Poster# PS2257

Karina Yazdanbakhsh | kyazdanbakhsh@nybc.org

Efficacy of Ianalumab in a Mouse Model of Autoimmune Hemolytic Anemia (AIHA)

Karina Yazdanbakhsh[1], Weili Bao[1], Shan Su[1], Yunfeng Liu[1], Marc Bigaud[2], Elissa Furutani[3], Tatjana Uffelmann[2], Pascal Edrich[2], Barbara Nüsslein-Hildesheim[2], Isabelle Isnardi[2], Christoph Burkhart[2]

[1]New York Blood Center, New York, NJ, USA;
[2]Novartis Pharma AG, Basel, Switzerland;
[3]Novartis Biomedical Research, Cambridge, MA, USA.

KEY FINDINGS & CONCLUSIONS

- In the Playfair-Marshall Clarke mouse model of AIHA, the anti-BAFF-R monoclonal antibody ianalumab (100 mg/kg; i.p.; weekly) led to B-cell depletion in both blood and spleen as well as a reduction in the levels of circulating anti-RBC autoantibodies which both correlated with the ianalumab plasma concentration.
- In addition, ianalumab reduced the frequency of splenic marginal zone B cells and corrected the imbalance between splenic Teff and protective Foxp3+ Treg.
- These data suggest that ianalumab may be effective in reducing anti-RBC autoantibodies in wAIHA. The observed restoration of Teff/Treg balance by ianalumab also highlights its immunomodulatory potential in inducing durable responses.
- Ongoing phase 3 studies in wAIHA (NCT05648968, in first-line (1L) and second-line (2L) primary ITP (NCT05653349 and NCT05653219, respectively), and in additional autoimmune indications will provide further evidence on the efficacy and safety of ianalumab in larger patient populations.

Scan to obtain:
- Poster

https://tinyurl.com/YazdanbakhshPS2257

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study is sponsored by Novartis Pharma AG.
Poster presented at EHA 2025, held on 12-15 June, 2025

INTRODUCTION

- Warm autoimmune hemolytic anemia (wAIHA) is caused by premature destruction of the patient's own red blood cells (RBC) by autoantibodies reactive at 37°C and can be life-threatening.[1]
- To date, approved therapies for the treatment of wAIHA are very limited.
- Ianalumab, a glycoengineered (afucosylated), fully human IgG1 monoclonal antibody directed against the B-cell–activating factor receptor (BAFF-R), depletes B cells through enhanced antibody-dependent cellular cytotoxicity (ADCC) with concurrent blockade of BAFF:BAFF-R–mediated signals (**Figure 1**).[2]

Figure 1. Ianalumab's dual mechanism of action

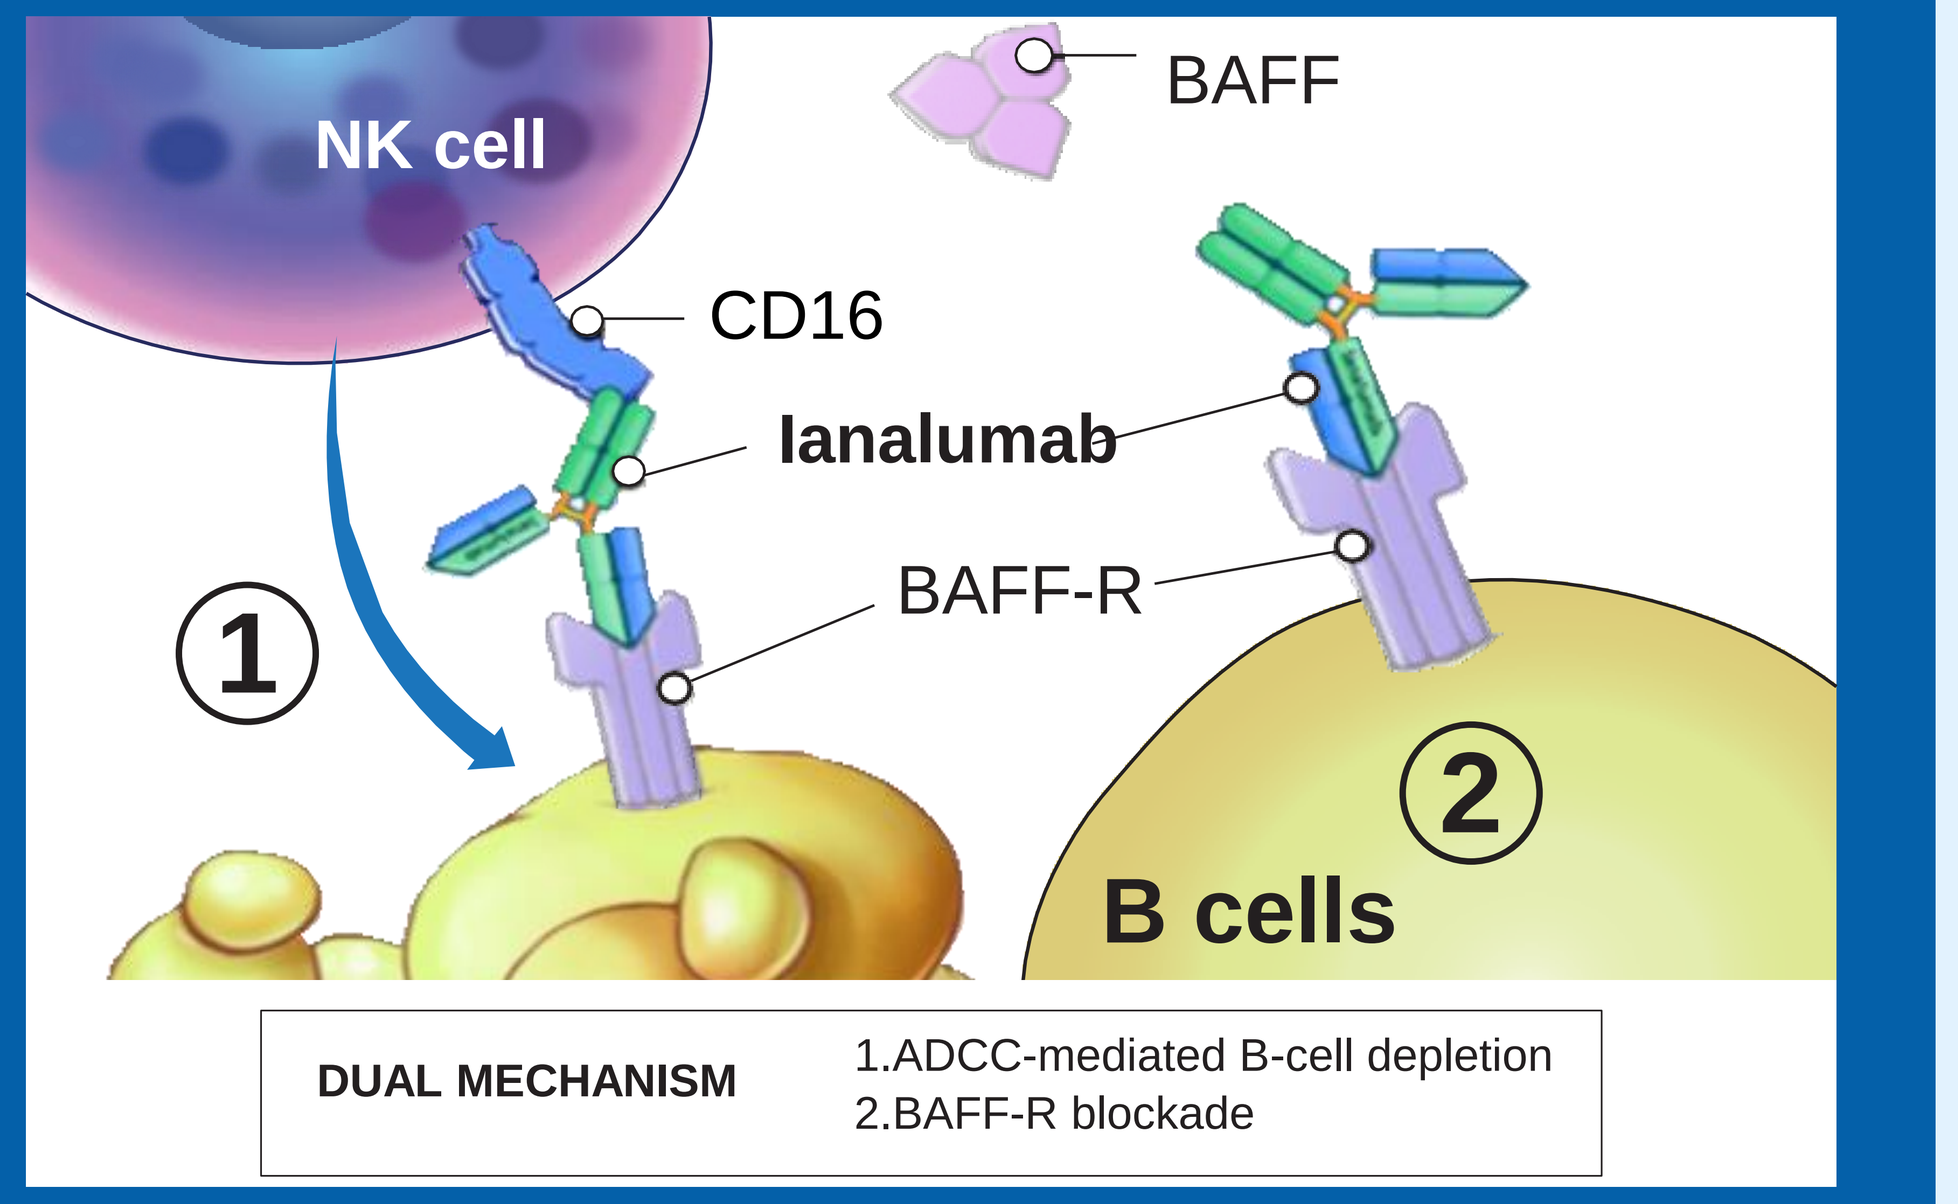
NK cell
BAFF
CD16
Ianalumab
BAFF-R
1
2
B cells
DUAL MECHANISM
1.ADCC-mediated B-cell depletion
2.BAFF-R blockade

