yes (N = 44)	29/44 (65.9)	[50.1-79.5]
	No (N = 50)	36/50 (72.0)	[57.5-83.8]
LDH at study entry	≤ULN (N = 53)	39/53 (73.6)	[59.7-84.7]
	>ULN (N = 41)	26/41 (63.4)	[46.9-77.9]
Prior R2 use	Pre-treated (N = 16)	12/16 (75.0)	[47.6-92.7]
	Naive (N = 78)	53/78 (67.9)	[56.4-78.1]
US sites	Yes (N = 26)	17/26 (65.4)	[44.3-82.8]
	No (N = 68)	48/68 (70.6)	[58.3-81.0]
Double refractory	Yes (N = 65)	44/65 (67.7)	[54.9-78.8]
	No (N = 29)	21/29 (72.4)	[52.8-87.3]
MTV at baseline	High tumor burden (N = 20)	8/20 (40.0)	[19.1-63.9]
	Low tumor burden (N = 72)	55/72 (76.4)	[64.9-85.6]

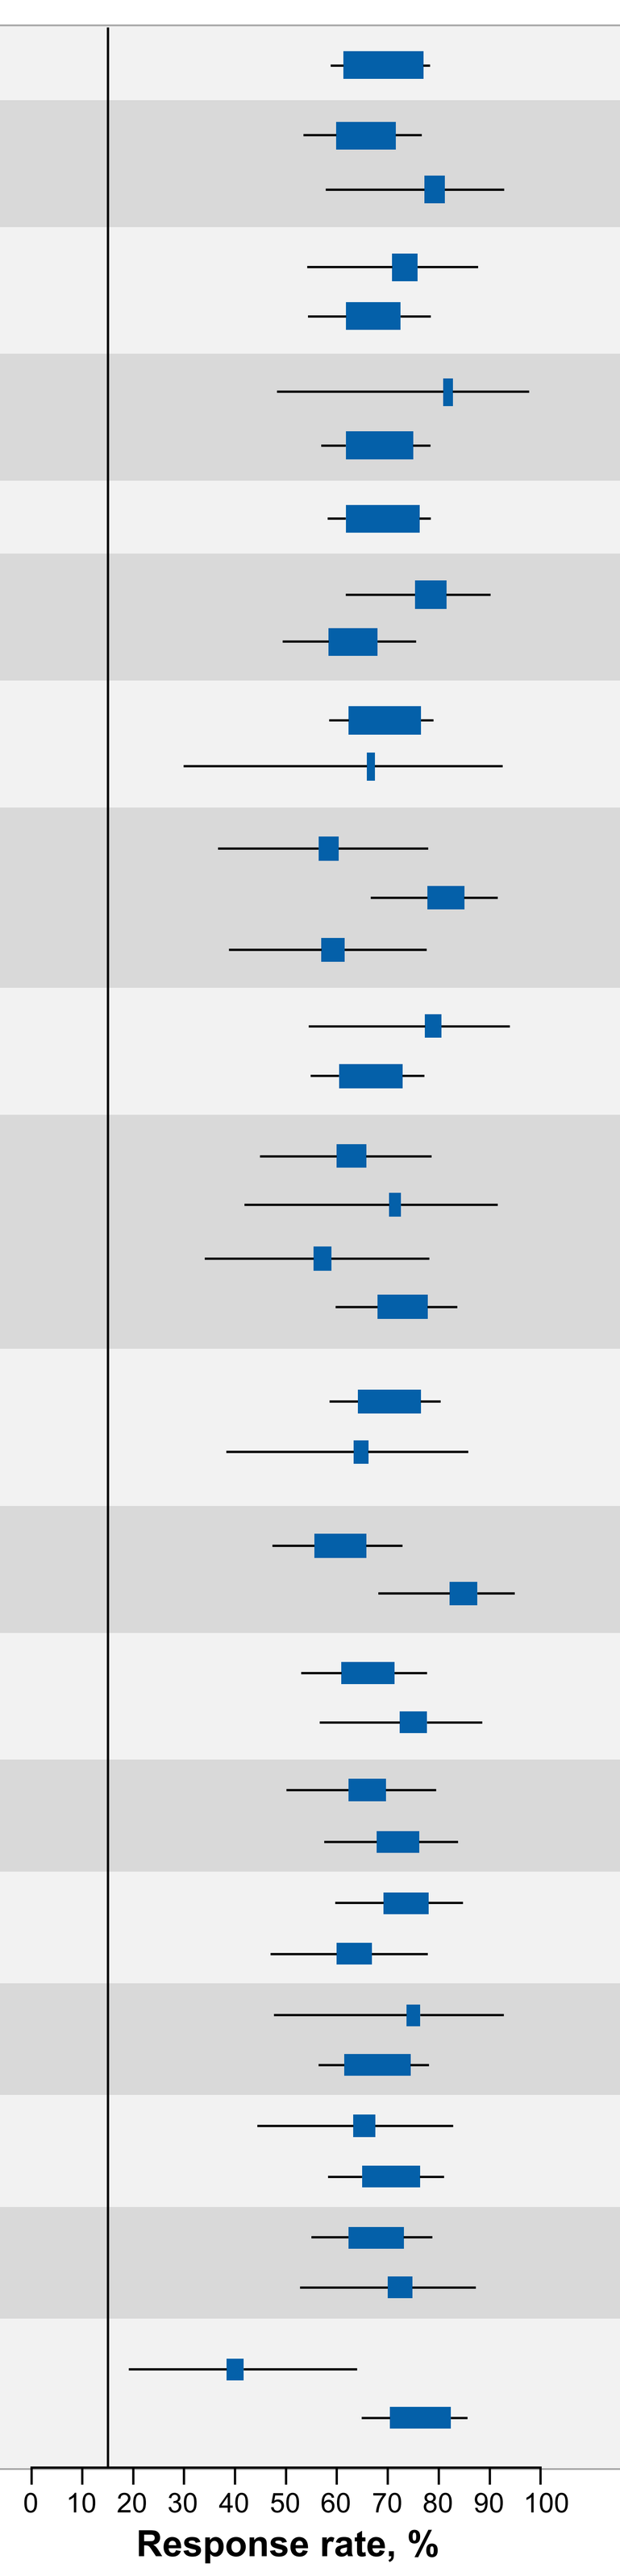
0
10
20
30
40
50
60
70
80
90
100
Response rate, %

