PF596

Markus Metzler | markus.metzler@uk-erlangen.de

New Pediatric Formulation of Asciminib in Children With Chronic Myeloid Leukemia in Chronic Phase: Second Interim Analysis of Pharmacokinetics, Safety and Growth Data from the ASC4Kids Study

Markus Metzler[1], Jessica Pollard[2,3], Hiroyuki Shimada[4], Shruti Kapoor[5], Matthias Hoch[6], Vishal Dhamal[7], Nithya Agrawal[5], Christine Rosko[5], Ana Paula Cardoso[5], Masakatsu Yanagimachi[8], Ho Joon Im[9], Nobuko Hijiya[10]

[1]Department of Pediatrics and Adolescent Medicine, University Hospital Erlangen, Erlangen, Germany; [2]Dana-Farber/Boston Children's Cancer and Blood Disorders Center, Boston, MA, USA; [3]Harvard Medical School, Boston, MA, USA; [4]Keio University School of Medicine, Tokyo, Japan; [5]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA; [6]Novartis Institute for BioMedical Research, Basel, Switzerland; [7]Novartis Healthcare Private Limited, Mumbai, India; [8]Kanagawa Children's Medical Center, Yokohama, Japan; [9]Department of Pediatrics, Asan Medical Center Children's Hospital, University of Ulsan College of Medicine, Seoul, Republic of Korea; [10]Pediatric Oncology, Columbia University Irving Medical Center, New York, NY, USA

KEY FINDINGS & CONCLUSIONS

- The confirmed PF asciminib dose of 1.3 mg/kg BID was well tolerated in pediatric pts aged 12–<18 years, with evidence of efficacy and no new safety signals observed
- Preliminary data suggest asciminib, unlike other TKIs, has a positive impact on growth for some pts
- A once-daily 2.6 mg/kg dose will be evaluated in Part 3 of this study

Scan to obtain:
- Poster

https://tinyurl.com/MetzlerPF596

Copies of this poster obtained through Quick Response (QR) code are for personal use only and may not be reproduced without permission of the authors.

This study was sponsored by Novartis Pharma AG, Basel, Switzerland.

Poster presented at EHA Annual Meeting 2025, Milan, Italy & Online, June 12–15 2025

INTRODUCTION

- More treatment options to improve efficacy and long-term safety to minimize adverse effects on growth are needed for pediatric pts with Ph+ CML-CP
 - Exposure to standard of care TKIs during active growth and puberty may affect growth rate, puberty, bone metabolism and fertility,[1–3] despite improved outcomes[1,4,5]
 - Approved TKIs (dasatinib, imatinib, and nilotinib) have been associated with growth delays in pediatric pts[6–8]
- Asciminib is the first BCR::ABL1 inhibitor that Specifically Targets the ABL Myristoyl Pocket (STAMP), approved for adults with:
 - Newly diagnosed (US)[9] or
 - Previously treated CML-CP (≥2 TKIs, worldwide)[9,10]
- The phase Ib/II, multi-center, open-label ASC4Kids study (NCT04925479) aims to identify the pediatric formulation (PF) dose (fed) leading to asciminib exposure comparable to the adult formulation (AF) dose (fasted) of 40 mg BID and assess its safety

METHODS

- The ASC4Kids study includes pts aged 1–<18 yrs with Ph+ CML-CP, without the T315I mutation, treated with ≥1 prior TKIs
- The primary objective of the study is to characterize the PK of the asciminib PF and identify a pediatric dose with the PF (in fed conditions) for which the exposure is comparable to that in adults treated with the AF given at 40 mg BID (under fasted conditions)
- Secondary endpoints include safety and molecular responses
- The study has 3 parts (**Figure 1**), and plans to enroll ≥30 evaluable pediatric pts to be treated with the PF
 - Part 1 (dose determination): The PF dose of 1.3 mg/kg BID was confirmed based on exposure in adult studies (at 40 mg BID) and no observed DLTs over the first 28 days
 - In Part 2 (dose expansion, BID): Additional pts are treated with the confirmed PF dose for further evaluation of exposure and DLTs (across Parts 1 + 2; 10 pts per group: 1–<12 yrs and 12–<18 yrs)
 - In Part 3 (dose expansion, QD): 10 more pts will be enrolled to receive PF 2.6 mg/kg QD (in fed conditions)
- An exploratory group of pts aged 14–<18 yrs who were treated with the AF of 40 mg BID (under fasted conditions) is also included (data not shown)
- This interim analysis with a data cutoff of August 19, 2024 was conducted after 10 evaluable pts in the PF 12–<18 yrs group had completed 28 days of treatment in Parts 1 + 2. PK is reported for the PF 12–<18 yrs group; safety, growth and efficacy is reported for the overall PF group