ADCC, antibody-dependent cellular cytotoxicity; BAFF, B-cell–activating factor; BAFF-R, BAFF receptor; CD16; cluster of differentiation 16; NK cell, natural killer cell.

METHODS

- To determine its efficacy in the inhibition of autoantibodies in wAIHA, ianalumab was studied in a Playfair-Marshall Clarke autoimmune hemolytic anemia (AIHA) mouse model which involves a repeated immunization protocol against rat RBC, resulting in the induction of anti-mouse RBC autoantibodies and development of anemia.[3]
- In this model, a 11-week-long immunization protocol (2 × 10^8 cells leuko-reduced rat RBC injected per mouse on a weekly basis) was used to induce AIHA.
- C57BL/6 (B6) mice were dosed with ianalumab (100 mg/kg; intraperitoneally (i.p.); weekly) from either 5 days prior to (n=15) or 3 days post (n=15) first rat RBC immunization. Control mouse groups included rat RBC-immunized B6 mice dosed with phosphate-buffered saline (PBS) (n=24 mice), non-diseased B6 mice dosed with ianalumab (n=6 mice) or PBS (n=6 mice), and unmanipulated B6 mice (n=6) (**Figure 2**).

Figure 2. Mouse AIHA model study design

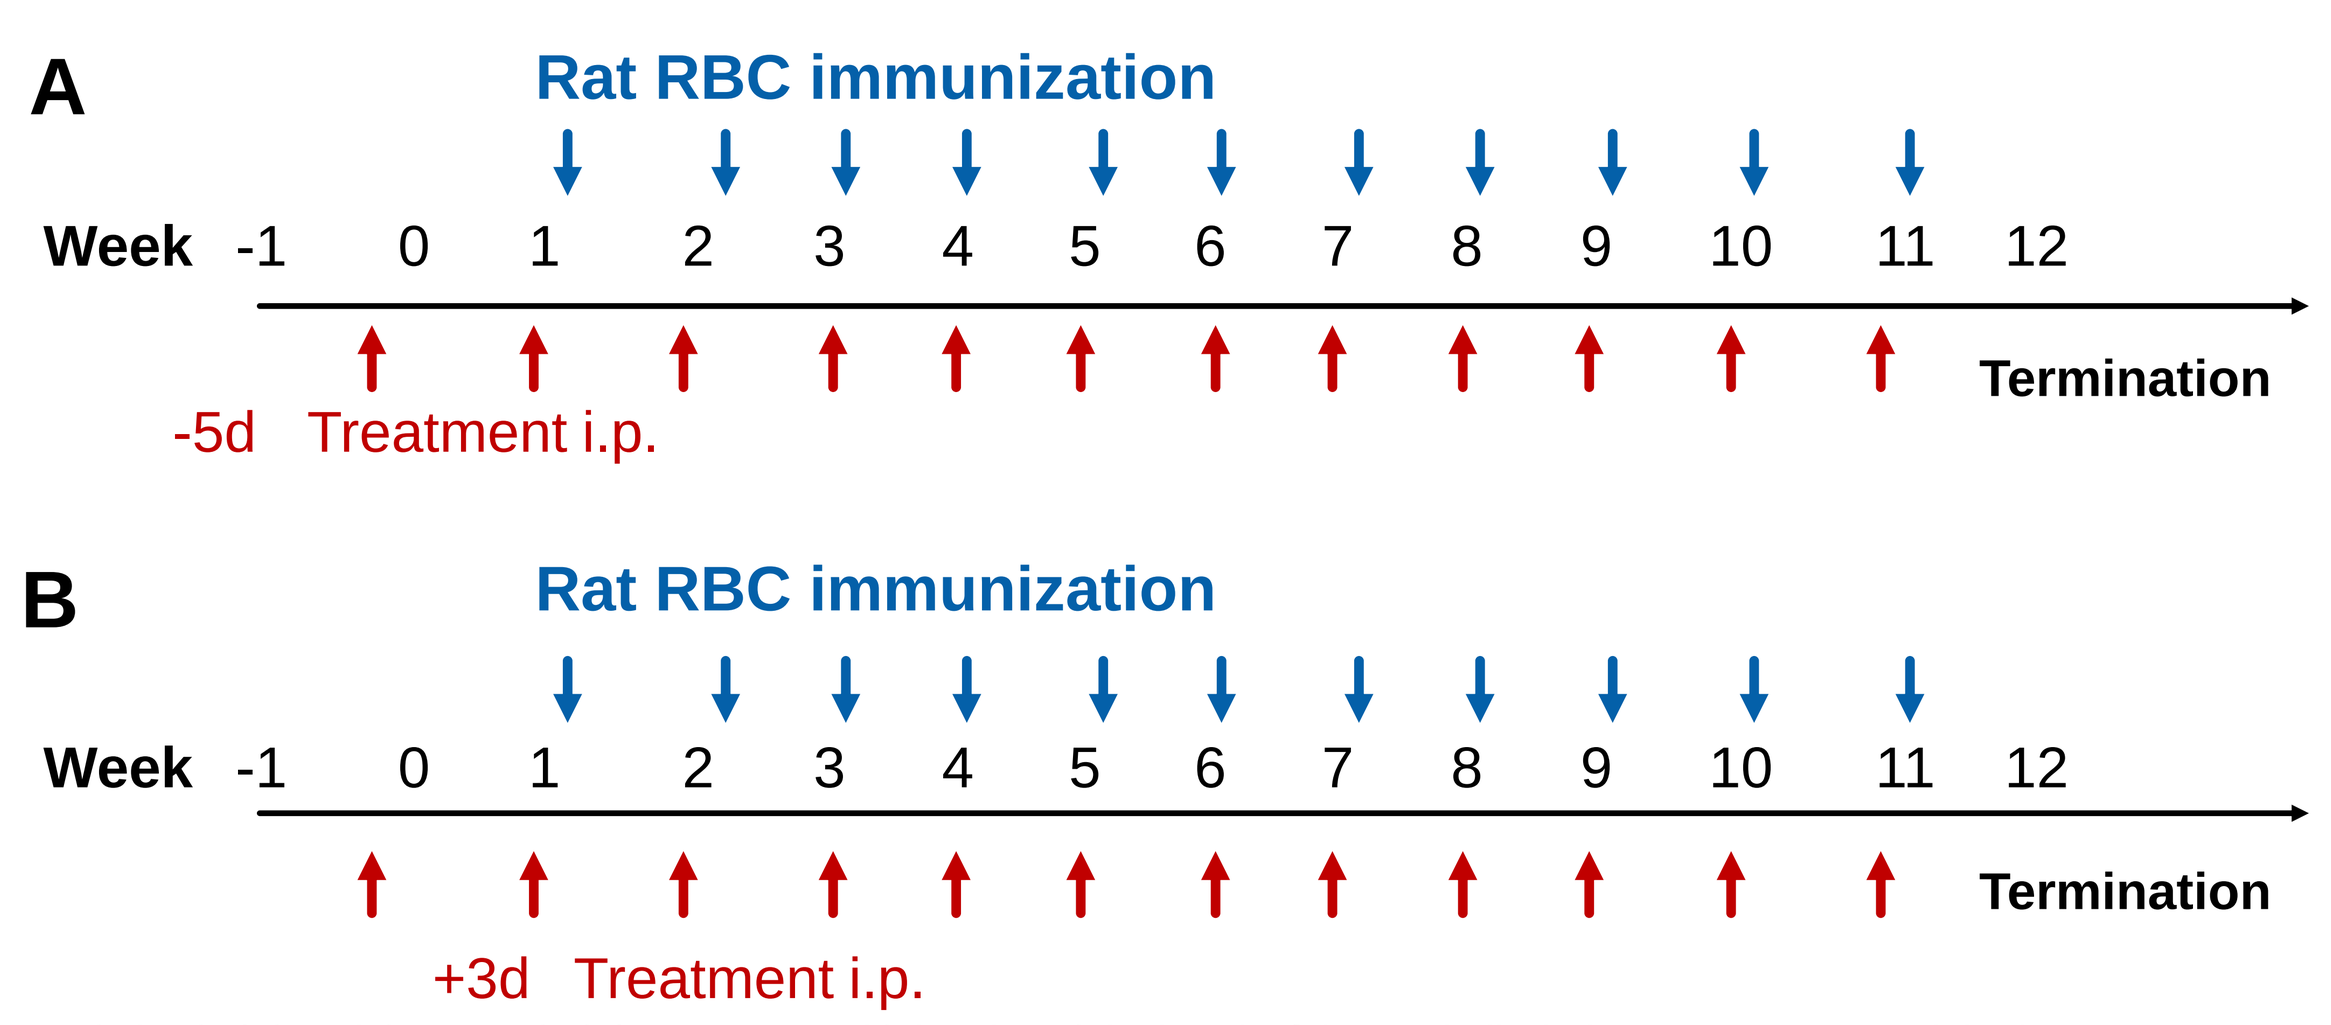
A
Rat RBC immunization
Week -1 0 1 2 3 4 5 6 7 8 9 10 11 12
Termination
-5d Treatment i.p.
B
Rat RBC immunization
Week -1 0 1 2 3 4 5 6 7 8 9 10 11 12
Termination
+3d Treatment i.p.