CRR, complete response rate; FLIPI, Follicular Lymphoma International Prognostic Index; HSCT, hematopoietic stem cell transplant; LDH, lactate dehydrogenase; MTV, metabolic tumor volume; PI3K, phosphatidylinositol-3-kinase; POD24, progression of disease within 2 years of frontline systemic therapy; R2, lenalidomide + rituximab; ULN, upper limit of normal.

- The estimated 48-month progression-free survival (PFS) was 50.2% as assessed by independent review committee (IRC) (**Figure 3**)
 - Patients who had a best overall response of complete response had a 48-month probability of PFS of 66.1%
 - Among patient subgroups at high risk, 48-month PFS by IRC was 45.5% (POD24), 45.5% (high FLIPI), 45.2% (bulky disease), 52.8% (double-refractory), and 23.2% (high tumor burden)

Figure 3. Progression-Free Survival

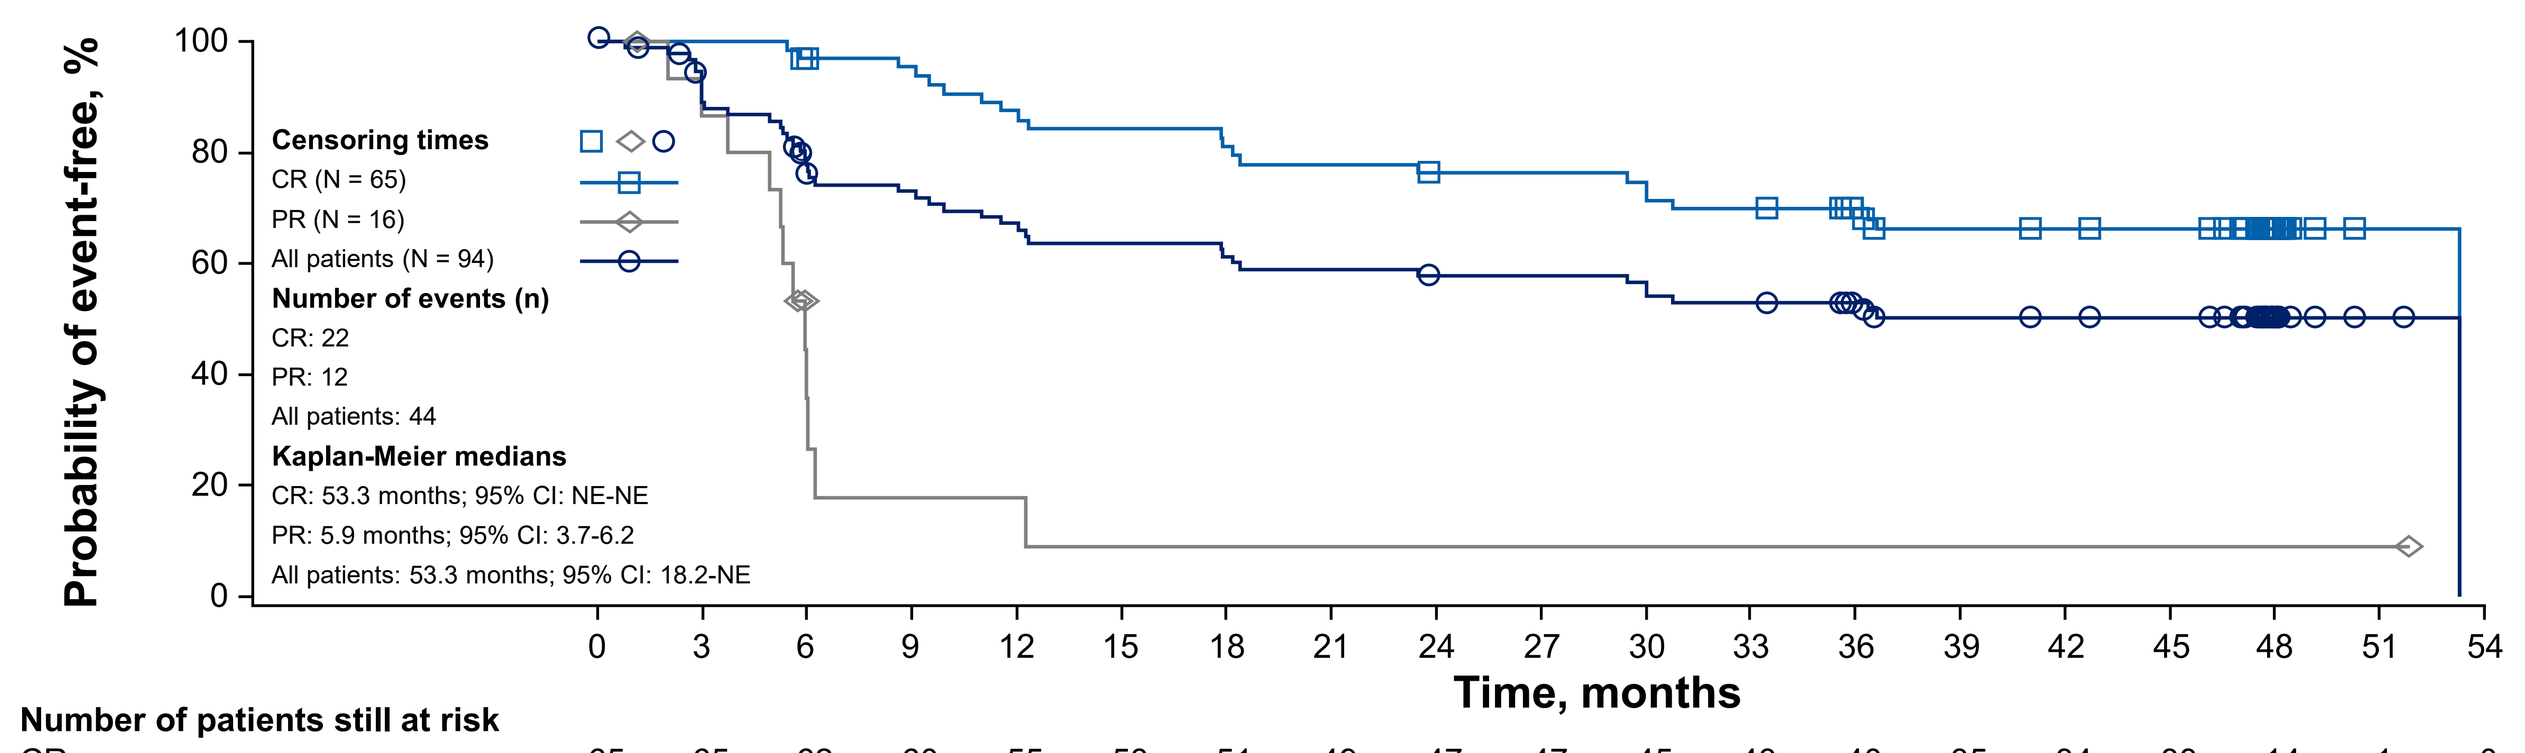
Probability of event-free, %
Time, months
Censoring times
CR (N = 65)
PR (N = 16)
All patients (N = 94)
Number of events (n)
CR: 22
PR: 12
All patients: 44
Kaplan-Meier medians
CR: 53.3 months; 95% CI: NE-NE
PR: 5.9 months; 95% CI: 3.7-6.2
All patients: 53.3 months; 95% CI: 18.2-NE