Figure 1. ASC4Kids study design

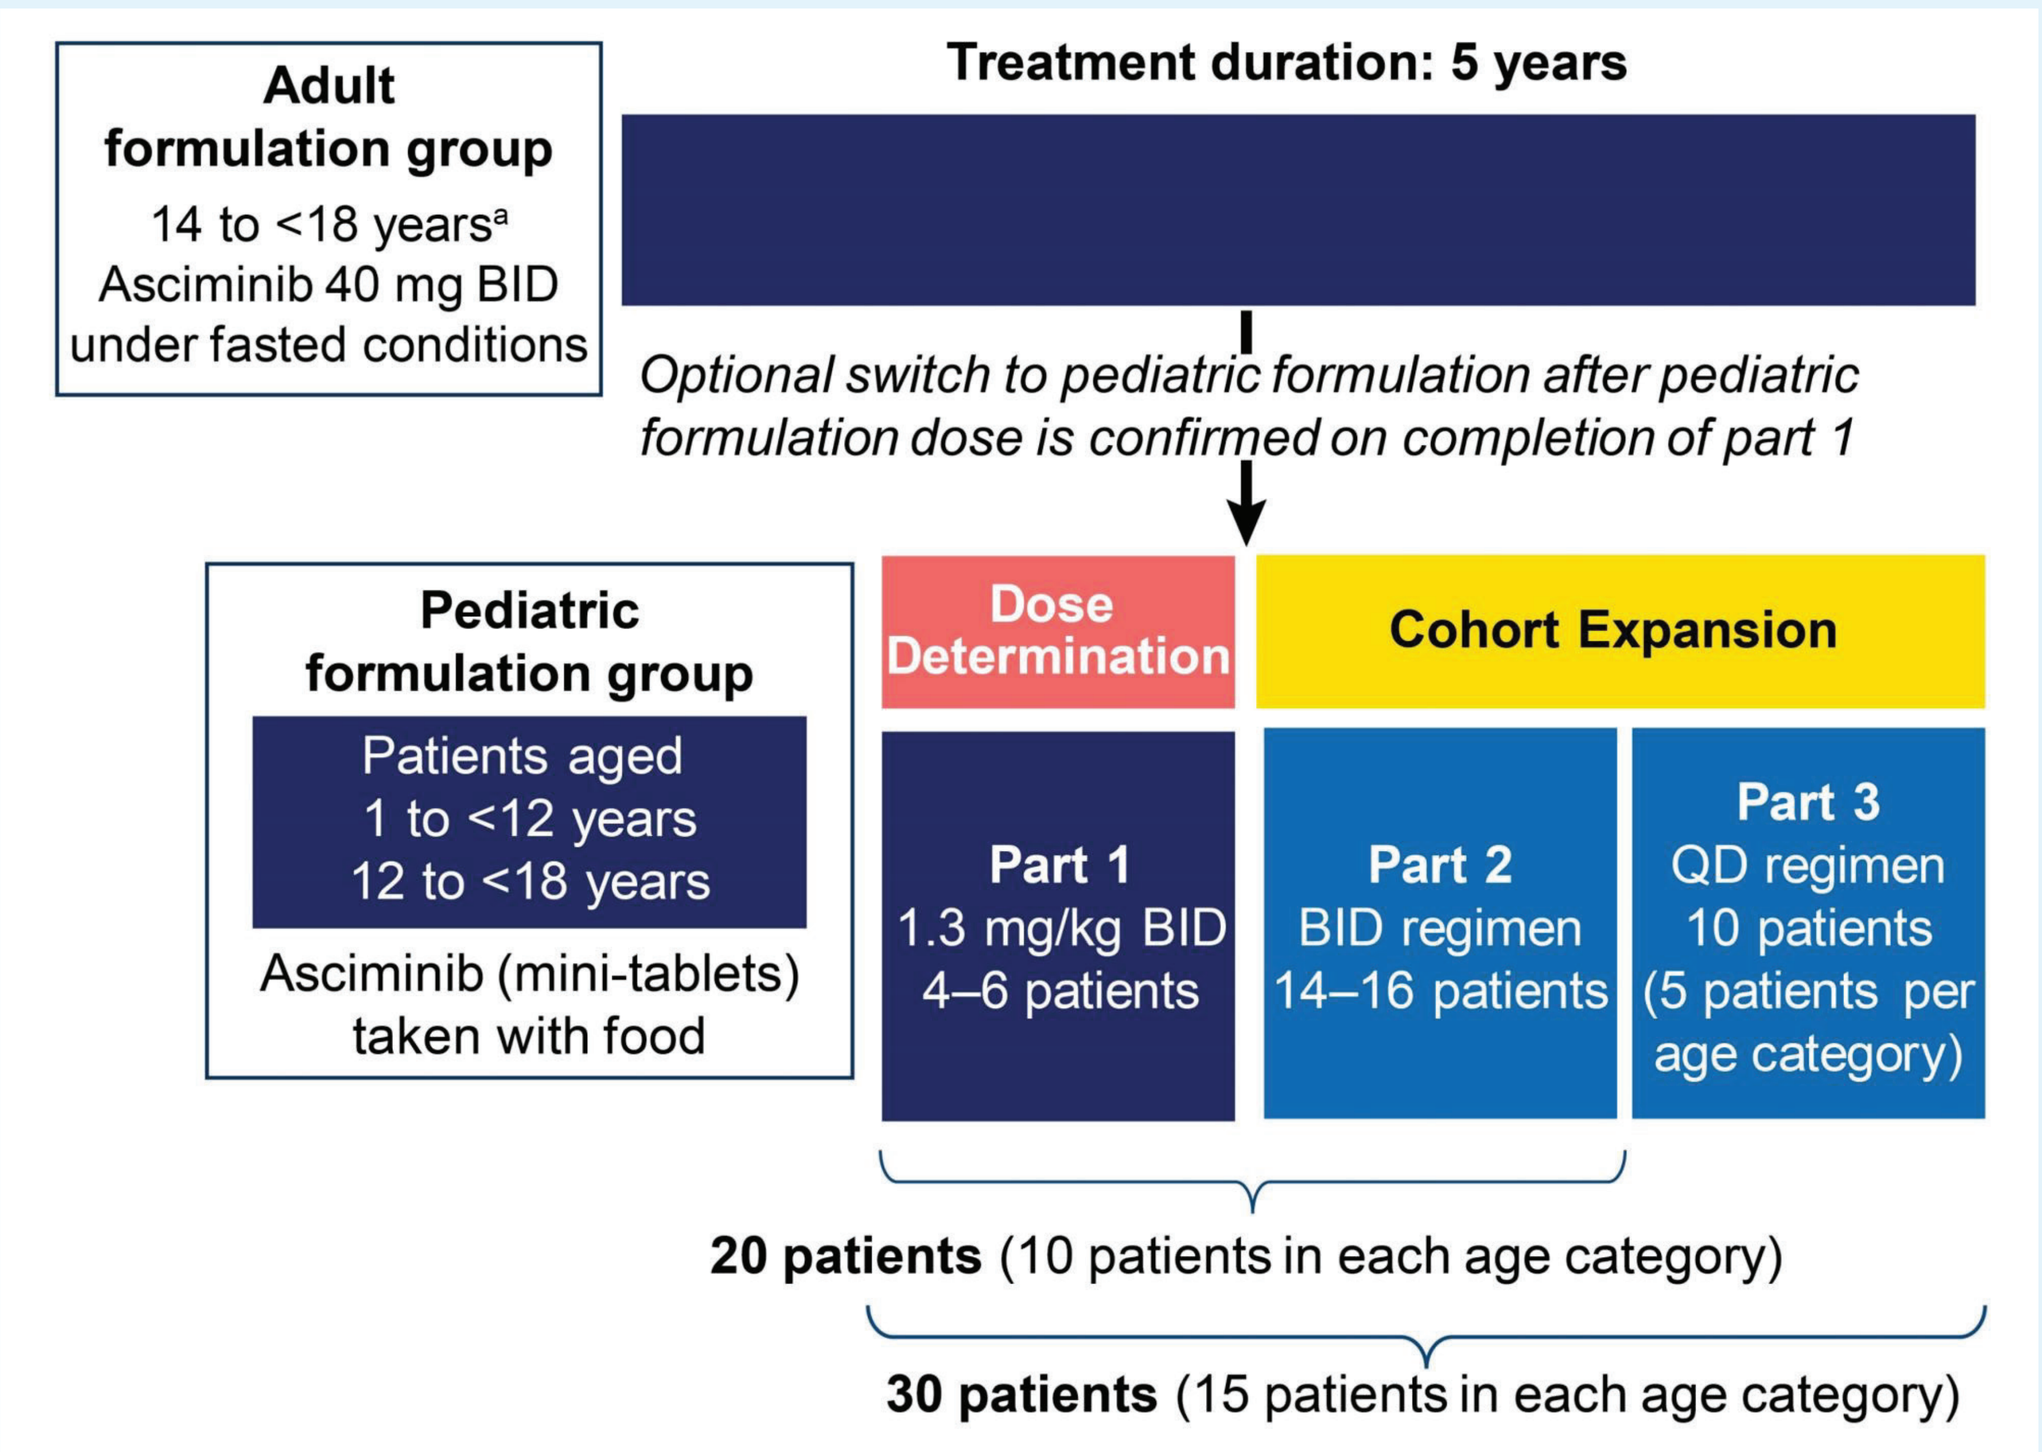
Adult formulation group
14 to <18 years[a]
Asciminib 40 mg BID under fasted conditions
Treatment duration: 5 years
Optional switch to pediatric formulation after pediatric formulation dose is confirmed on completion of part 1
Pediatric formulation group
Patients aged
1 to <12 years
12 to <18 years
Asciminib (mini-tablets) taken with food
Dose Determination
Cohort Expansion
Part 1
1.3 mg/kg BID
4–6 patients
Part 2
BID regimen
14–16 patients
Part 3
QD regimen
10 patients
(5 patients per age category)
20 patients (10 patients in each age category)
30 patients (15 patients in each age category)