Prophylactic (A) and therapeutic (B) treatments with ianalumab in the Playfair-Marshall Clarke AIHA mouse model. Blue arrows indicate the time points of the injection of leuko-reduced rat RBC; red arrows indicate the time points of ianalumab injection. AIHA, autoimmune hemolytic anemia; i.p., intraperitoneally; RBC, red blood cell.

RESULTS

Ianalumab significantly depletes B cells

- Results from two separate studies were pooled and analyzed. Mean circulating B-cell counts in the blood were significantly reduced within the first 3 weeks of weekly dosing with ianalumab (p<0.0001; **Figure 3**), while no effect was observed on non-B lineage cell populations.

B-cell depletion correlates with ianalumab exposure

- The degree of B-cell depletion in both blood and spleen of mice correlated with ianalumab plasma concentration (p<0.005) as well as a reduction in the levels of circulating anti-RBC autoantibodies (p=0.005, **Figure 4**).

B-cell depletion correlates with the presence of anti-drug antibodies

- Moreover, low ianalumab plasma concentration was associated with the presence of anti-drug antibodies (ADA), consistent with the immune response in mice exposed to human antibodies **(Figure 5**).

Ianalumab reduces splenic marginal zone B cells

- Within the splenic B compartment, the frequency of marginal zone (CD21/35+IgM+) B-cell subset, implicated in autoreactive B-cell responses, was significantly reduced in ianalumab-treated rat RBC-immunized mice relative to PBS-treated rat RBC-immunized mice (p<0.0001) (**Figure 6**).

Ianalumab corrects the imbalance between effector and regulatory T cells

- Additionally, in the spleen, ianalumab treatment corrected the imbalance between CD4+CD44^{high} effector T cells (Teff) and protective CD4+Foxp3+ regulatory T cells (Treg) (p<0.0001) in rat RBC-immunized mice, reversing the Teff/Treg ratio skewed toward Teff (which favors autoimmunity as seen in PBS-treated rat RBC-immunized mice) to levels comparable to those in unmanipulated mice or non-disease mice treated with ianalumab or PBS (**Figure 7**).