Number of patients still at risk	0	3	6	9	12	15	18	21	24	27	30	33	36	39	42	45	48	51	54
CR	65	65	62	60	55	53	51	49	47	47	45	43	40	35	34	33	14	1	0
PR	16	13	4	2	2	1	1	1	1	1	1	1	1	1	1	1	1	1	0
All patients	94	80	67	62	57	54	52	50	48	48	46	44	41	36	35	34	15	2	0

CR, complete response; NE, not estimable; PR, partial response.

- Median overall survival (OS) was not reached as assessed by IRC (**Figure 4**); 48-month OS was 79.3% in efficacy-evaluable patients
 - Among patient subgroups at high risk, 48-month OS by IRC was 80.8% (POD24), 73.2% (high FLIPI), 73.0% (bulky disease), 83.7% (double-refractory), and 65.5% (high tumor burden)

Figure 4. Overall Survival

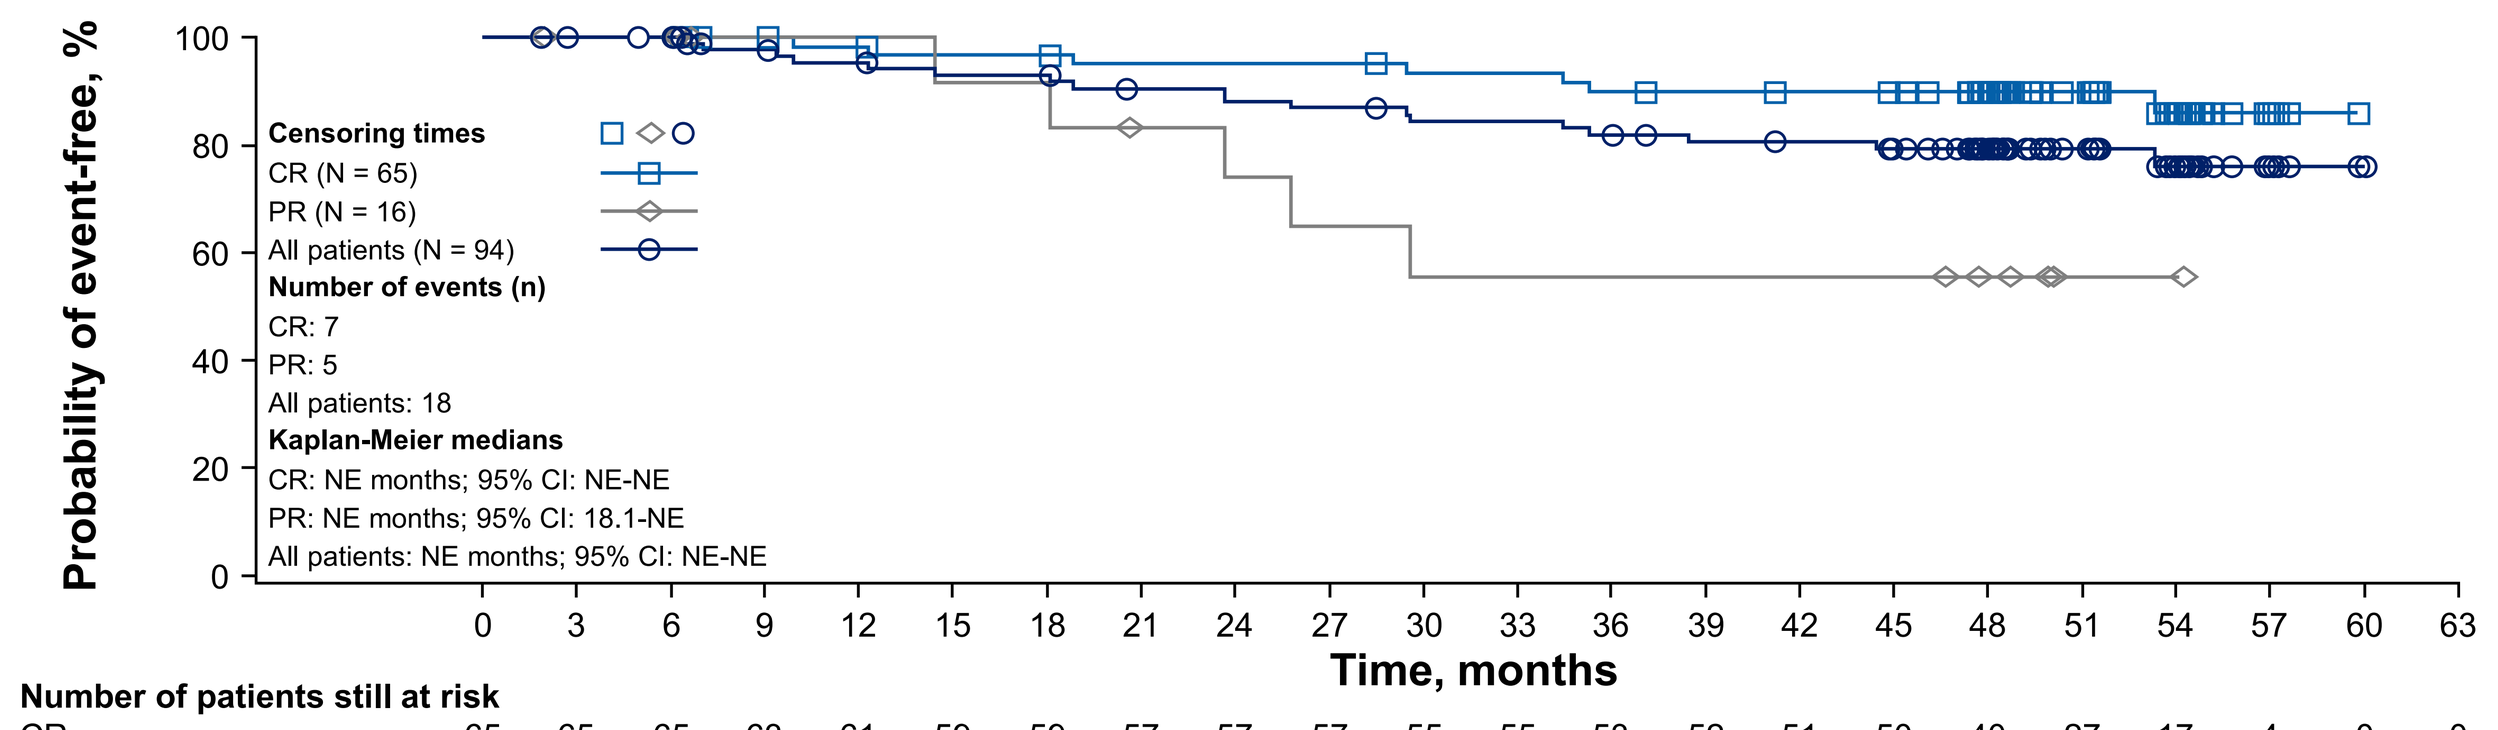
Probability of event-free, %
Time, months
Censoring times
CR (N = 65)
PR (N = 16)
All patients (N = 94)
Number of events (n)
CR: 7
PR: 5
All patients: 18
Kaplan-Meier medians
CR: NE months; 95% CI: NE-NE
PR: NE months; 95% CI: 18.1-NE
All patients: NE months; 95% CI: NE-NE