[a]must weigh ≥40 kg.

RESULTS

Patient demographics and characteristics

- 19 pts were enrolled in the PF group; 7 in the 1–<12 yrs and 12 in the 12–<18 yrs groups, respectively (**Table 1**)
- Median duration of asciminib treatment was 36.7 weeks (range 3.7–102.6)
- At data cutoff (August 19, 2024), all pts continued to receive treatment

Table 1. Patient demographics and disease characteristics

	Pediatric formulation 1 to <12 years (N=7)	Pediatric formulation 12 to <18 years (N=12)	Pediatric formulation All (N=19)
Age (years), median (range)	8.3 (2–11.5)	13.0 (12–17)	12.3 (2–17)
Male, n (%)	3 (42.9)	6 (50.0)	9 (47.4)
Height (cm), median (range)	122.0 (83.0–153.5)	150.8 (139.8–172.9)	146.0 (83.0–172.9)
Weight (kg), (median range)	23.9 (10.8–54.0)	46.0 (28.4–88.2)	36.0 (10.8–88.2)
BMI (kg/m^2), median (range)	16.1 (15.5–22.9)	19.1 (14.0–35.9)	16.9 (14.0–35.9)

PK

- For the older PF group (12–<18 yrs), 10 pts were evaluable for PK. The average asciminib exposure with PF 1.3 mg/kg BID (median [range]) was comparable to that observed in adult studies (10th and 90th percentile of median values averaged from Phase 3 ASCEMBL trial data on adults treated with asciminib [data not published]) as shown in **Figure 2**:
 - AUC_{last}: 7091 (2701.3–12518.3) vs 5130 (2683.7–8476.3) hr*ng/mL in ASCEMBL
 - C_{max}: 1031 (343.0–2160.0) vs 939 (473.0–1547.0) ng/mL in ASCEMBL
- Final interim analysis data for the younger PF group (12–<18 yrs) are pending