Figure 3. B-cell depletion induced by ianalumab in blood

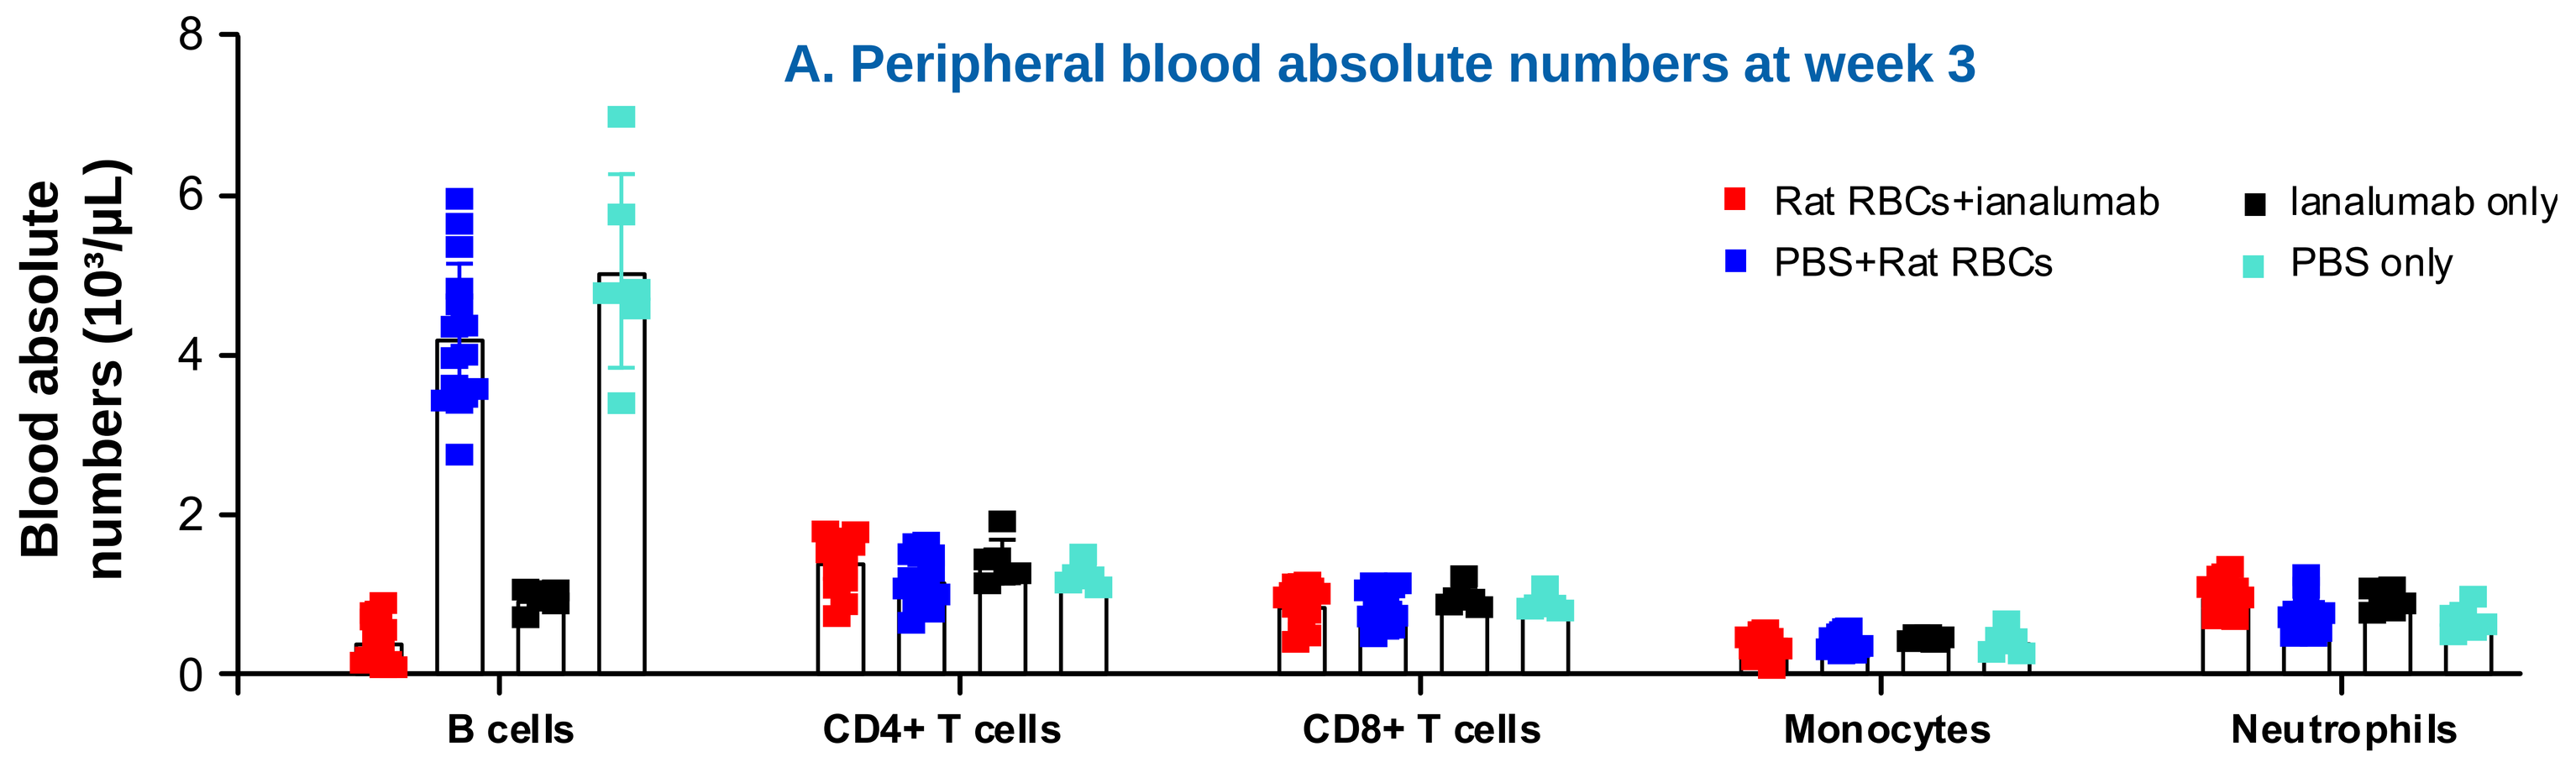
A. Peripheral blood absolute numbers at week 3
Blood absolute numbers (10³/µL)
Rat RBCs+ianalumab
PBS+Rat RBCs
Ianalumab only
PBS only
B cells
CD4+ T cells
CD8+ T cells
Monocytes
Neutrophils

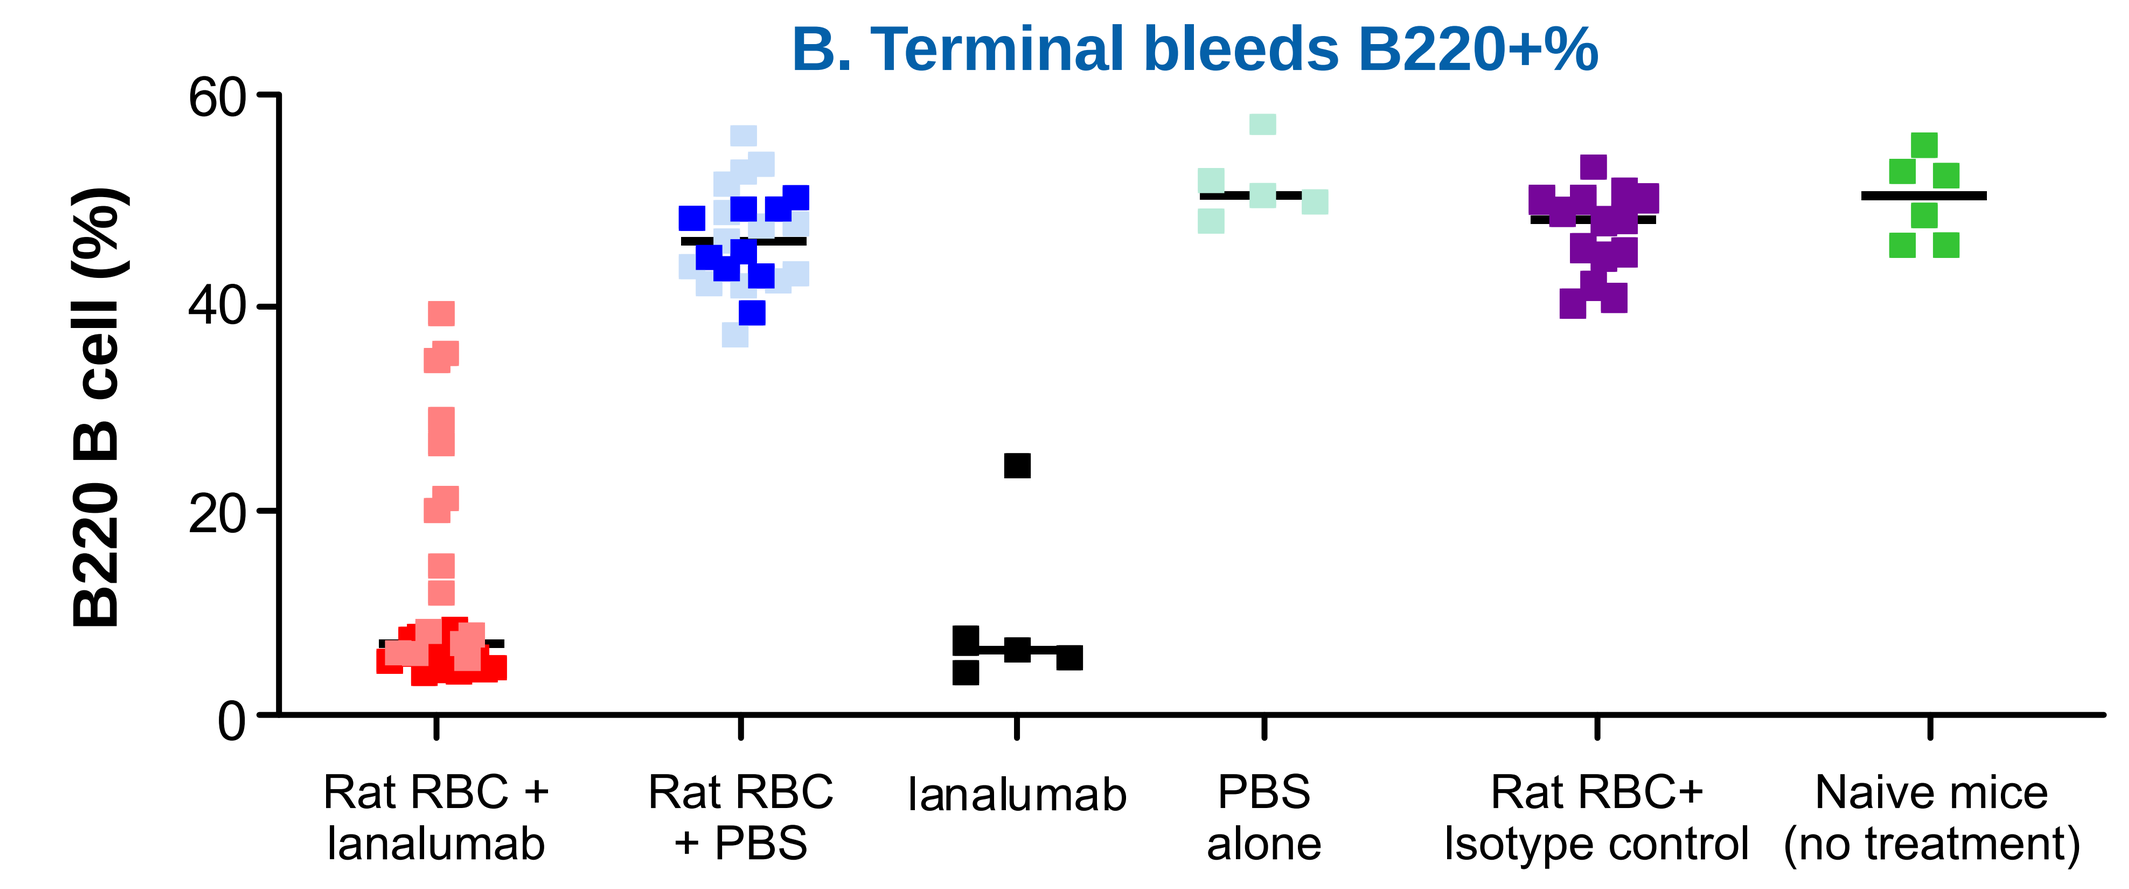
B. Terminal bleeds B220+%
B220 B cell (%)
Rat RBC + Ianalumab
Rat RBC + PBS
Ianalumab
PBS alone
Rat RBC+ Isotype control
Naive mice (no treatment)