Number of patients still at risk	0	3	6	9	12	15	18	21	24	27	30	33	36	39	42	45	48	51	54	57	60	63
CR	65	65	65	63	61	59	59	57	57	57	55	55	53	52	51	50	40	27	17	4	0	0
PR	16	15	15	12	12	11	11	9	8	7	6	6	6	6	6	6	4	1	1	0	0	0
All patients	94	92	91	84	81	78	78	74	72	71	68	68	65	63	62	59	45	29	19	5	1	0

CR, complete response; NE, not estimable; PR, partial response.

- Median time to next treatment among all patients and patients with a best overall response of complete response was not reached (**Figure 5**)

Figure 5. Time to Next Treatment

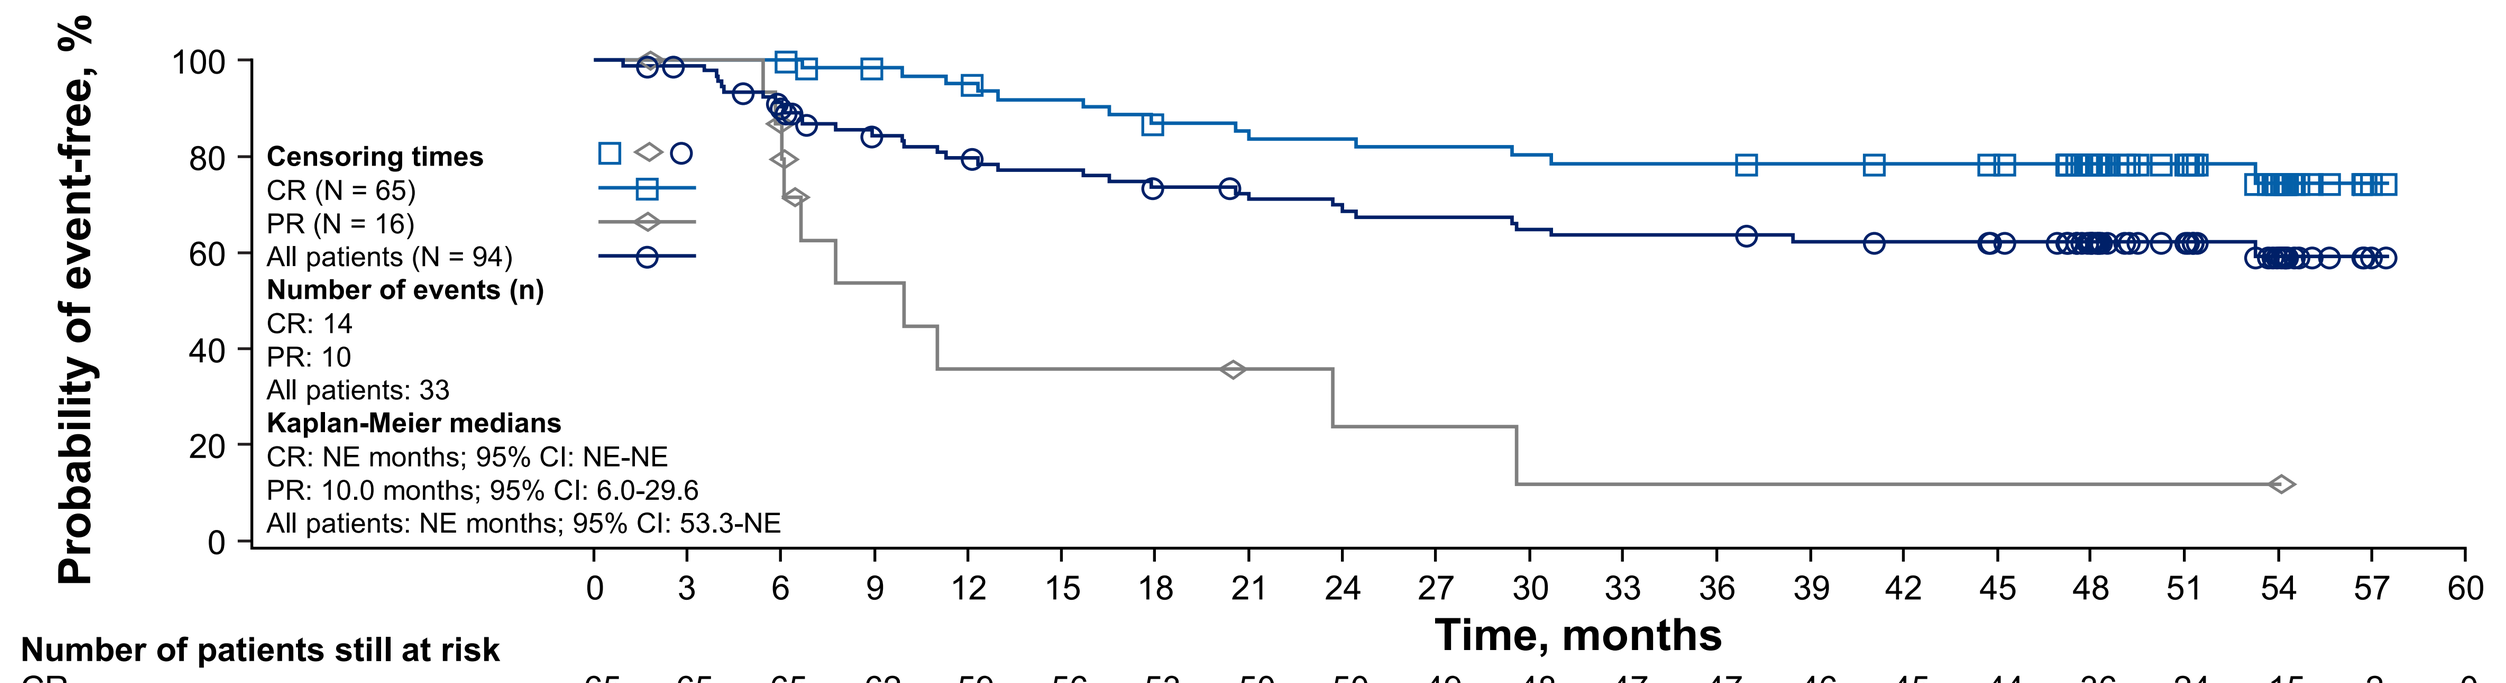
Probability of event-free, %
Time, months
Censoring times
CR (N = 65)
PR (N = 16)
All patients (N = 94)
Number of events (n)
CR: 14
PR: 10
All patients: 33
Kaplan-Meier medians
CR: NE months; 95% CI: NE-NE
PR: 10.0 months; 95% CI: 6.0-29.6
All patients: NE months; 95% CI: 53.3-NE