Figure 2. Asciminib exposure by AUC_{last} and C_{max} in the PF group

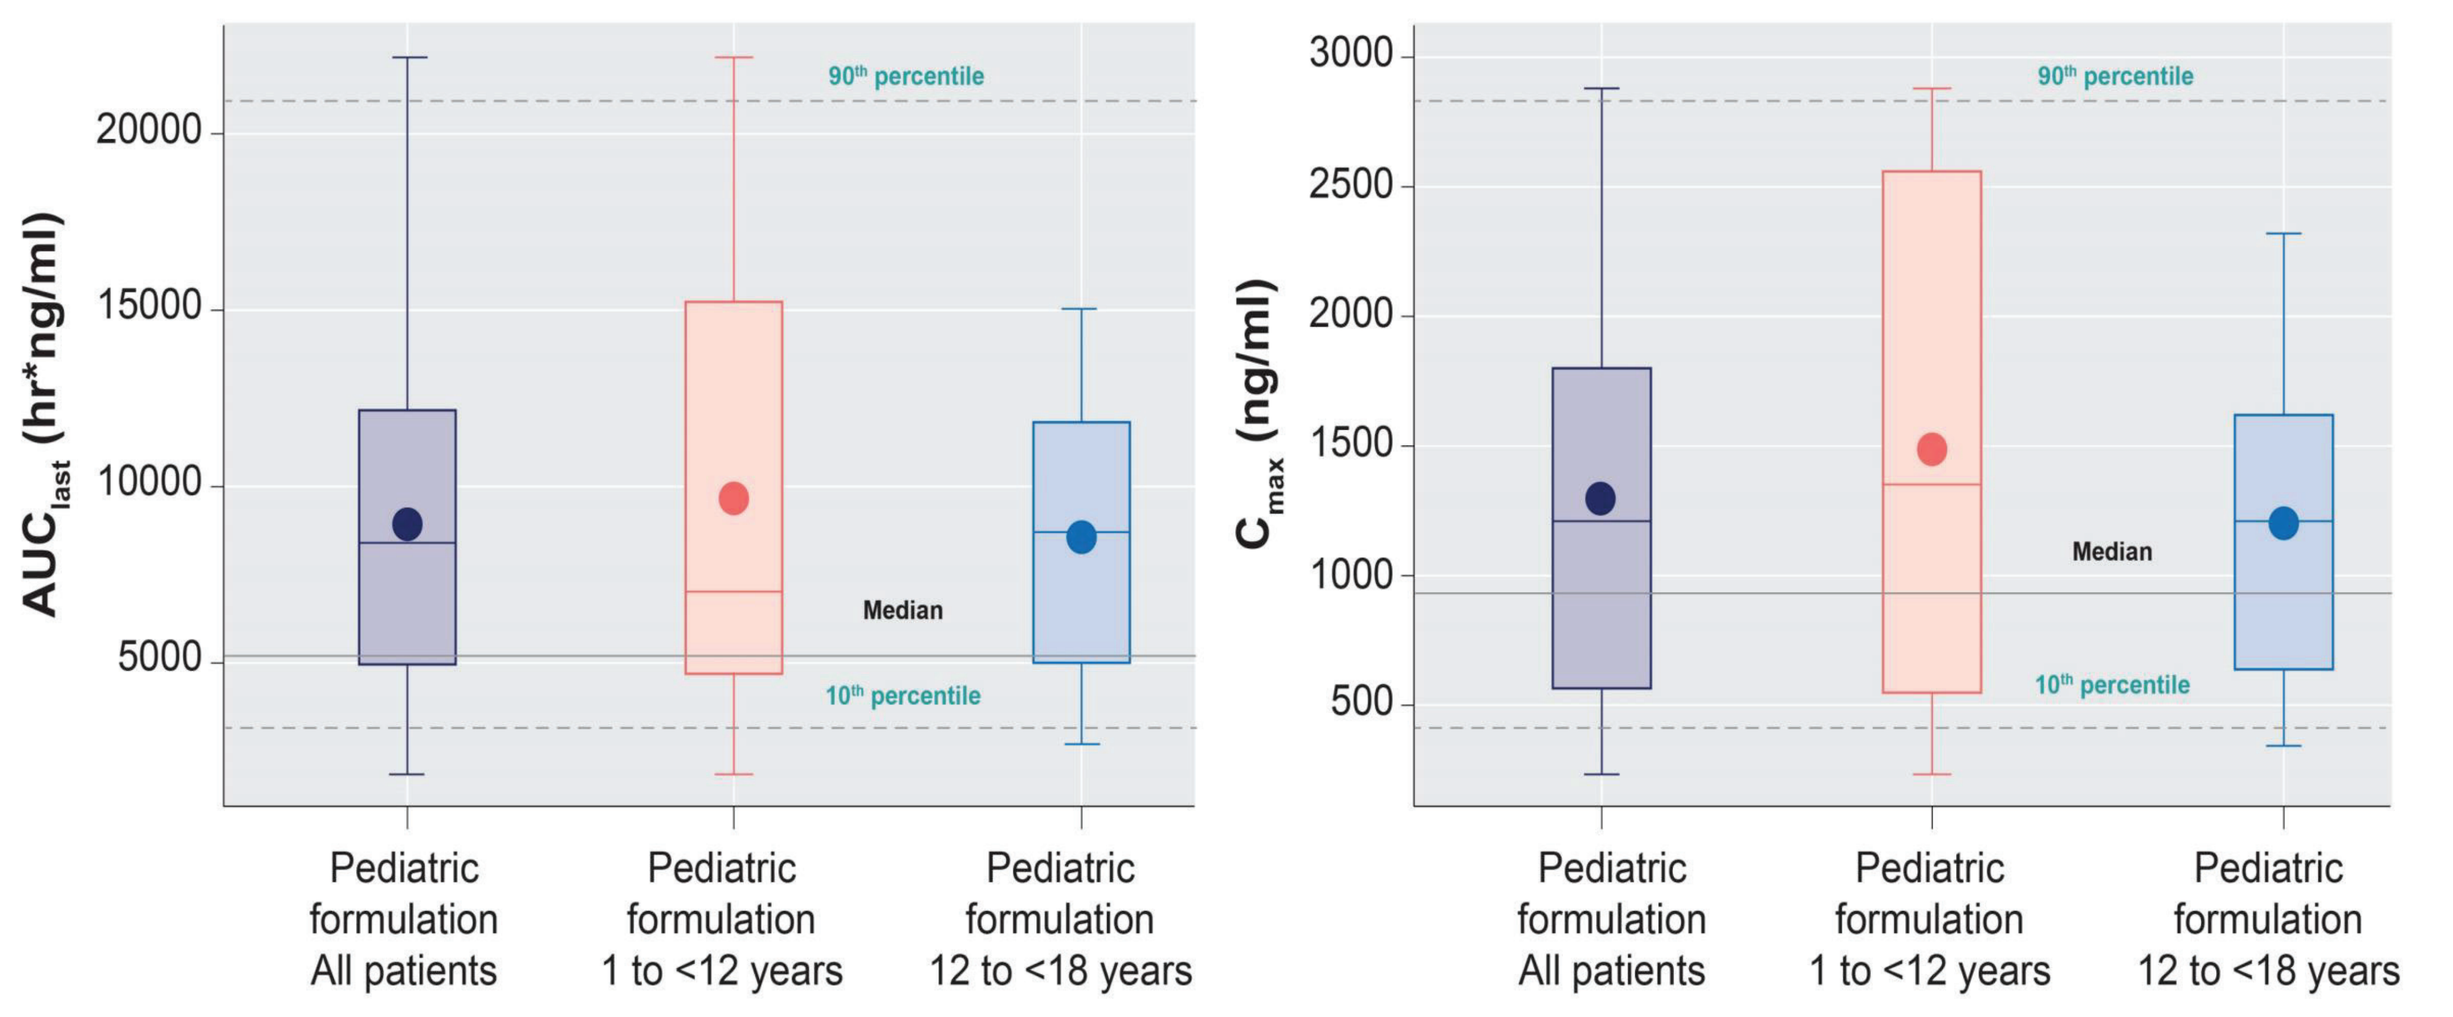
AUClast (hr*ng/ml)
20000
15000
10000
5000
Cmax (ng/ml)
3000
2500
2000
1500
1000
500
90th percentile
Median
10th percentile
Pediatric formulation All patients
Pediatric formulation 1 to <12 years
Pediatric formulation 12 to <18 years

Boxes show the IQR range of data within each group. Full line represents median (with dotted lines representing 10th and 90th percentile) of PK parameters averaged from the ASCEMBL study (Week 2 Day 1 visit, n=14, assessing adults with Ph+ CML-CP treated with ≥2 TKIs. Circles within the box plots represent the mean values. Lower and upper whiskers extend to the most extreme points within 1.5x of Q1 and Q3, respectively.