A. Absolute counts of B cells and non-B-cell lineages at week 3 post-treatment with ianalumab from study 1. **B.** B-cell frequency in terminal bleeds from both prophylactic (bold colors) and therapeutic (light colors) studies were analyzed by flow cytometry.

PBS, phosphate-buffered saline; RBC, red blood cell.

Figure 4. Correlation between ianalumab plasma levels and the degree of B-cell depletion and reduction in anti-RBC auto-antibodies

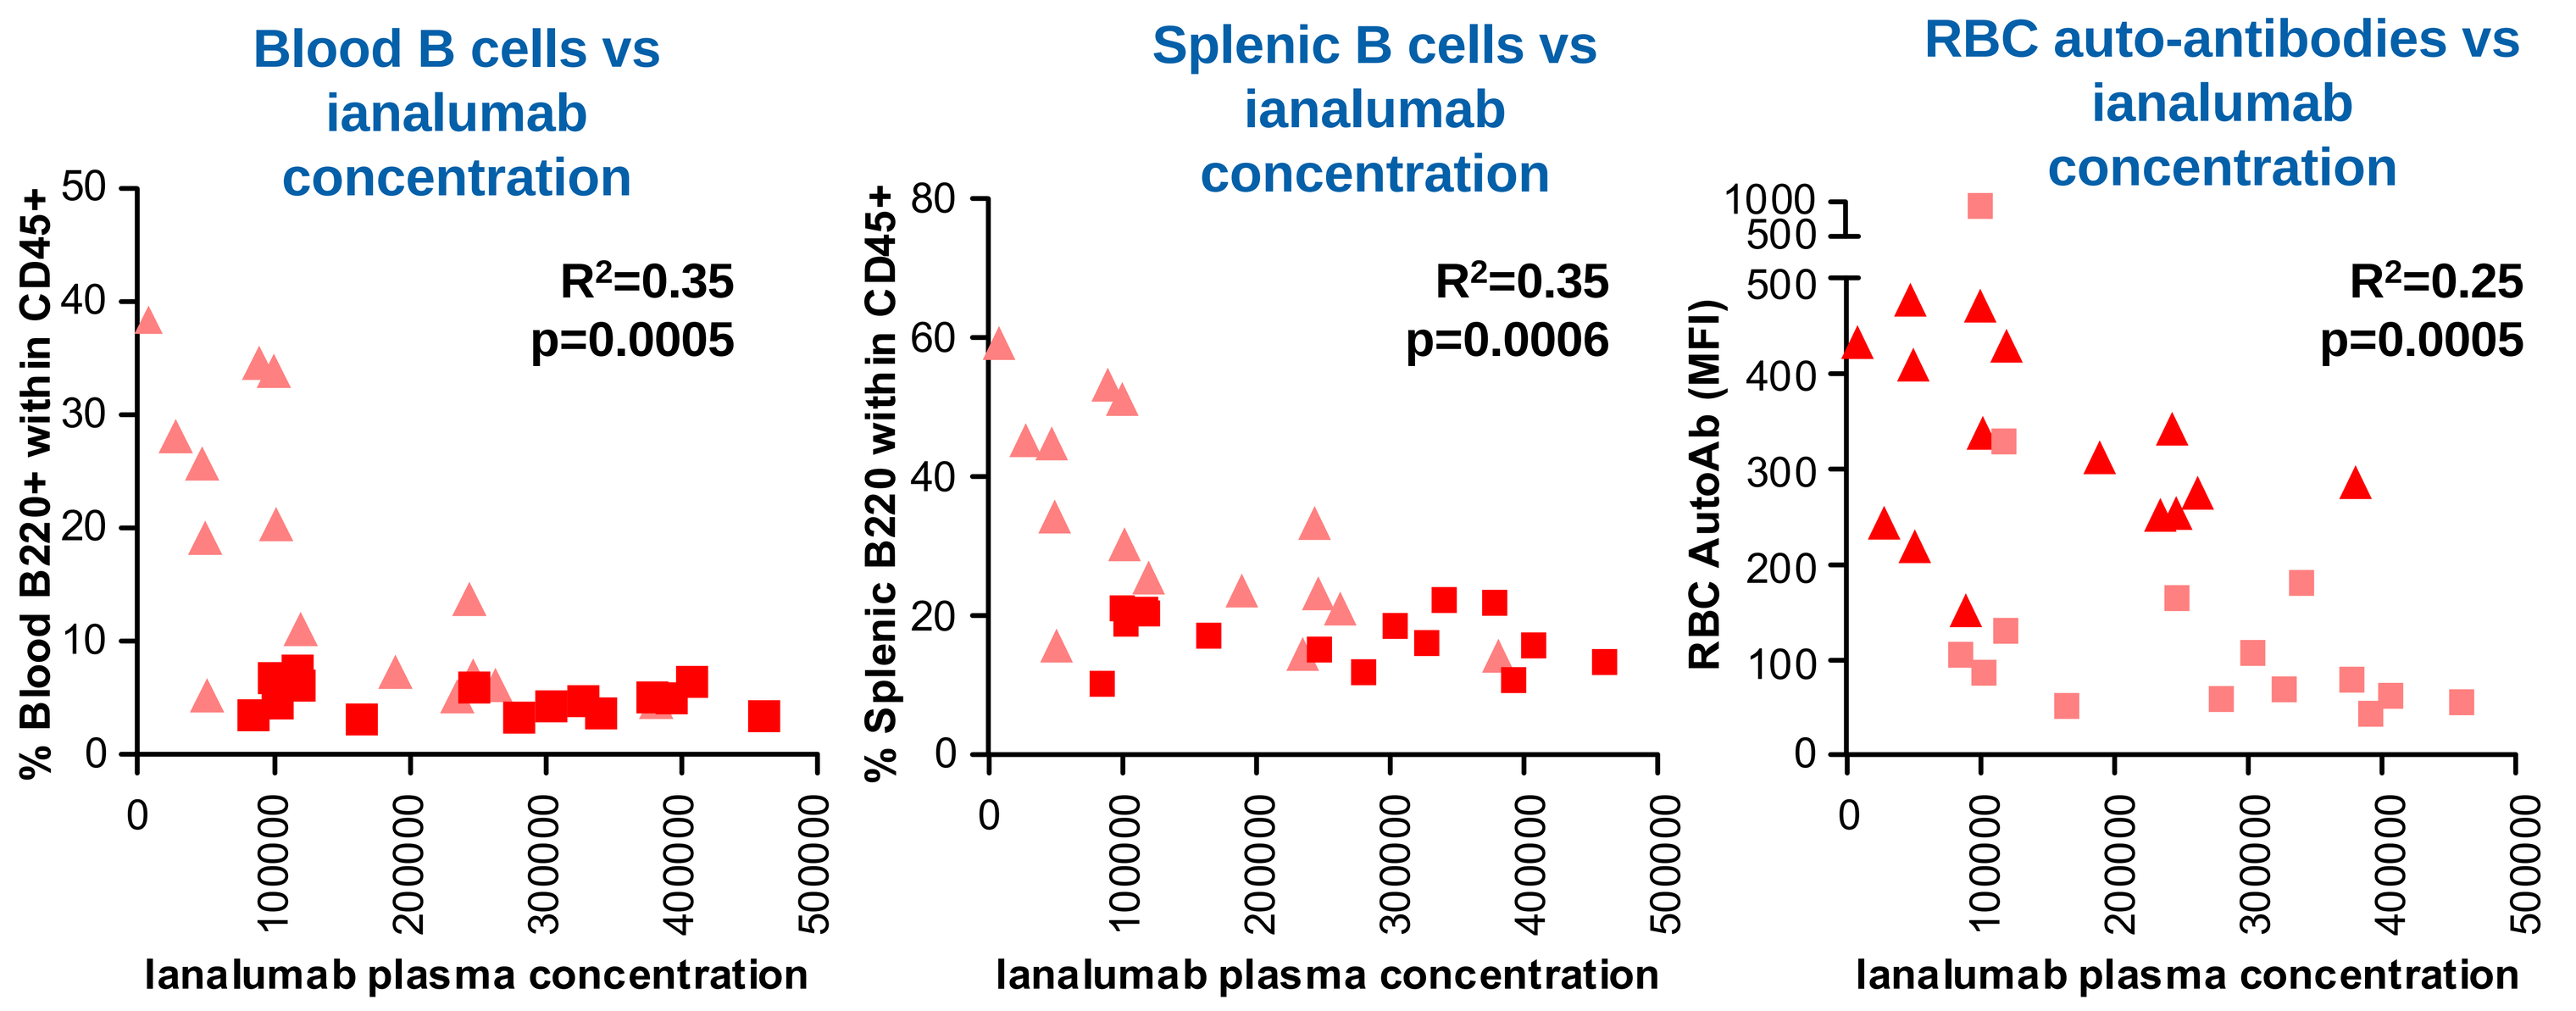
Blood B cells vs ianalumab concentration
% Blood B220+ within CD45+
R²=0.35
p=0.0005
Splenic B cells vs ianalumab concentration
% Splenic B220 within CD45+
R²=0.35
p=0.0006
RBC auto-antibodies vs ianalumab concentration
RBC AutoAb (MFI)
R²=0.25
p=0.0005
Ianalumab plasma concentration