Number of patients still at risk	0	3	6	9	12	15	18	21	24	27	30	33	36	39	42	45	48	51	54	57	60
CR	65	65	65	62	59	56	53	50	50	49	48	47	47	46	45	44	36	24	15	2	0
PR	16	15	13	6	4	4	4	3	2	2	1	1	1	1	1	1	1	1	1	0	0
All patients	94	91	83	72	67	64	61	57	56	54	52	51	51	49	48	46	37	25	16	2	0

CR, complete response; NE, not estimable; PR, partial response.

- MRD data were available for 31 of 94 patients (33.0%)
 - 90.3% of evaluable patients (28/31) achieved MRD negativity at any time point (**Table 3**)
 - 63.6% of patients with MRD-negative status at month 6 (7/11) are ongoing without relapse
 - All 5 patients with MRD-positive status at month 6 relapsed

Table 3. MRD-Negative Rate by Timing After Tisagenlecleucel Infusion

	MRD-negative (N = 31[a]), % (n/N)
Day 28	82.0 (22/27)
Month 3	75.0 (12/16)
Month 6	69.0 (11/16)
Month 12	76.0 (13/17)
Any time	90.0 (28/31)

MRD, minimal residual disease.
[a]Patients evaluated for MRD response.

- CAR transgene persistence was observed for up to 1680 days; median Tlast (time to last measurable concentration) was 210 days (range: 13-1680)

References

1. Fowler NH et al. Target Oncol. 2024;19(4):495-510. **2.** Link BK et al. Br J Haematol. 2019;184(4):660-663. **3.** Casulo C et al. J Clin Oncol. 2015;33(23):2516-2522. **4.** Sortais C et al. Ann Hematol. 2020;99(7):1595-1604. **5.** Jacobson CA et al. Lancet Oncol. 2022;23(1):91-103. **6.** Fowler NH et al. Nat Med. 2022;28(2):325-332. **7.** Dreyling M et al. Blood. 2024;143(17):1713-1725. **8.** Kymriah. Prescribing information. Novartis Pharmaceuticals Corp. **9.** Delfau-Larue MH et al. Blood Adv. 2018;2(7):807-816.

Acknowledgements

The authors sincerely thank the patients, their families, and the principal investigators and support staff.
All analyses in this presentation were conducted by Novartis Pharmaceuticals Corporation.
Medical writing support was provided by Jay Shah, PhD, of Novartis Healthcare Pvt Ltd and was funded by Novartis Pharmaceuticals Corporation.
Previously presented at the 2024 ASH Annual Meeting and Exposition; December 9-13, 2024; San Diego, CA, and Virtual. Poster: 3034.

Disclosures

Catherine Thieblemont: *Current Employment and Ended employment in the past 24 months:* University of Paris; *Consultancy and Honoraria:* Kite/Gilead, Takeda, BMS/Celgene, AbbVie, BeiGene, Janssen, Novartis, Regeneron; *Honoraria:* Bayer, Roche, Cellectis, ADC Therapeutics, AstraZeneca, Incyte, Sanofi, Amgen; *Research Funding:* Kite/Gilead, AbbVie, Roche.