Safety

- Nearly all pts (18/19) in the overall PF group experienced at least one AE (**Table 2**)
 - The most common AEs were vomiting and upper respiratory tract infection
 - Grade ≥3 AEs were reported in 2 pts (leukopenia and neutropenia, 1 pt; abdominal pain, 1 pt)
- No Grade 4 AEs, serious AEs, DLTs or AEs leading to discontinuation were reported
- AEs leading to dose interruption and/or adjustment occurred in 3 pts (**Table 3**)

Table 2. AEs (≥10%) by preferred term for the overall PF group

	Pediatric formulation 1 to <12 years (N=7)		Pediatric formulation 12 to <18 years (N=12)		Pediatric formulation All (N=19)	
	All grades, n (%)	Grade ≥3, n (%)	All grades, n (%)	Grade ≥3, n (%)	All grades, n (%)	Grade ≥3, n (%)
Number of pts with at least 1 event	6 (85.7)	1 (14.3)	12 (100)	1 (8.3)	18 (94.7)	2 (10.5)
Vomiting	3 (42.9)	0	3 (25.0)	0	6 (31.6)	0
Upper respiratory tract infection	3 (42.9)	0	1 (8.3)	0	4 (21.1)	0
Diarrhea	2 (28.6)	0	1 (8.3)	0	3 (15.8)	0
Pyrexia	2 (28.6)	0	1 (8.3)	0	3 (15.8)	0
Abdominal pain	1 (14.3)	0	2 (16.7)	1 (8.3)	3 (15.8)	1 (5.3)
Stomatitis	1 (14.3)	0	2 (16.7)	0	3 (15.8)	0
Fatigue	1 (14.3)	0	2 (16.7)	0	3 (15.8)	0
Headache	1 (14.3)	0	2 (16.7)	0	3 (15.8)	0
Rash	1 (14.3)	0	2 (16.7)	0	3 (15.8)	0
Neutropenia	2 (28.6)	1 (14.3)	0	0	2 (10.5)	1 (5.3)
Bone pain	1 (14.3)	0	1 (8.3)	0	2 (10.5)	0
Blood alkaline phosphatase increased	0	0	2 (16.7)	0	2 (10.5)	0
Cough	2 (28.6)	0	0	0	2 (10.5)	0

Table 3. AEs leading to dose interruption and/or adjustment in the overall PF group

AE	Grade	Event	Duration (days)	Related to asciminib	Status at data cutoff
1 pt (8 yrs old)	3	Worsening neutropenia	14	✓	Resolving
1 pt (8 yrs old)	1	Vomiting	2	X	Resolved
1 pt (13 yrs old)	1	Vomiting and abdominal pain	1	X	Resolved

Growth

- For height percentile shift from baseline to data cutoff in the overall PF group; 9 pts stayed in the same percentile, 4 dropped and 6 increased
- Asciminib had a positive effect on height velocity in most patients in the overall PF group (**Figure 3**)
- Four of 19 pts had notably low (SDS <-2) bone age at baseline; 3 of these also at Week 52

Figure 3. Height development in male and female pts in the overall PF group during asciminib therapy