Correlative analysis of B cells in peripheral blood, spleen and plasma anti-RBC auto-antibody levels with ianalumab plasma concentration in AIHA induced mice treated with ianalumab in prophylactic (bold red) and therapeutic (light red) studies.
AIHA, autoimmune hemolytic anemia; CD, cluster of differentiation; RBC, red blood cell.

Figure 5. Correlation of ADA with ianalumab concentration in blood

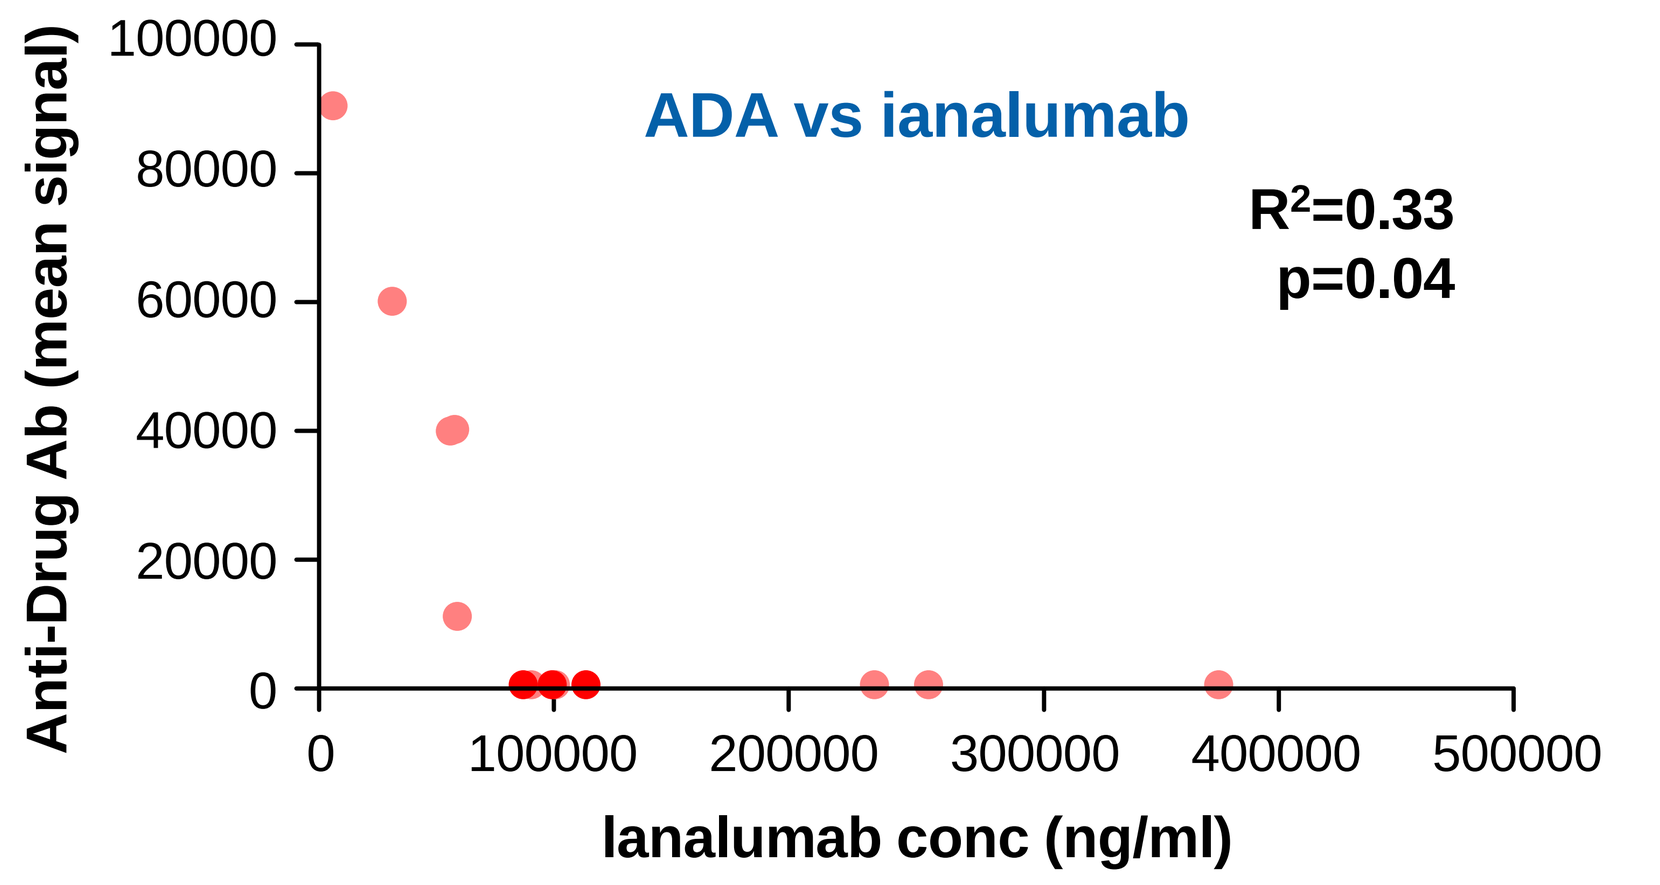
ADA vs ianalumab
Anti-Drug Ab (mean signal)
R²=0.33
p=0.04
Ianalumab conc (ng/ml)

Anti-ianalumab antibodies in mouse plasma were measured by electrochemiluminescence using biotin-labeled ianalumab for capture and sulfo tag-labeled ianalumab for detection. Correlative analysis of ADA with ianalumab plasma concentration in AIHA-induced mice treated with ianalumab in prophylactic (dark red) and therapeutic (light red) studies.