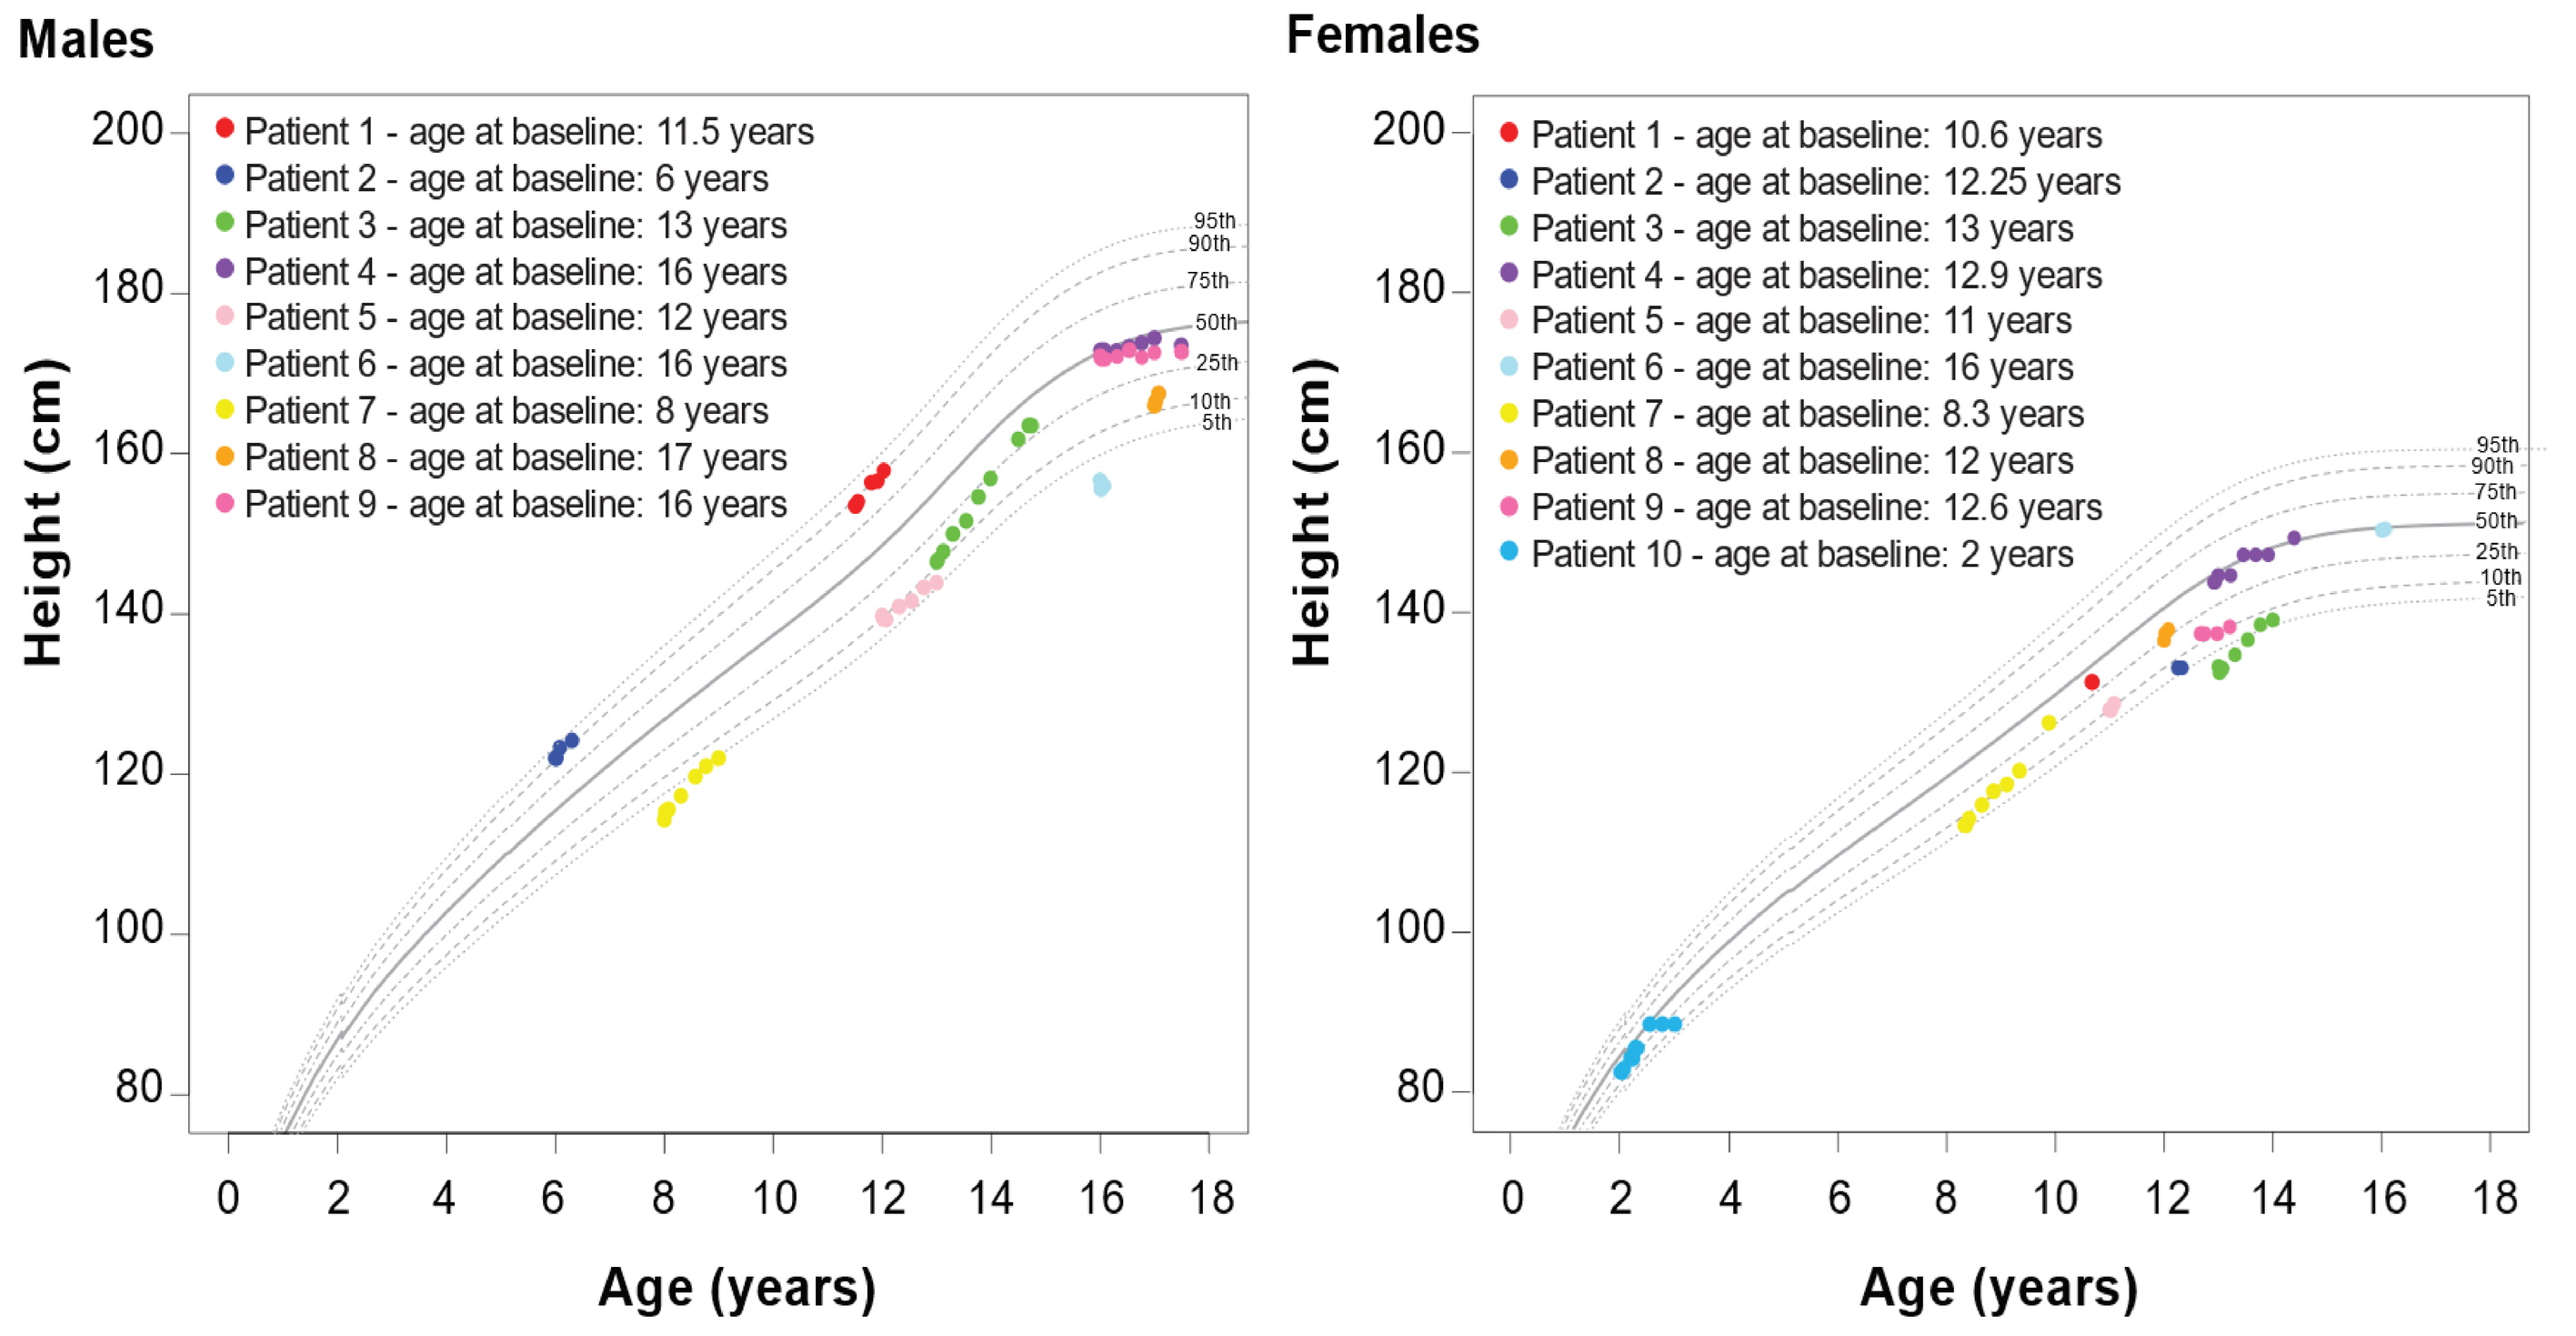
Males
Patient 1 - age at baseline: 11.5 years
Patient 2 - age at baseline: 6 years
Patient 3 - age at baseline: 13 years
Patient 4 - age at baseline: 16 years
Patient 5 - age at baseline: 12 years
Patient 6 - age at baseline: 16 years
Patient 7 - age at baseline: 8 years
Patient 8 - age at baseline: 17 years
Patient 9 - age at baseline: 16 years
Females
Patient 1 - age at baseline: 10.6 years
Patient 2 - age at baseline: 12.25 years
Patient 3 - age at baseline: 13 years
Patient 4 - age at baseline: 12.9 years
Patient 5 - age at baseline: 11 years
Patient 6 - age at baseline: 16 years
Patient 7 - age at baseline: 8.3 years
Patient 8 - age at baseline: 12 years
Patient 9 - age at baseline: 12.6 years
Patient 10 - age at baseline: 2 years
95th
90th
75th
50th
25th
10th
5th
Height (cm)
Age (years)