ADA, anti-drug antibodies; AIHA, autoimmune hemolytic anemia

Figure 6. Reduction of splenic marginal B cells

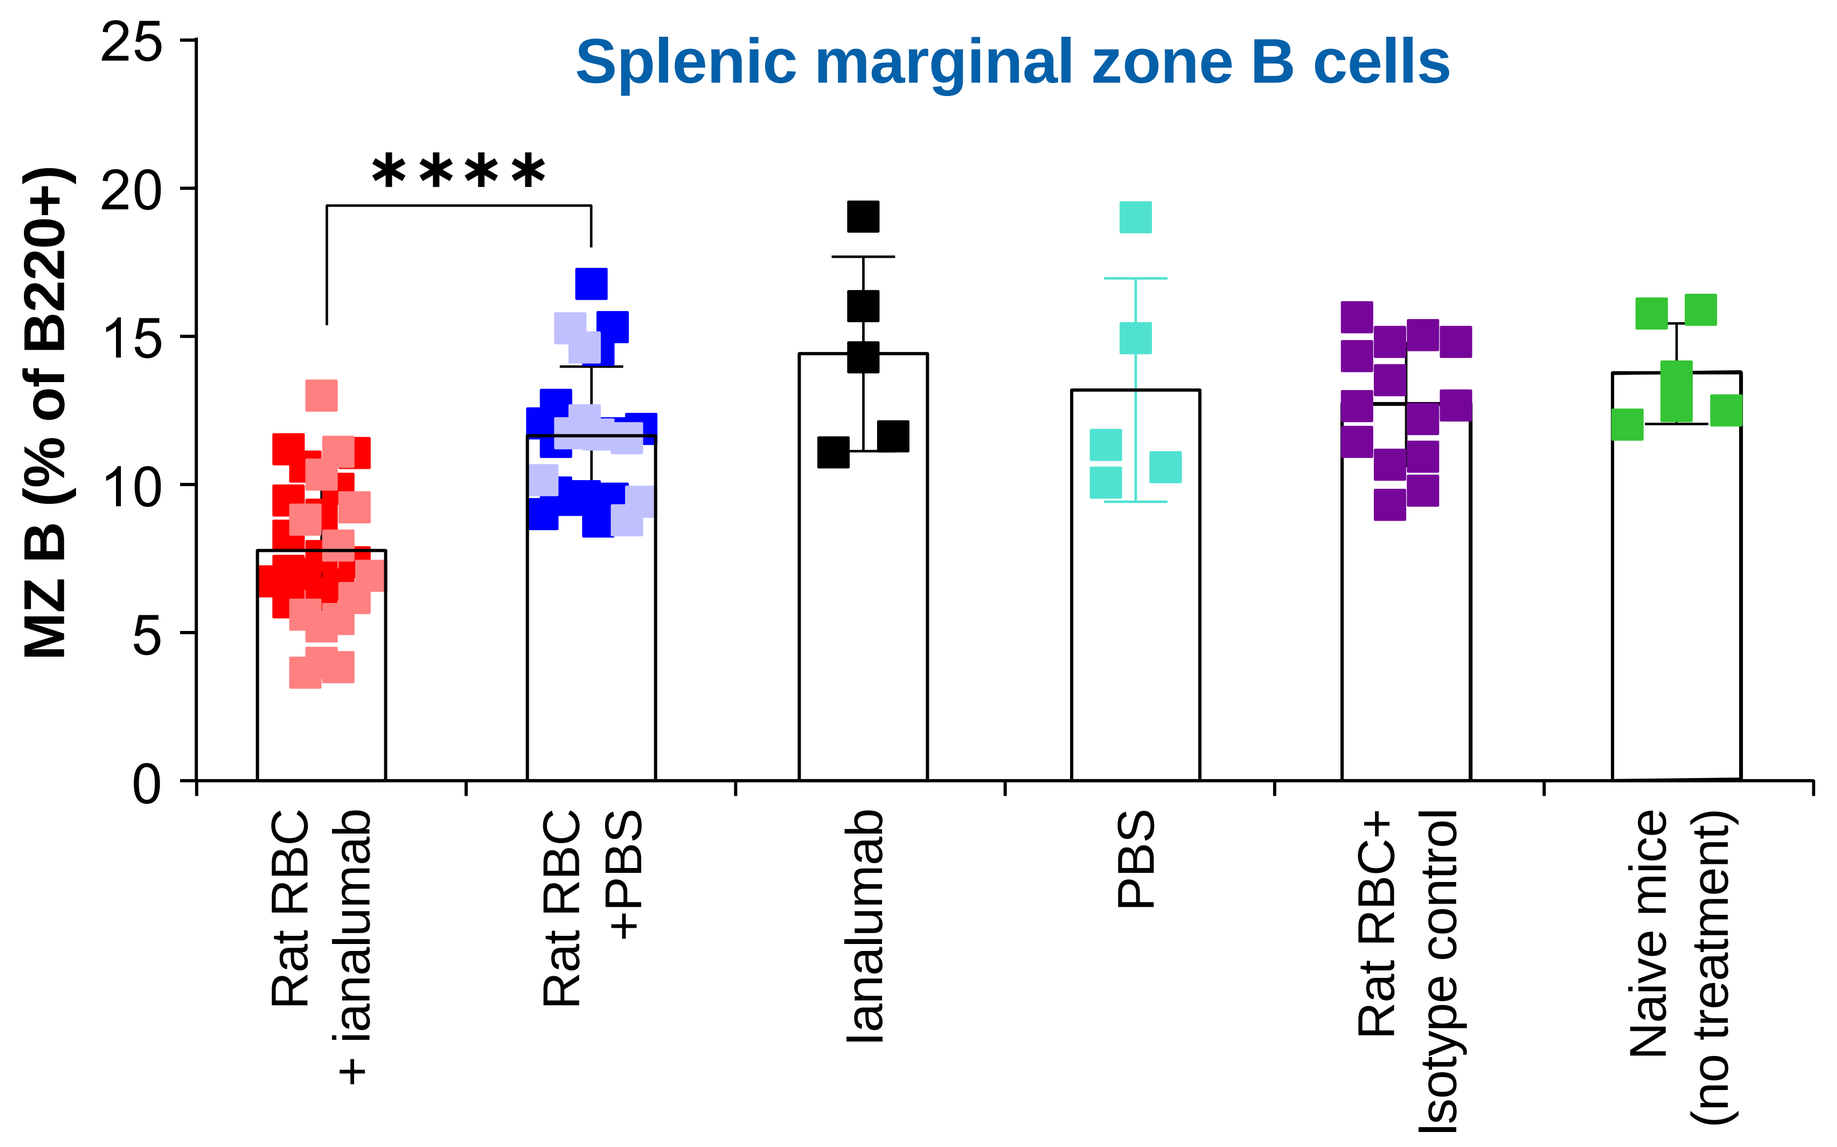
Splenic marginal zone B cells
MZ B (% of B220+)

Rat RBC + ianalumab
Rat RBC +PBS
Ianalumab
PBS
Rat RBC+ Isotype control
Naive mice (no treatment)

Frequency of MZ B cells within splenic B (B220+) cells were analyzed by flow cytometry in both prophylactic (bold colors) and therapeutic (light colors) studies.

MZ, marginal zone; PBS, phosphate-buffered saline; RBC, red blood cell..