Efficacy

- For almost all patients, BCR::ABL1^{IS} levels generally remained stable through Week 40 (**Figure 4**)
 - At baseline,16/19 pts in the PF group had BCR::ABL1^{IS} ≤10%; 6 were in MMR (BCR::ABL1^{IS} ≤0.1%)
 - Of pts evaluable at Weeks 28 and 40, 10/11 and 9/9 pts had BCR::ABL1^{IS} ≤1%, and 7/11 6/9 pts were in MMR, respectively

Figure 4. Asciminib efficacy in individual pts from the PF group

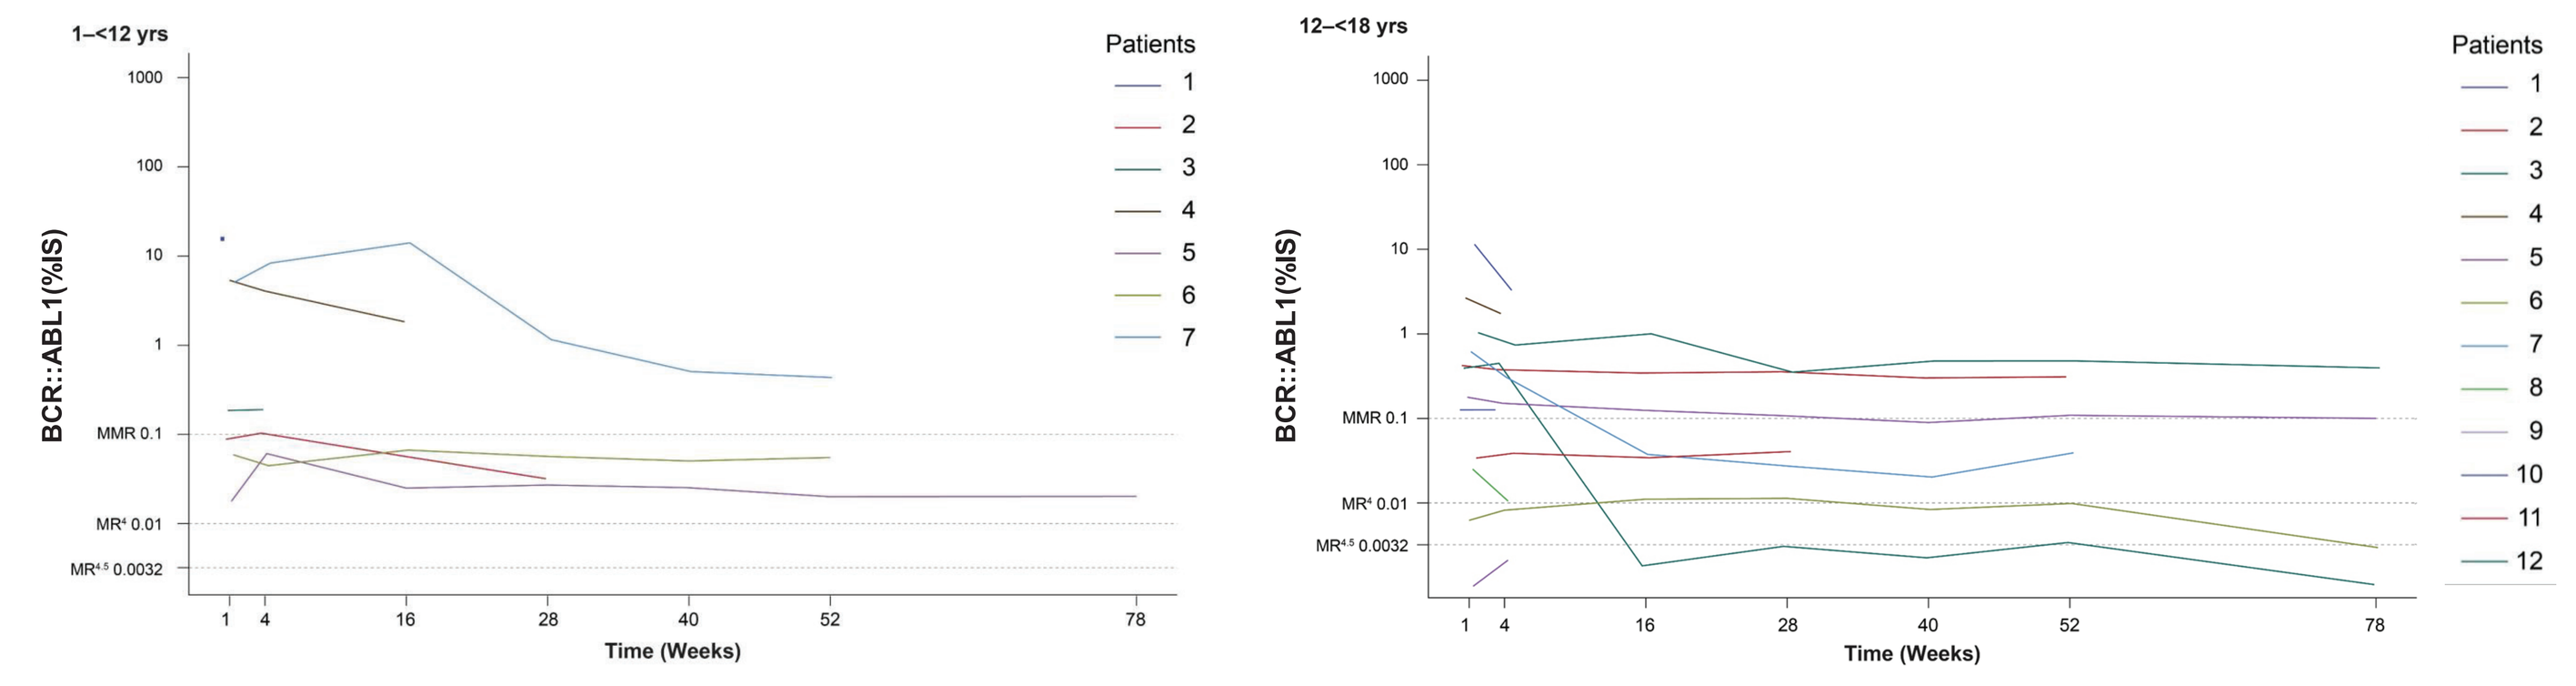
1–<12 yrs
12–<18 yrs
BCR::ABL1(%IS)
MMR 0.1
MR4 0.01
MR4.5 0.0032
Time (Weeks)
Patients

Pt 1 in the 1–<12 yrs age group only has the baseline assessment, as shown as a dot on the graph.

Acknowledgements

The authors would like to thank the pts and their families, as well as the investigators and staff at the participating study sites. Editorial assistance was provided by Emma Richards-Sirianni, PhD, of Novartis UK Ltd., and was funded by Novartis Pharmaceuticals Corporation in accordance with Good Publication Practice (GPP 2022) guidelines (https://www.ismpp.org/gpp-2022).

Abbreviations

AE, adverse event; AF, adult formulation; AUC_{last}, area under the curve from dosing to the time of the last measured concentration; BID, twice daily; BMI, body mass index; C_{max}, maximum plasma concentration; CML-CP, chronic myeloid leukemia in chronic phase; CTCAE, Common Terminology Criteria for Adverse Events; DLT, dose-limiting toxicity; IQR, interquartile range; kg, kilogram; MedDRA, Medical Dictionary for Regulatory Activities; mg, milligram; MMR, major molecular response (BCR::ABL1^{IS} ≤0.1%); MR^4, deep molecular response (BCR::ABL1^{IS} ≤0.01%), $MR^{4.5}$, deep molecular response (BCR::ABL1^{IS} ≤0.0032%); PF, pediatric formulation; Ph+, Philadelphia chromosome-positive; PK, pharmacokinetic; pt(s), patient(s); QD, once daily; SDS, standard deviation score; TKI, tyrosine kinase inhibitor; US, United States; yrs, years.

References

1. Hijiya N, Suttorp M. *Blood.* 2019;133(22):2374–2384.
2. Samis J, et al. *Pediatr Blood Cancer.* 2016;63(8):1332–1338.
3. Hijiya N, et al. *Blood.* 2016;127(4):392–399.
4. Bower H, et al. *J Clin Oncol.* 2016;34(24):2851–2857.
5. Nevejan L, et al. *Eur J Haematol.* 2024;112(3):458–465.
6. Carofiglio F, et al. *Int J Mol Sci.* 2020;21(12):4469.
7. Gore L, et al. *J Clin Oncol.* 2018;36(13):1330–1338.
8. Hijiya N, et al. *Blood Adv.* 2021;5(14):2925–2934.
9. Scemblix (asciminib) Prescribing information (PI). Available from: https://www.novartis.com/us-en/sites/novartis_us/files/scemblix.pdf. Accessed May 2025.
10. Scemblix (asciminib). https://www.ema.europa.eu/en/documents/product-information/scemblix-epar-product-information_en.pdf. Accessed May 2025.