Figure 7. Ianalumab corrected the Teff vs Treg imbalance

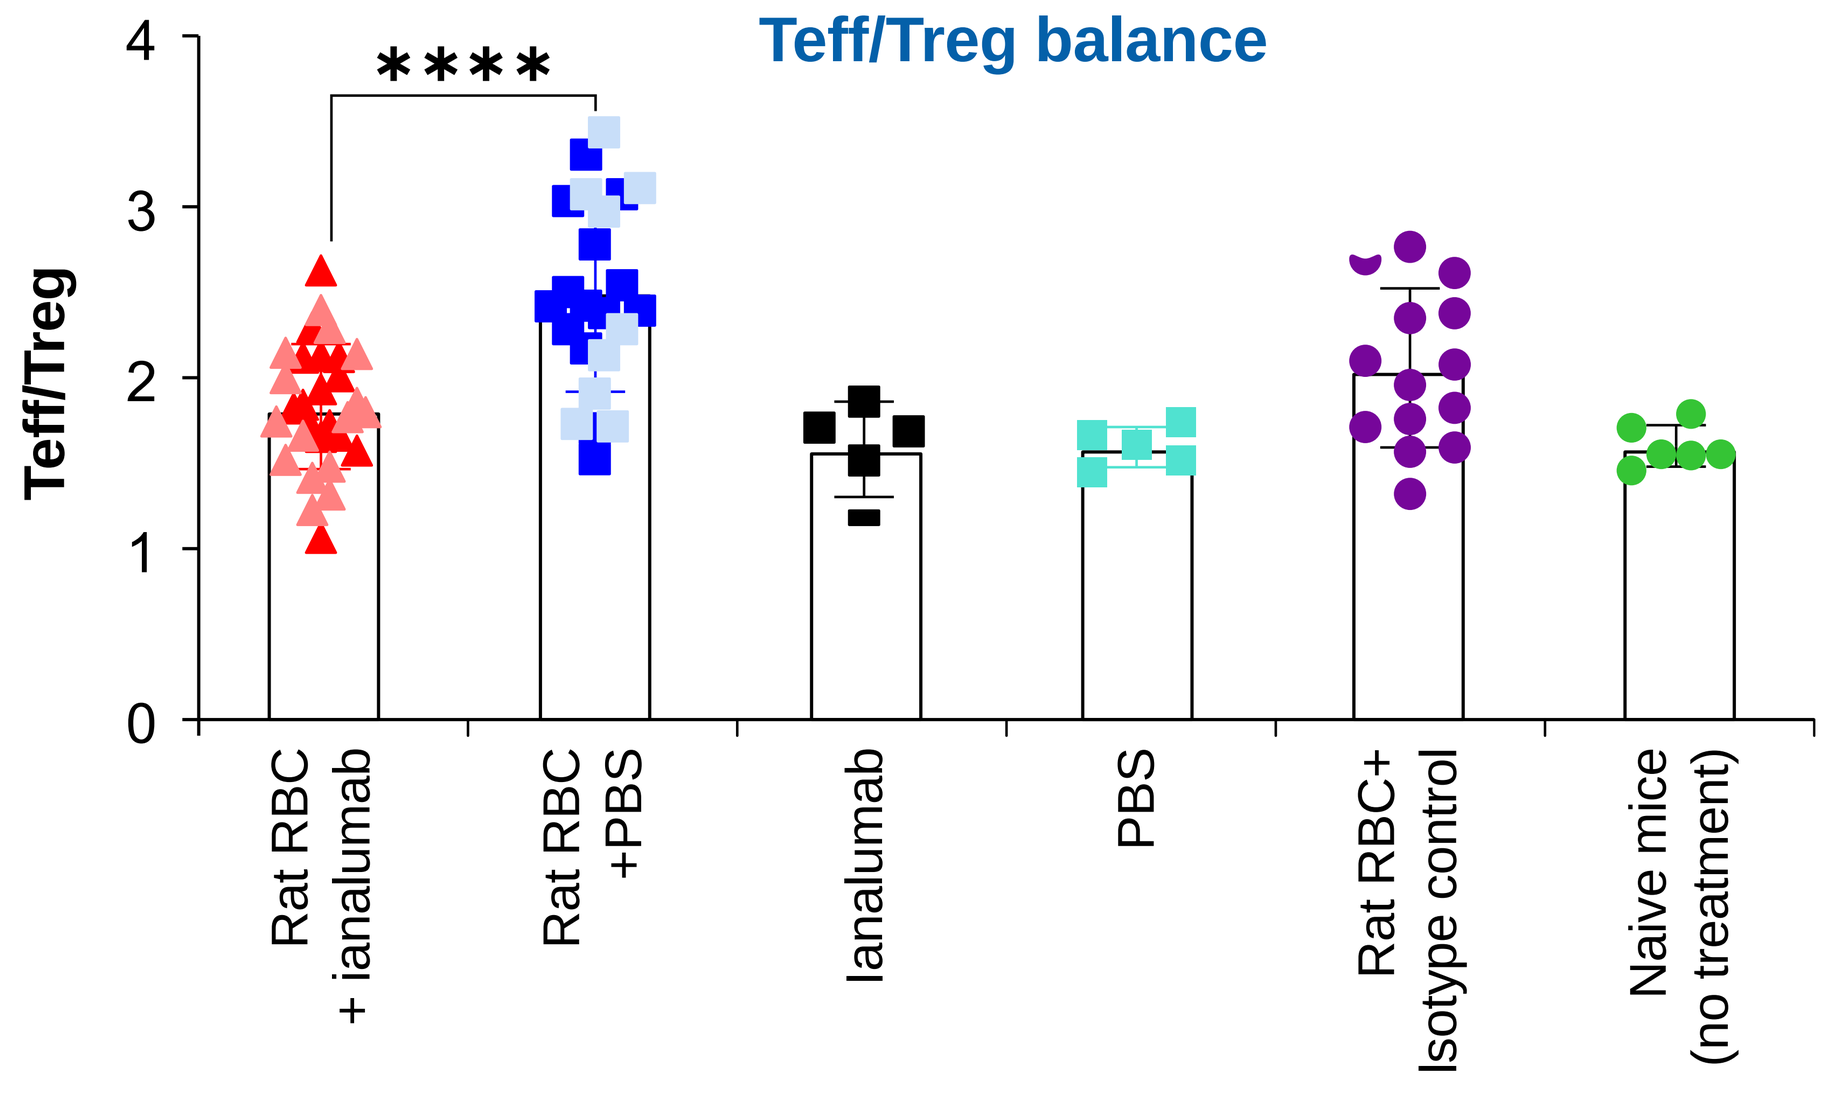
Teff/Treg balance
Teff/Treg

Rat RBC + ianalumab
Rat RBC +PBS
Ianalumab
PBS
Rat RBC+ Isotype control
Naive mice (no treatment)

Ratio of splenic effector T cells (Teff: CD4+CD44+) to T regulatory cells (Tregs:CD4+Foxp3+) in prophylactic (bold colors) and therapeutic (light colors) studies.

PBS, phosphate-buffered saline; RBC, red blood cells; Teff, effector T cells; Treg, regulatory T cells.

References

1. Barcellini W, Fattizzo B. *Front Immunol*. 2020;11:946.
2. McWilliams EM, et al. *Blood Adv*. 2019;3:447-60.
3. Playfair JHL, Marshall-Clarke S. *Nat New Biol*. 1973; 243:213-214.

Acknowledgments

All authors participated in the development of the poster. The authors thank Lalitha Venkateswaran and Deepak Kumar (Novartis Healthcare Pvt. Ltd., India) for editorial support, which was funded by Novartis Pharma AG, Basel, Switzerland in accordance with Good Publication Practice (GPP 2022) guidelines (https://www.ismpp.org/gpp-2022).

Disclosures

KY has received research funding from Novartis Pharma AG, Switzerland and Zenas Biopharma, US. WB, SS, and YL have nothing to declare. MB, TU, PE, BN, II, and CB are employees of Novartis Pharma AG, Basel, Switzerland and hold company stocks. EF is an employee of Novartis Biomedical Research, Cambridge, USA and holds company stocks.